Dual PI3K-δ,γ Inhibitor Duvelisib Reduces Immunosuppressive Tregs and Myeloid Cells, Enhancing Efficacy of Checkpoint and Co-Stimulatory Antibodies in a B Cell Lymphoma Model

Poster 33

Jonathan A. Pachter and David T. Weaver

Verastem, Inc., Needham, MA

Concept

- Duvelisib is clinically active as a monotherapy in B cell malignancies
- PI3K-δ inhibition is known to reduce immunosuppressive Tregs
- PI3K-γ inhibition is known to reduce immunosuppressive myeloid cells
- Duvelisib may potentiate efficacy of checkpoint & co-stimulatory antibodies against B cell malignancies

Abstract

BACKGROUND: Duvelisib is a dual inhibitor of PI3K-δ and PI3K-γ which has shown clinical activity as monotherapy in CLL/SLL, FL and T cell lymphoma.[1] Recent publications have demonstrated that PI3K-δ inhibition reduces immunosuppressive Tregs[2] whereas PI3K-γ inhibition reduces immunosuppressive myeloid cells.[3] Hence, we postulated that duvelisib may augment the efficacy of immune checkpoint or co-stimulatory antibodies.

METHODS: Mice bearing syngeneic A20 B cell lymphoma tumors (60-90 mm^3) were treated with vehicle, duvelisib, anti-PD-1, anti-PD-1 + duvelisib, anti-OX40, or anti-OX40 + duvelisib. Tumor volumes were measured by caliper. Tregs, macrophages and MDSCs were quantified by flow cytometry from mice bearing A20 tumors after 8 days of treatment.

RESULTS: In the A20 model, duvelisib, anti-PD-1 and anti-OX40 treatments each induced tumor growth delay. When duvelisib and anti-PD-1 were combined in mice with pre-existing A20 tumors, strong anti-tumor synergy was observed. When anti-OX40 and duvelisib were combined, tumor regression was observed which correlated with strong reduction of tumor Tregs, M2 macrophages and MDSCs. To assess immune memory, tumor-free mice following anti-OX40 alone or anti-OX40 + duvelisib were injected with A20 cells in the contralateral flank with no further treatment. Whereas mice that had received anti-OX40 alone grew new tumors upon A20 re-challenge, all tumor-free mice that had received anti-OX40 + duvelisib did not grow tumors upon re-challenge and showed elevated memory T cells in blood and spleen. These findings indicate that anti-OX40 + duvelisib treatment established immune memory.

CONCLUSIONS: The dual inhibition of PI3K-δ and PI3K-γ appears to make duvelisib especially effective in reducing both lymphoid and myeloid immuno-suppressive populations, enhancing the anti-tumor efficacy of immune checkpoint and co-stimulatory antibodies. These data support potential clinical exploration of duvelisib in combination with checkpoint or co-stimulatory antibodies.

