FAK/PYK2 Inhibitors VS-6063 and VS-4718 Enhance Immune Checkpoint Inhibitor Efficacy

Jennifer E. Ring[1], Yajuan Li[1], Kam Sprott[1], Irina M. Shapiro[1], David DeNardo[2], Yan Wang[1], David T. Weaver[1], and Jonathan A. Pachter[1]

[1]Verastem Inc., Boston, MA; [2]Washington University, St. Louis, MI

ABSTRACT

Although durable responses to single agent immune checkpoint inhibitors have been reported, additional approaches are needed to extend this therapeutic benefit to a greater proportion of cancer patients. Focal Adhesion Kinase (FAK) and the closely related family member PYK2 are potentially valuable targets in this regard due to the roles of these protein kinases in regulating key cellular populations in the tumor microenvironment. In addition to the potency of the small molecule FAK/PYK2 inhibitors VS-6063 and VS-4718 to target cancer stem cells, we have also reported that these agents inhibited monocyte-derived macrophages, decreased IL-6 production from macrophages in vitro, and reduced tumor-associated macrophages in xenograft models.

We now report that VS-6063 and VS-4718 stimulate proliferation of CD8+ cytotoxic T cells.Both FAK/PYK2 inhibitors dose-dependently increased proliferation of primary CD8+ T cells isolated from healthy donor.

Based on the observed enhancement of CD8+ T cells and previously noted inhibition of tumor-associated macrophages, we investigated whether FAK/PYK2 inhibitors potentiate the anti-tumor efficacy of an anti-PD-1 monoclonal antibody in syngeneic mouse tumor models. Combination of VS-4718 with anti-PD-1 extended the median overall survival to 42 days relative to 21, 25 and 28 day median overall survival with vehicle control, single agent anti-PD-1 and single agent VS-4718, respectively. Moreover, on day 56, 30% of mice treated with the VS-4718/anti-PD-1 combination were alive in contrast to the vehicle control, single agent VS-4718, and single agent anti-PD-1 groups in which no mice survived. Studies are currently underway to better understand the immune cell changes in these tumors following VS-4718 and anti-PD-1 combination therapy.

These data provide a rationale for clinical trials in cancer patients to test whether a FAK/PYK2 inhibitor in combination with an immune checkpoint inhibitor could increase the breadth of responsive tumor types, increase the number of responders, and confer a more durable anti-tumor response.

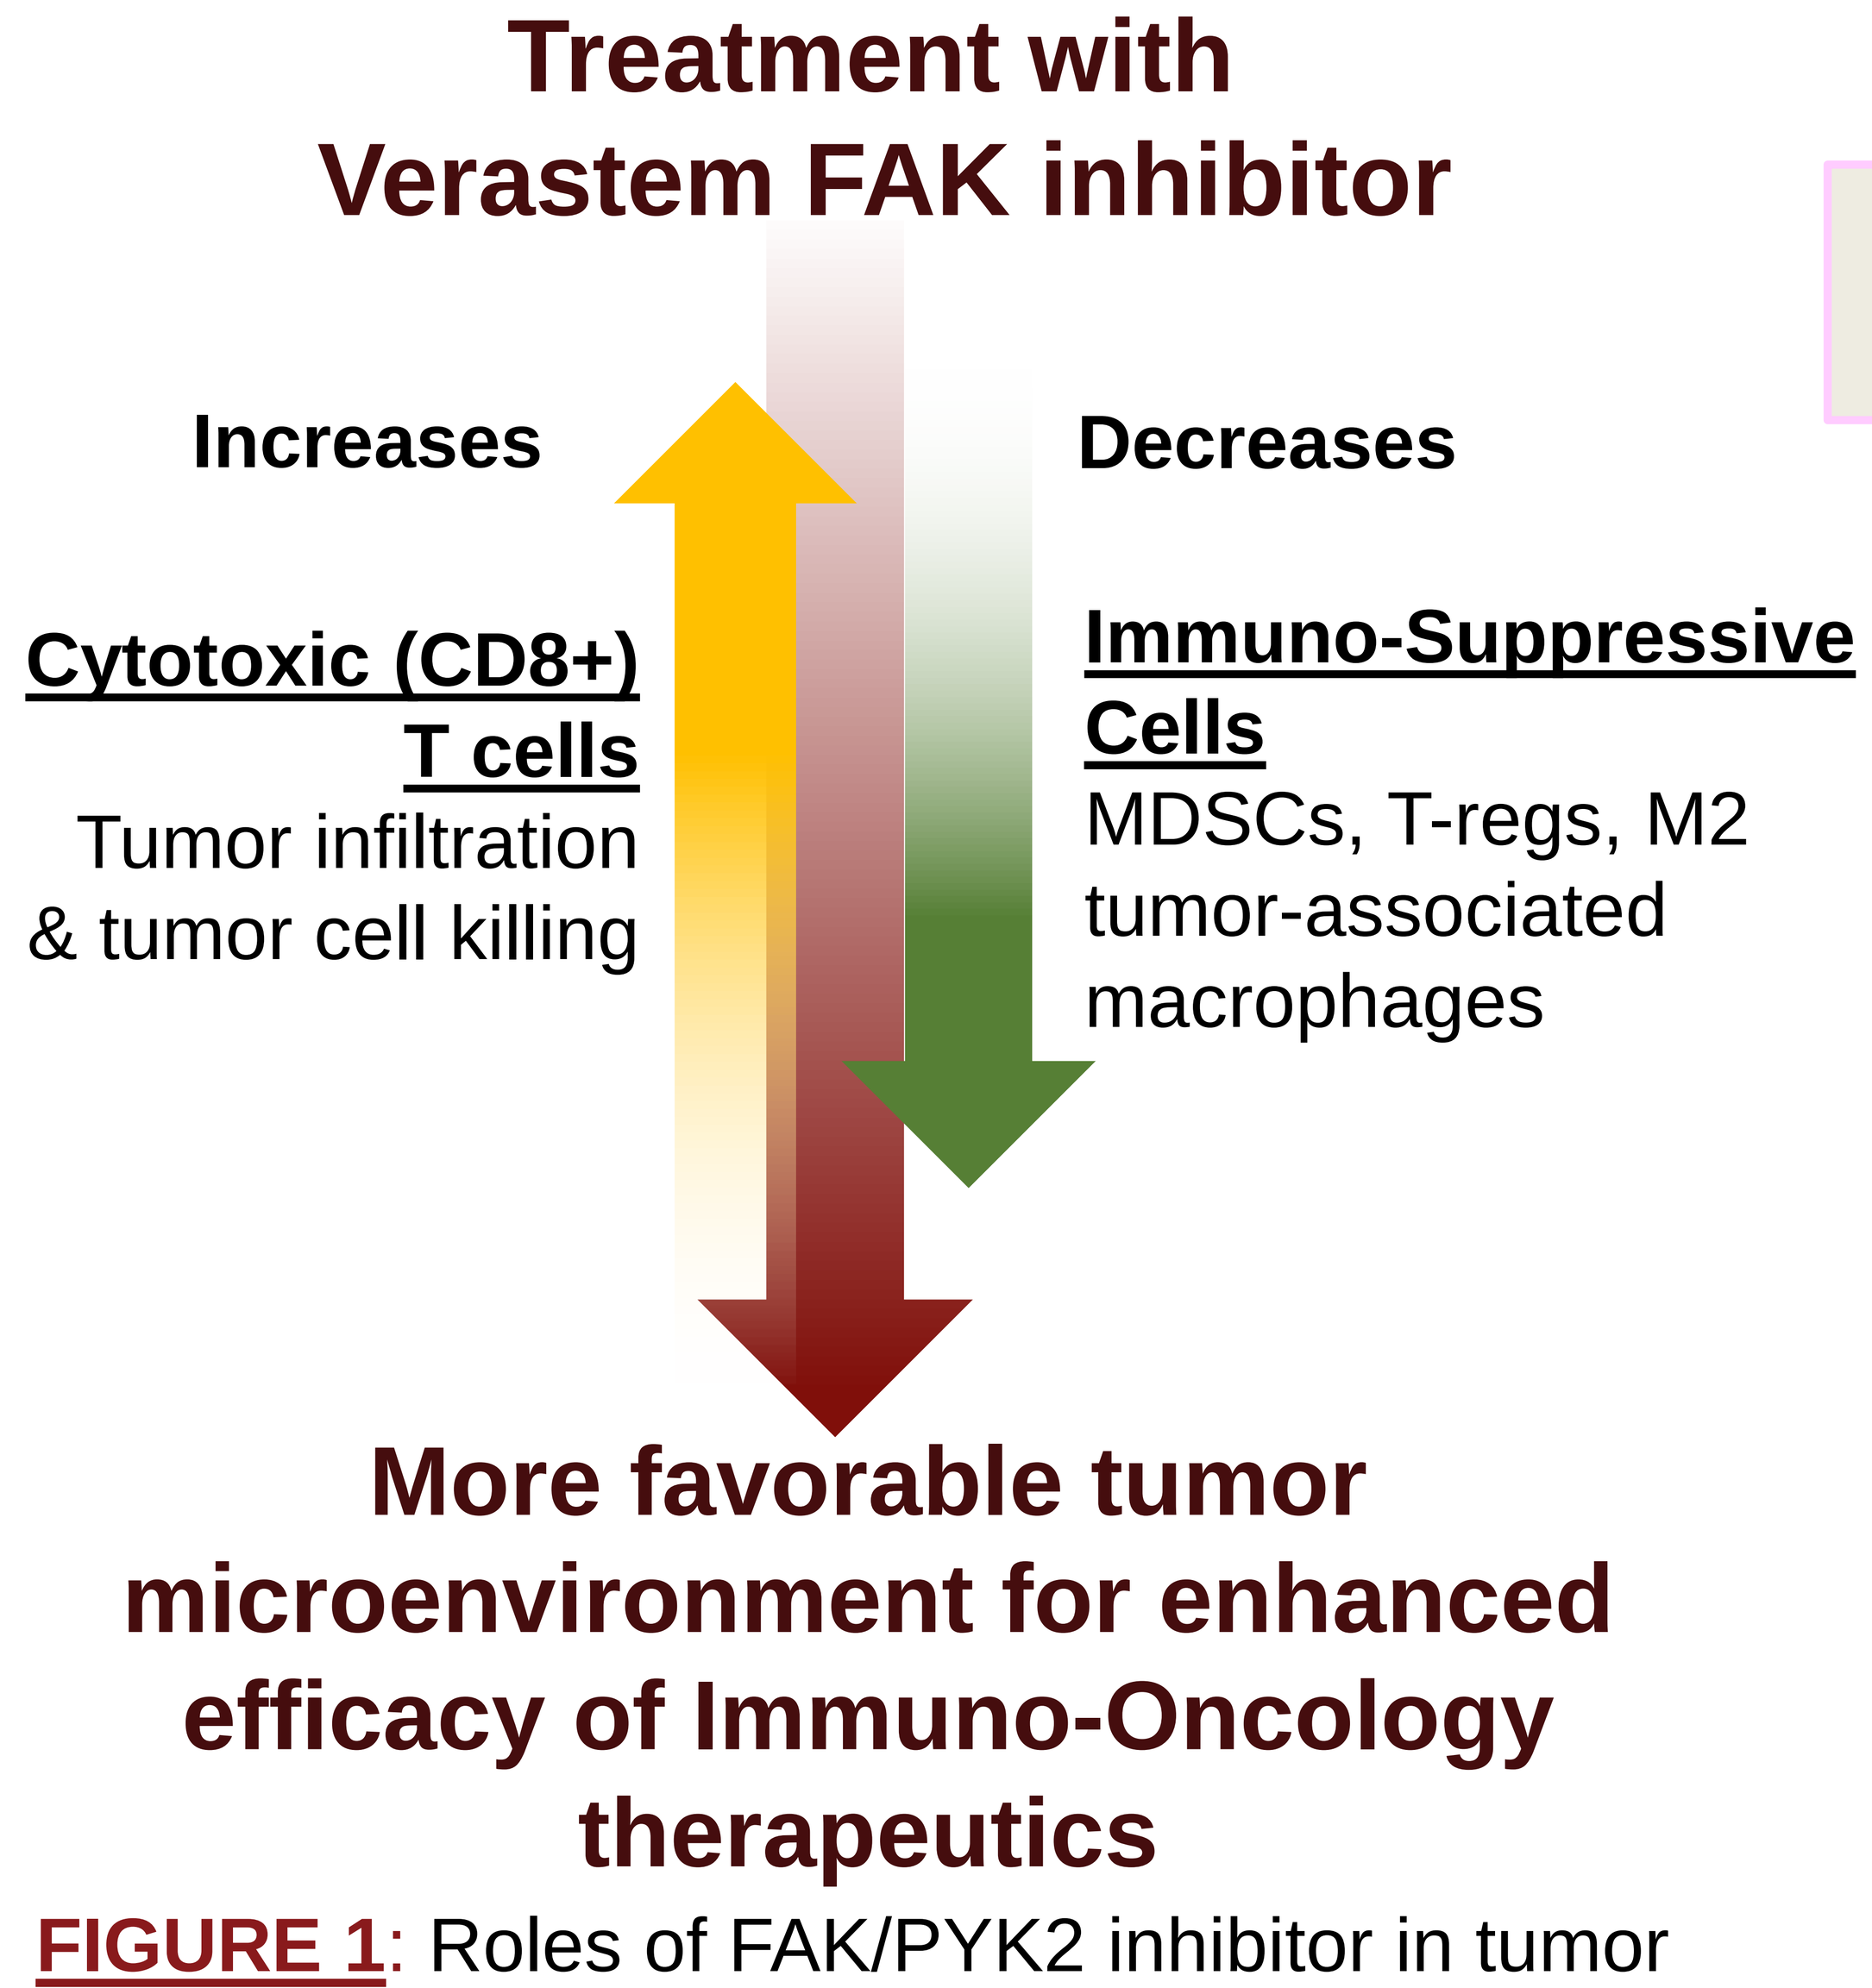

Treatment with
Verastem FAK inhibitor
Increases
Decreases
Cytotoxic (CD8+) T cells
Tumor infiltration & tumor cell killing
Immuno-Suppressive Cells
MDSCs, T-regs, M2 tumor-associated macrophages
More favorable tumor microenvironment for enhanced efficacy of Immuno-Oncology therapeutics

FIGURE 1: Roles of FAK/PYK2 inhibitor in tumor micro-environment

RESULTS

Combination of VS-4718 + anti-PD1 improves survival of MC38 colorectal syngeneic tumor bearing mice

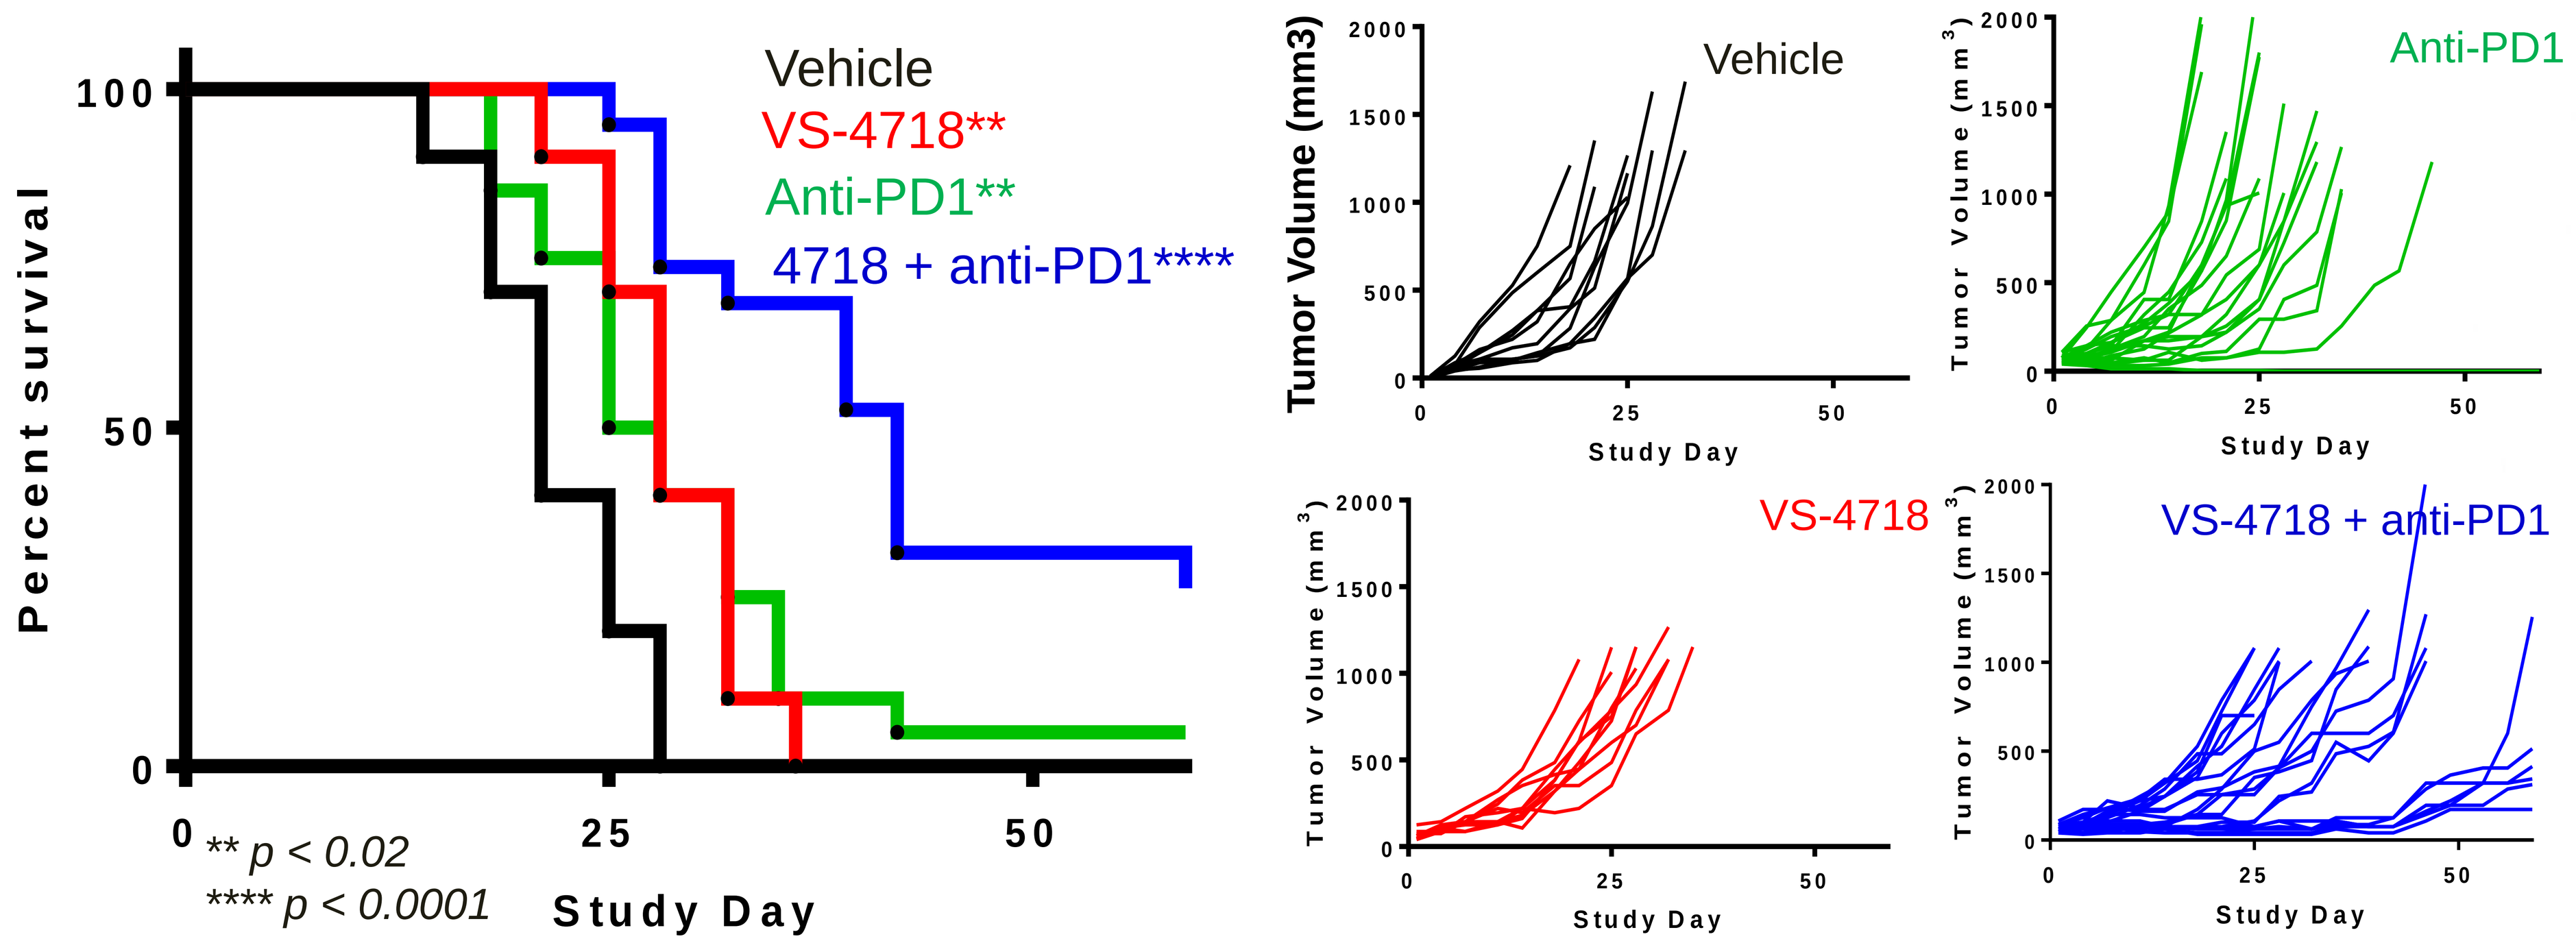

Vehicle
VS-4718**
Anti-PD1**
4718 + anti-PD1****
Percent survival
100
50
0
0
25
50
Study Day
** p < 0.02
**** p < 0.0001
Tumor Volume (mm3)
Study Day
Vehicle
Anti-PD1
VS-4718
VS-4718 + anti-PD1

FIGURE 2: Syngeneic MC38 tumors were randomized once tumors reached 50-100mm^3 and treated with either vehicle, VS-4718 (50 mg/kg, BID), anti-PD1 (clone RMP1-14, 5 mg/kg on days 1, 4, 8, 11) or VS-4718 + anti-PD1 until tumors reached 1000mm^3 as a surrogate survival endpoint

Decreased Tregs and increased CD8+ T cells with VS-4718 + anti-PD1 combination in MC38 tumors

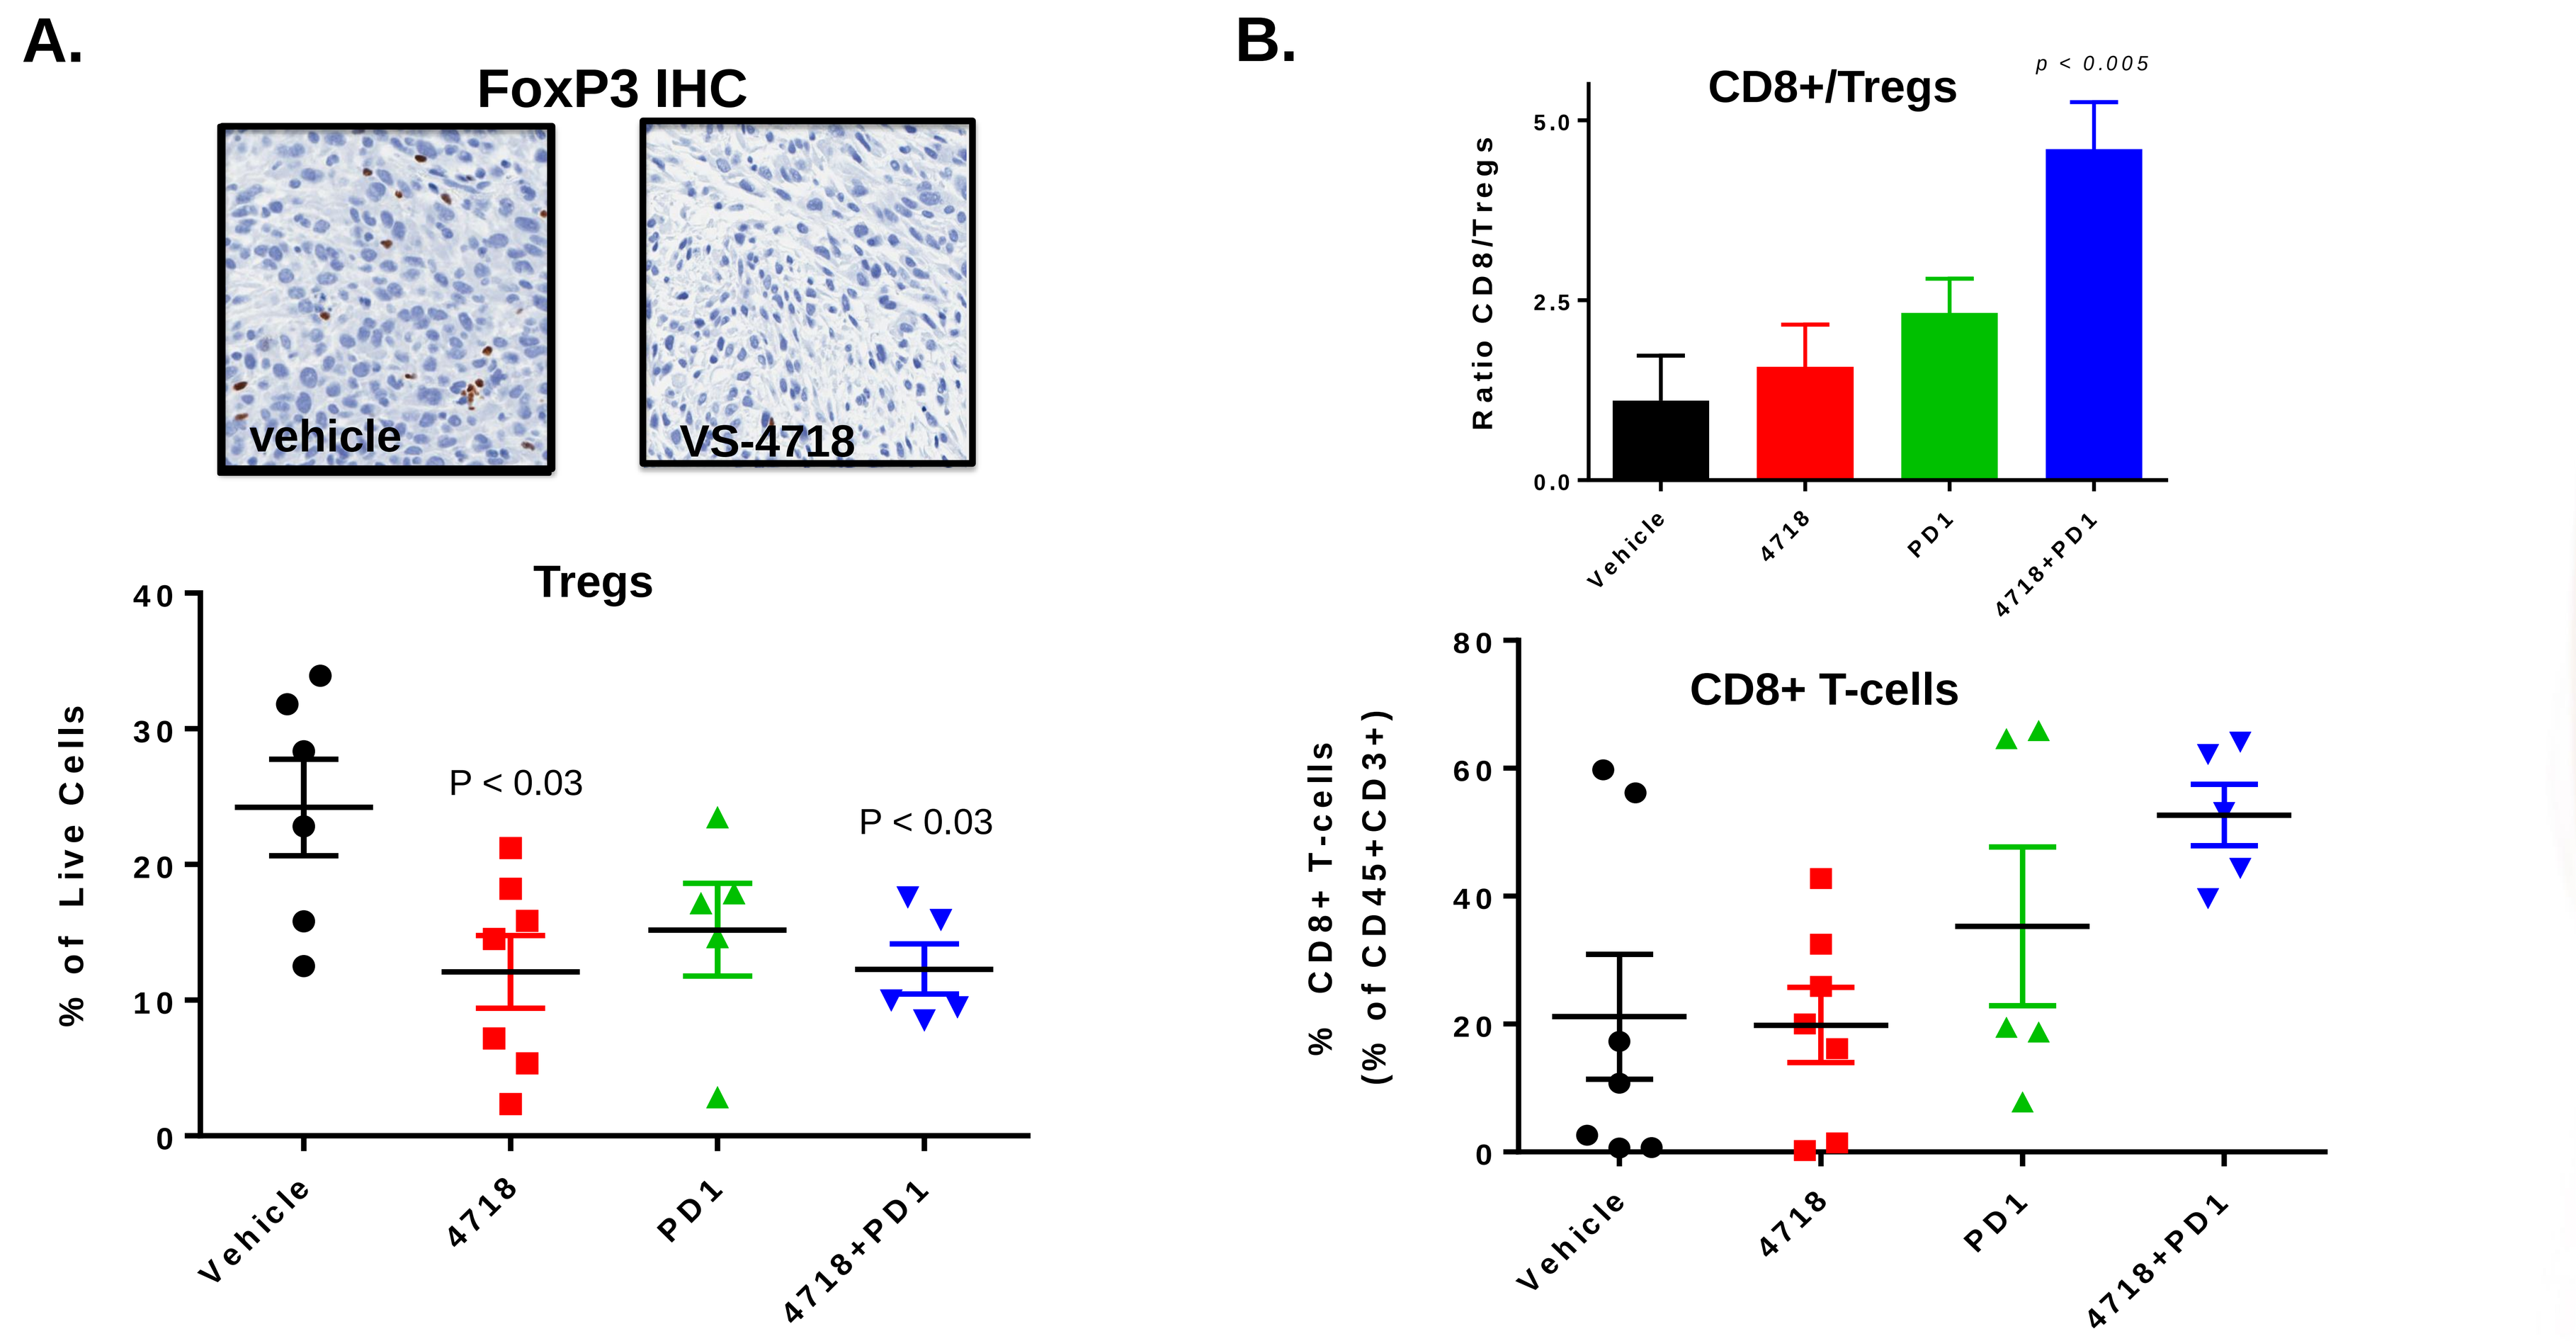

A.
FoxP3 IHC
vehicle
VS-4718
Tregs
% of Live Cells
P < 0.03
P < 0.03
Vehicle
4718
PD1
4718+PD1
B.
CD8+/Tregs
p < 0.005
Ratio CD8/Tregs
CD8+ T-cells
% CD8+ T-cells (% of CD45+CD3+)

FIGURE 3: Syngeneic MC38 tumors were randomized once tumors reached 50-100mm^3 and then treated with either vehicle, VS-4718 (50 mg/kg, BID), anti-PD1 (clone RMP1-14, 5 mg/kg on days 1, 4, 8, 11) or VS-4718 + anti-PD1 for 12 days at which point tumors were harvested and embedded in FFPE for staining Foxp3 by IHC (A) or processed live for flow cytometry (B).

Increased inflammation with VS-4718 + anti-PD1 combination in MC38 tumors

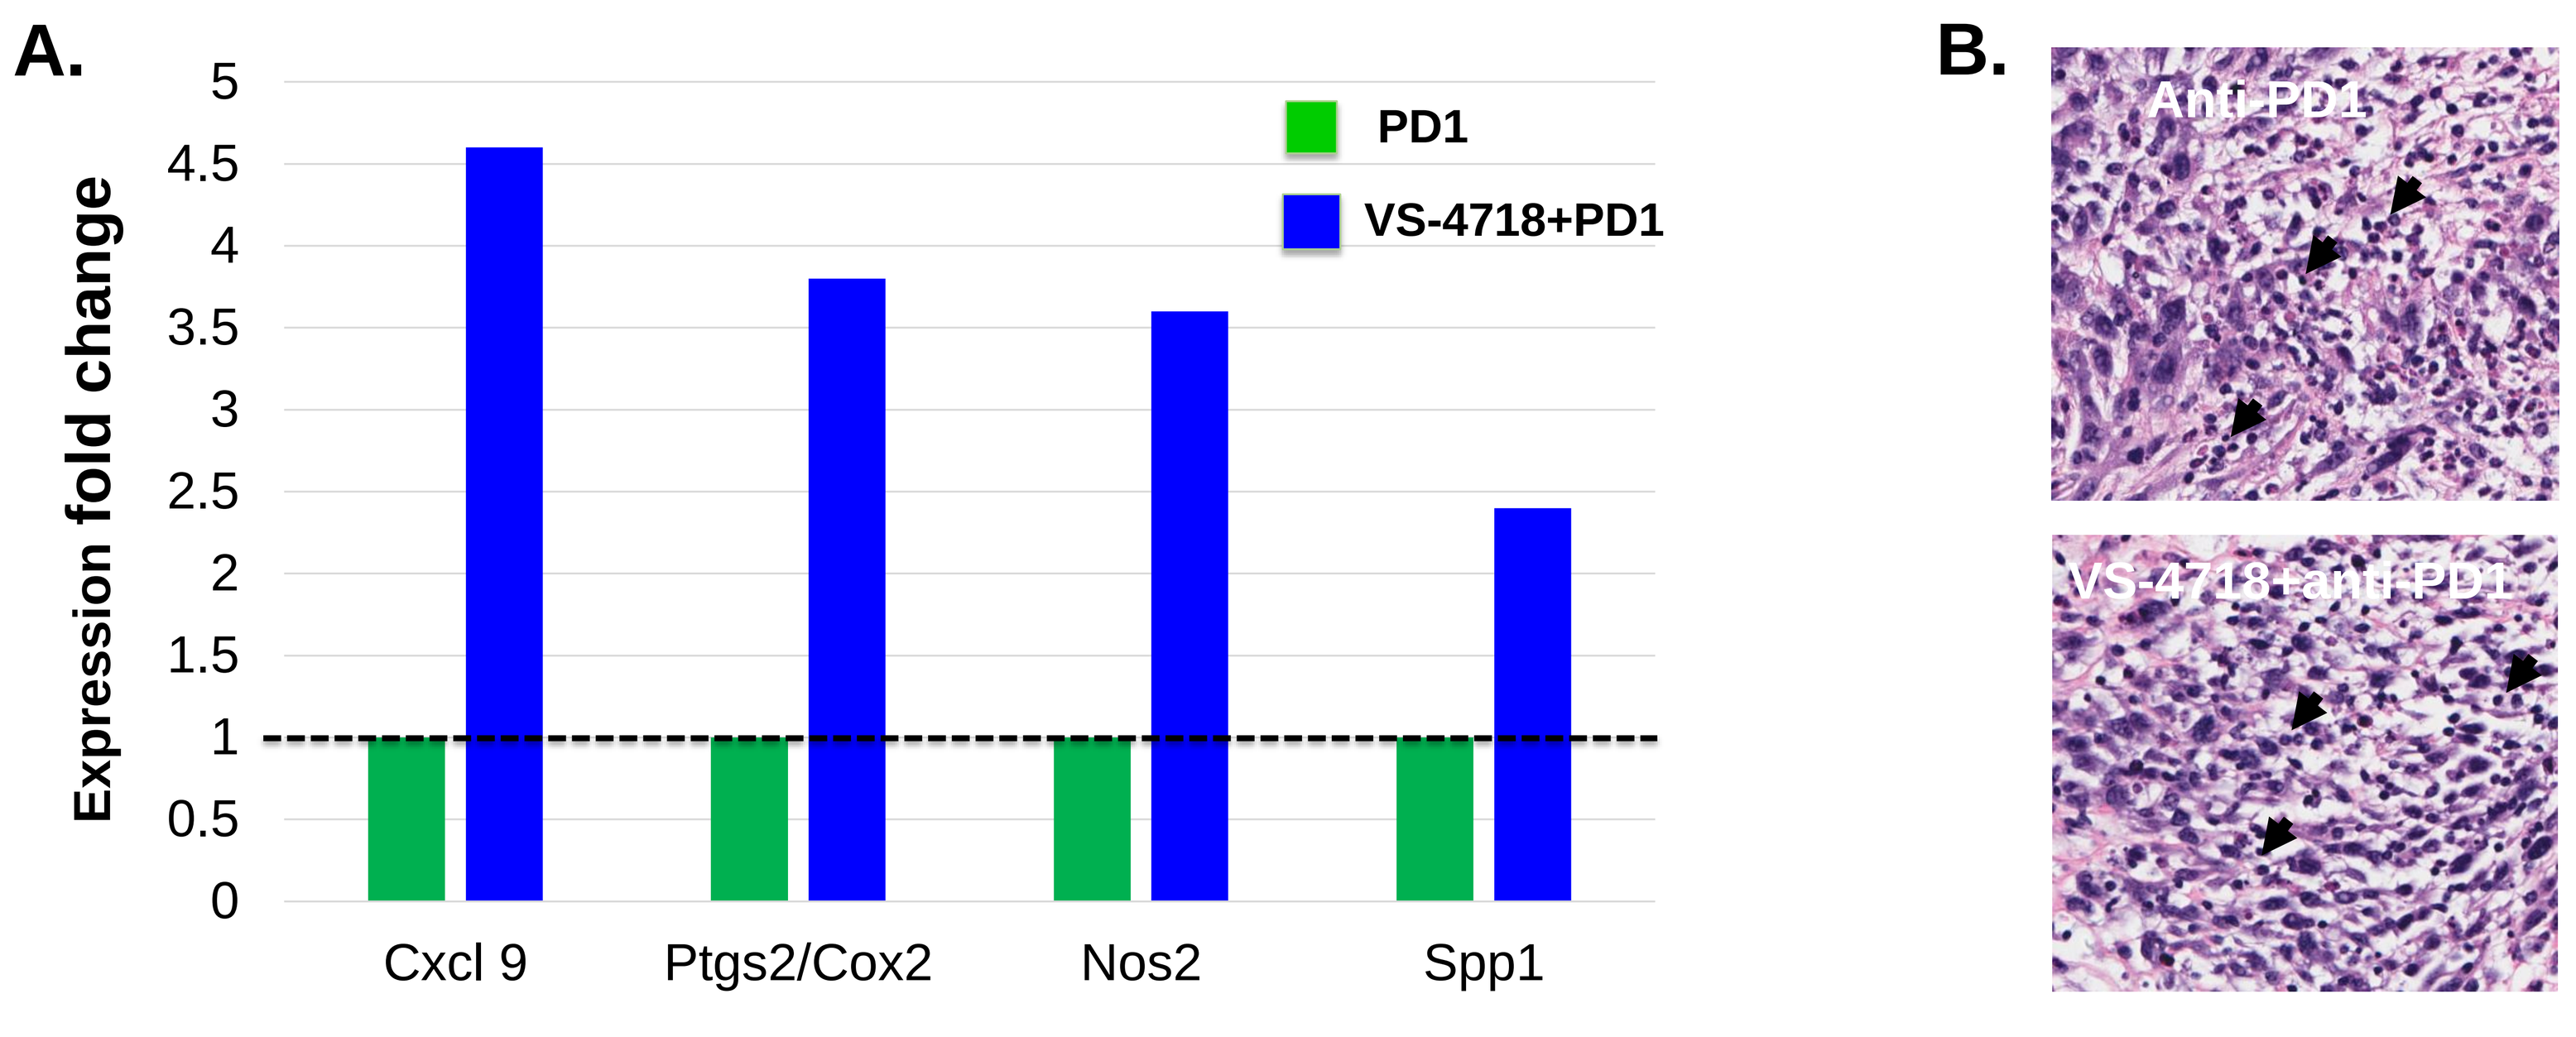

A.
PD1
VS-4718+PD1
Expression fold change
Cxcl 9
Ptgs2/Cox2
Nos2
Spp1
B.
Anti-PD1
VS-4718+anti-PD1

Figure 4: Syngeneic MC38 tumors were randomized once tumors reached 50-100mm^3 and then treated with either anti-PD1 (clone RMP1-14, 5 mg/kg on days 1, 4, 8, 11) or VS-4718 (50 mg/kg, BID) + anti-PD1 for 12 days at which point tumors were harvested in RNALater and analyzed for changes in inflammatory cytokines/chemokines by qPCR (A) or embedded in FFPE and stained with H&E (B).

VS-4718 and VS-6063 enhance human CD8+ T cell proliferation

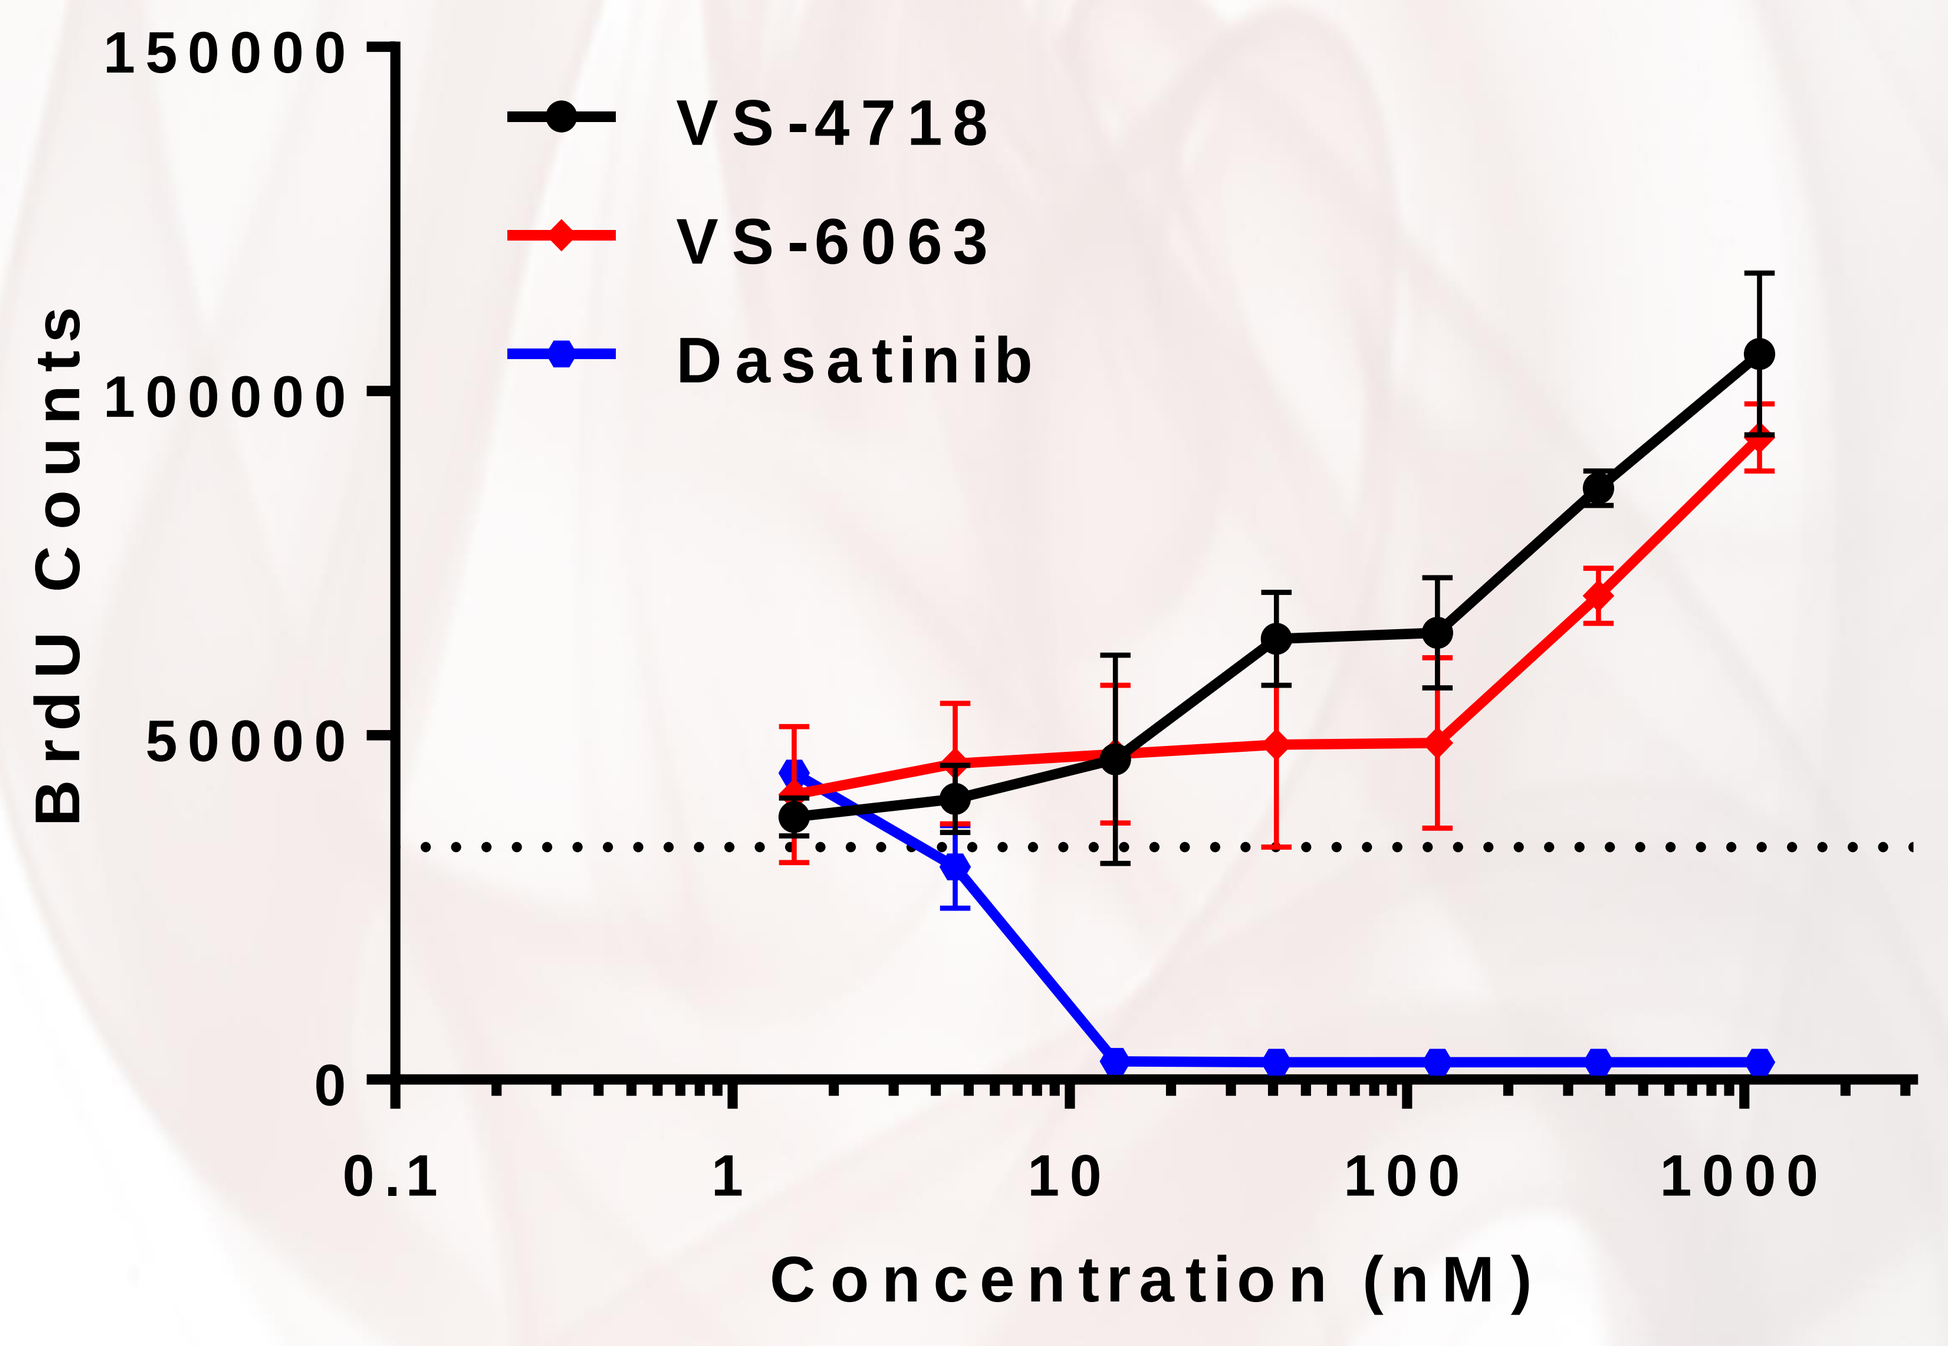

VS-4718
VS-6063
Dasatinib
BrdU Counts
Concentration (nM)

FIGURE 5: CD8+ T cells were isolated from hPBMCs by magnetic bead separation and then plated on CD3-coated plates in the presence or absence of VS-4718 or VS-6063 for 72 hours. Assay wells were pulsed with BrdU for the last 3-4 hours and subjected to a BrdU-incorporation assay for the determination of proliferating cells. Data are presented as a fold-change of DMSO control wells.

VS-4718 and VS-6063 decrease CD8+ T cell exhaustion markers

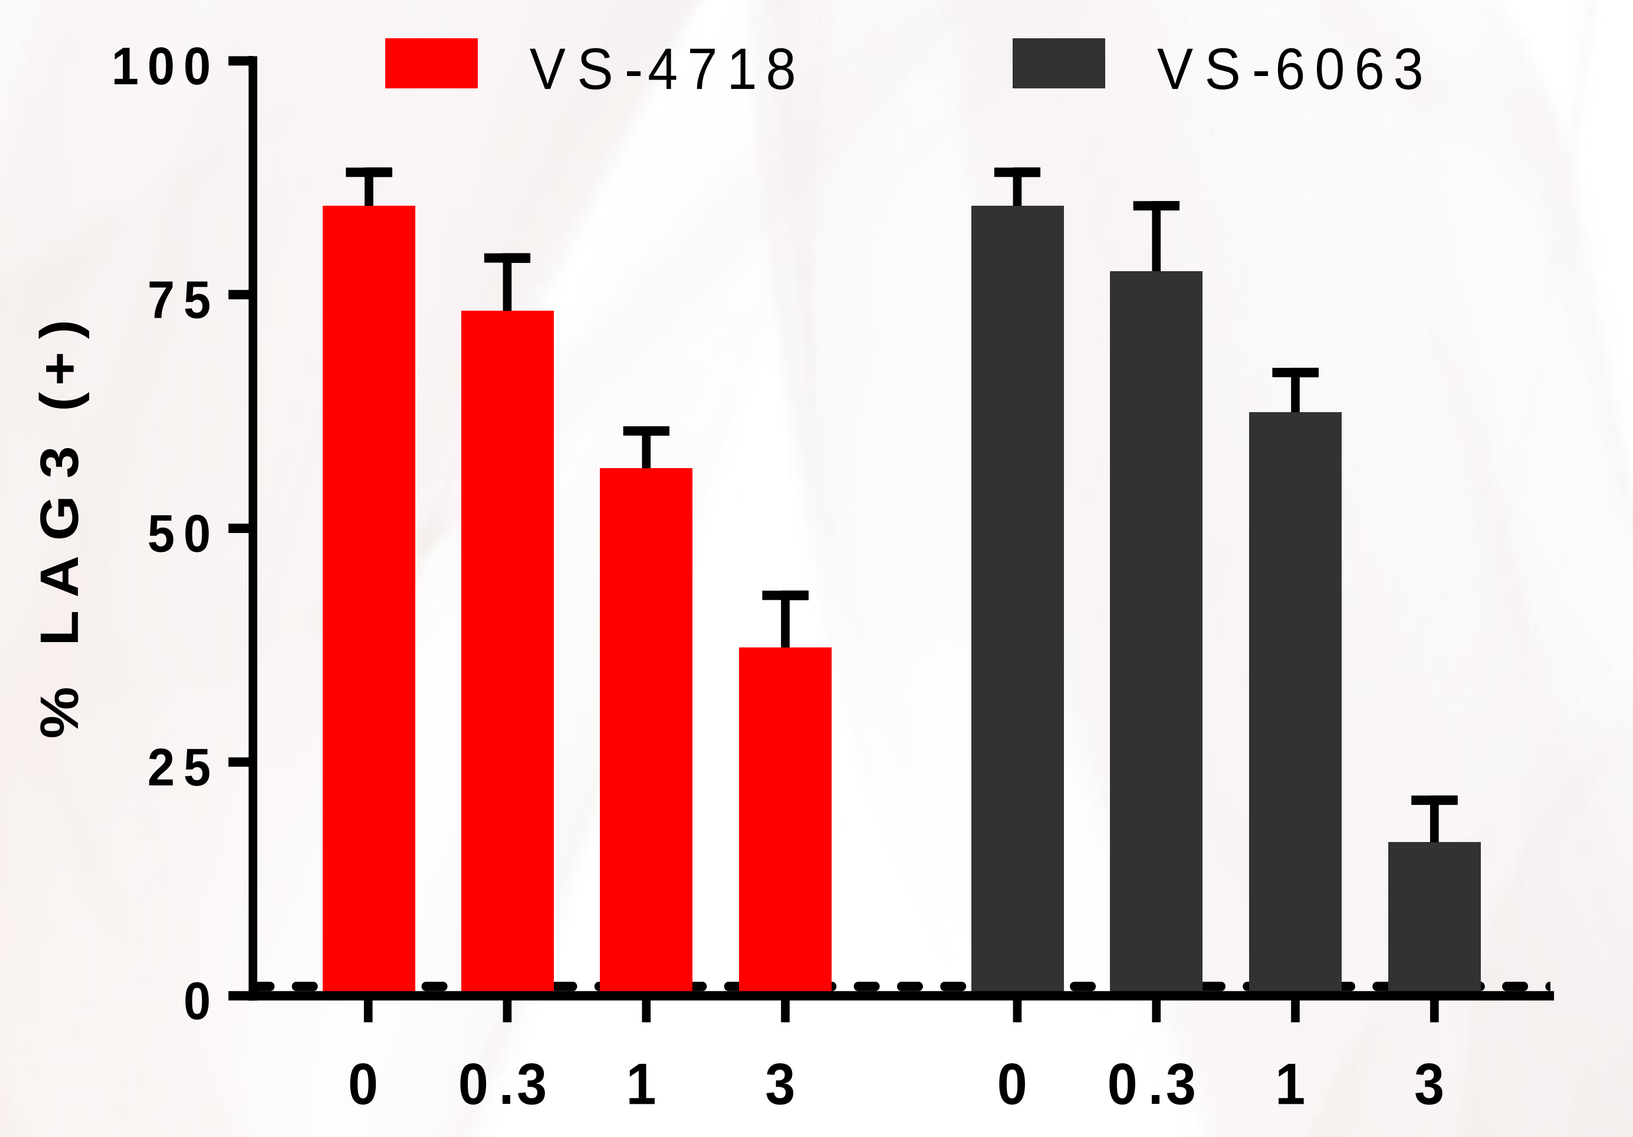

VS-4718
VS-6063
% LAG3 (+)
0
0.3
1
3

FIGURE 6: CD8+ T cells isolated as in Figure 5 were plated on anti-CD3 coated plates in the presence of VS-4718 or VS-6063 for 72-hours and then harvested and stained with anti-LAG3 by flow cytometric analysis. Similar reduction of PD1 is also observed.

FAK inhibitor enhances CD8+ cytolytic activity against cognate tumor cells

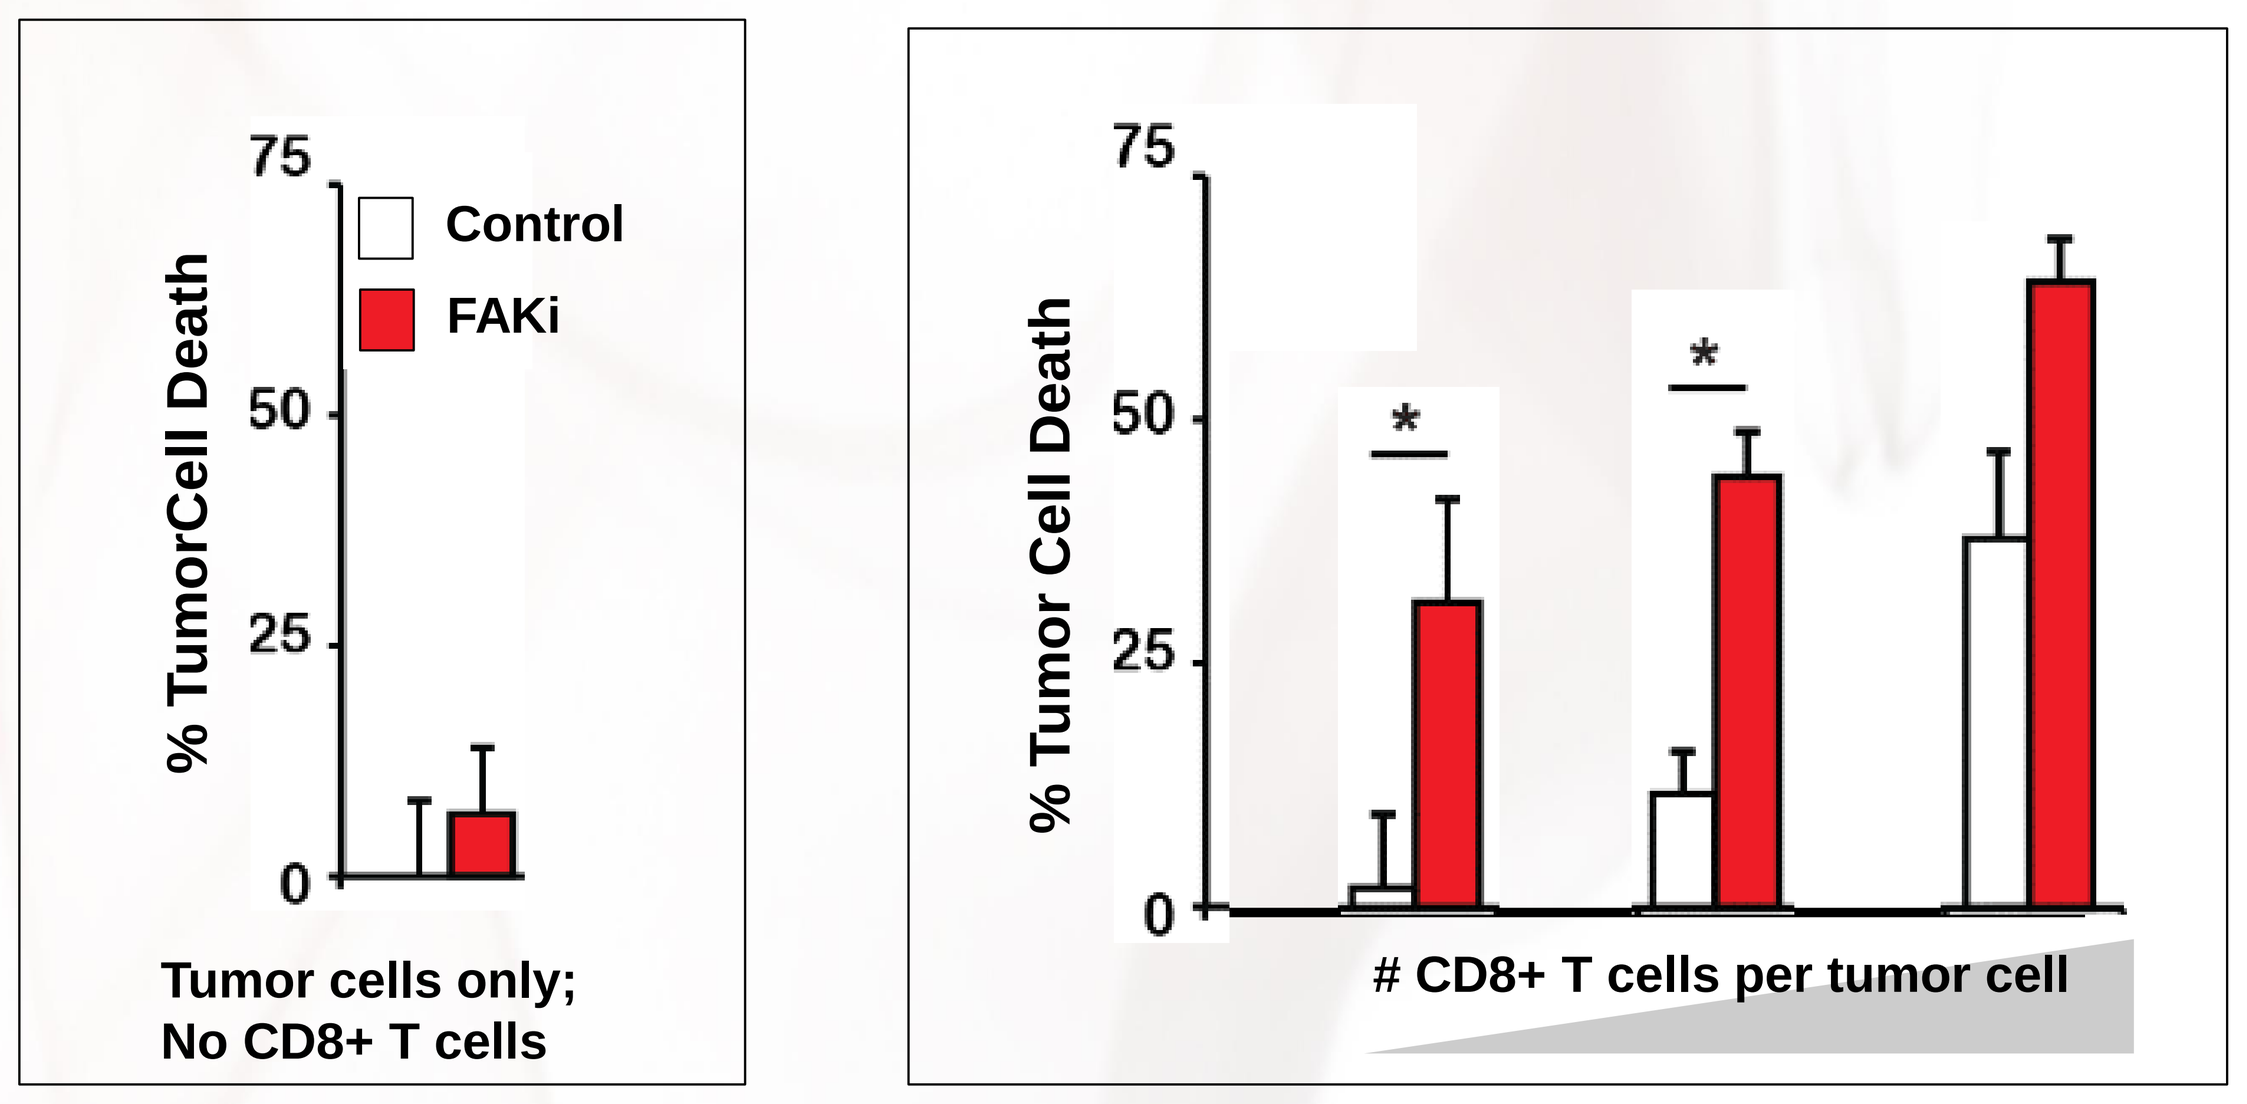

Control
FAKi
% TumorCell Death
Tumor cells only; No CD8+ T cells
% Tumor Cell Death
CD8+ T cells per tumor cell

FIGURE 7: Antigen-specific CD8+ T cells were co-cultured with cognate luciferized tumor cells at various E:T ratios in the presence of 1 μM VS-4718 on top of Matrigel. After 24 hours, cultured were analyzed for luciferase as a measure of tumor cell viability.

SUMMARY

- **Combination of FAK inhibitor + anti-PD1 improves survival of MC38 tumor-bearing animals**
- **FAK inhibitor + anti-PD1 combination induces decreased Tregs and increased CD8+ T cells and inflammatory factors in the MC38 tumors**
- **FAK inhibitors increase CD8+ T cell proliferation, decrease CD8+ T cell exhaustion markers, and increase T cell-mediated tumor cell killing in vitro**
- **These data provide rationale for clinical trials with a FAK inhibitor in combination with an immune checkpoint inhibitor**