Duvelisib is a Dual Inhibitor of PI3K-δ & PI3K-γ

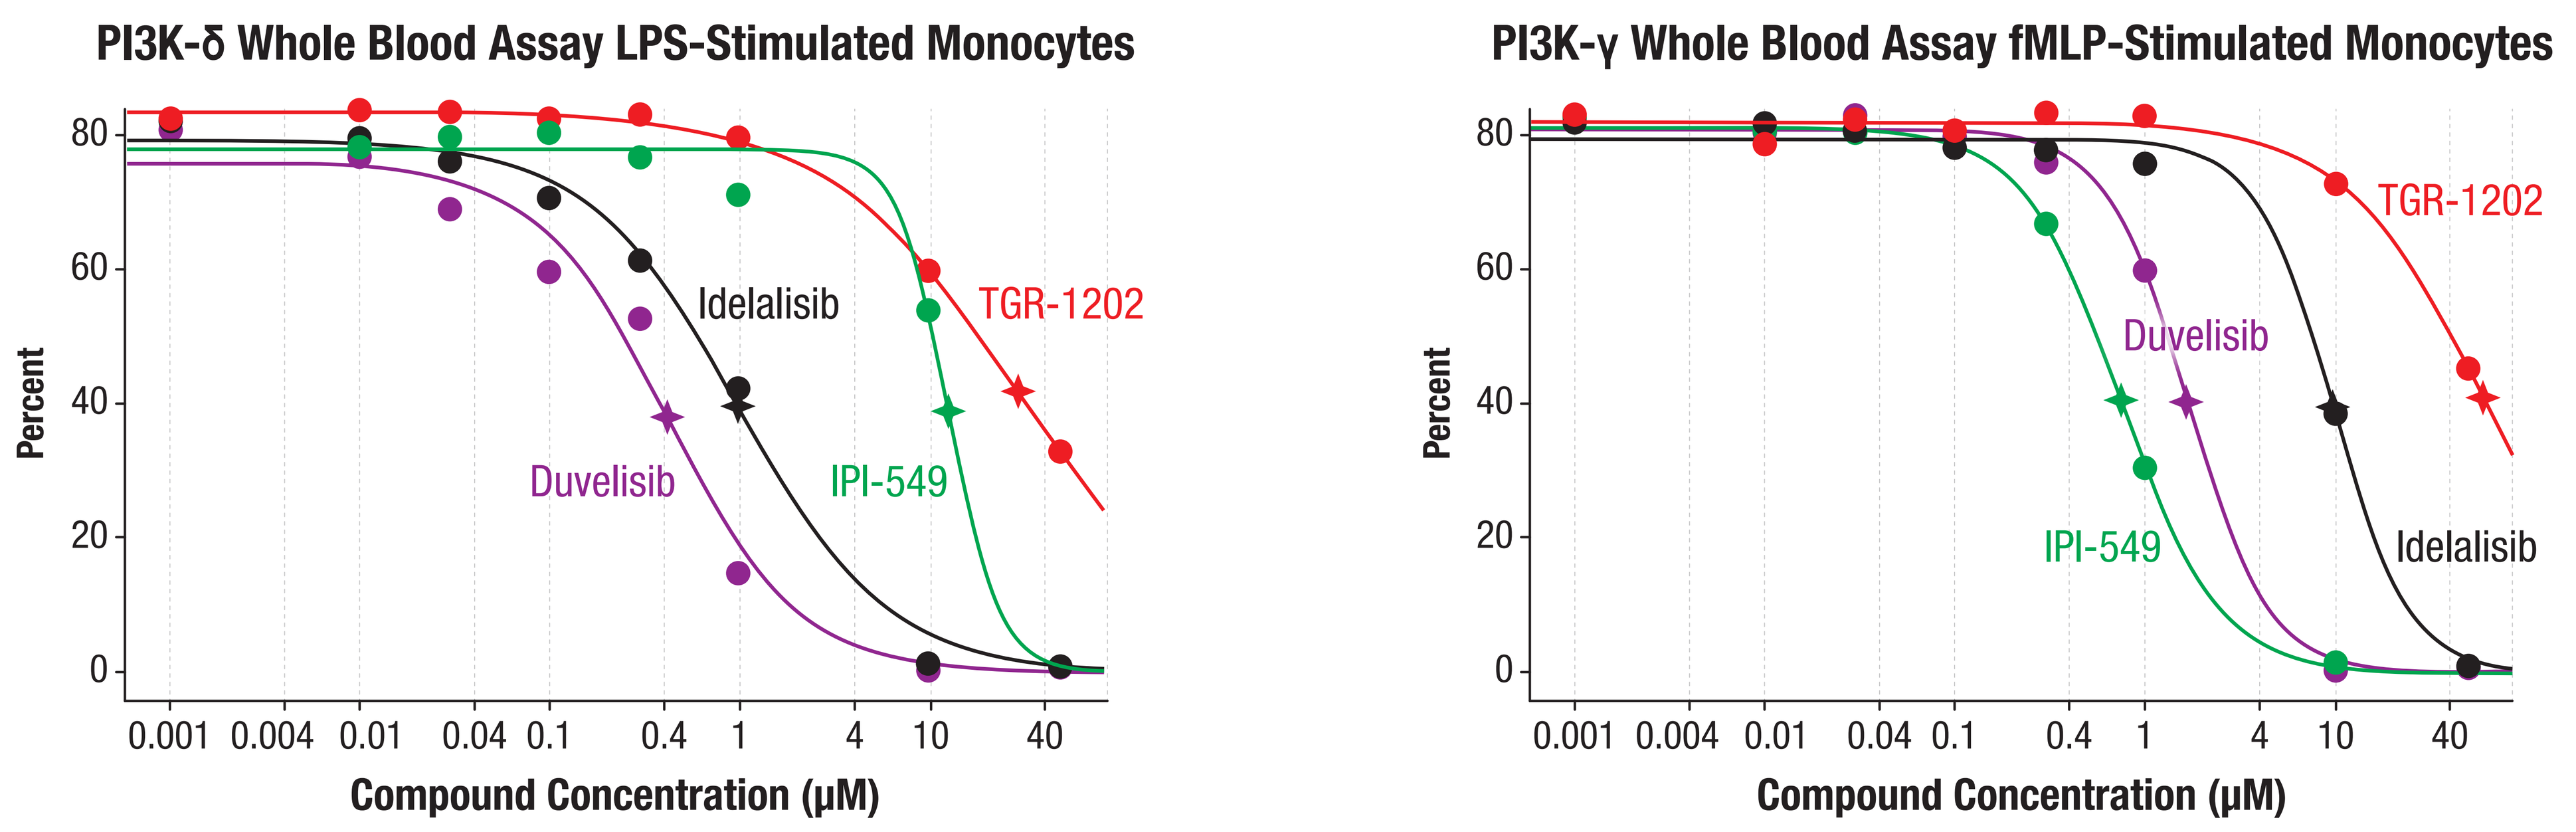
PI3K-δ Whole Blood Assay LPS-Stimulated Monocytes
PI3K-γ Whole Blood Assay fMLP-Stimulated Monocytes
Percent
Compound Concentration (μM)
Duvelisib
Idelalisib
IPI-549
TGR-1202

Drug	Mechanism of Action	PI3K-δ IC_{50} (μM)	PI3K-γ IC_{50} (μM)	C_{max} (ss) (μM)
Duvelisib	Dual PI3K-δ/PI3K-γ Inhibitor	0.4 ± 0.1	1.6 ± 0.2	2.5
Idelalisib	PI3K-δ Inhibitor	1.0 ± 0.2	9.4 ± 2.3	4.8
IPI-549	PI3K-γ Inhibitor	12 ± 0.5	0.5 ± 0.2	9.1
TGR-1202		25 ± 8	55 ± 16	3.1

Figure 1: Duvelisib is a dual inhibitor of PI3K-δ & PI3K-γ. Inhibition of LPS-stimulated monocytes and fMLP-stimulated monocytes were used to measure whole blood potencies of PI3K inhibitors against PI3K-δ and PI3K-γ, respectively. The graphs show dose responses with monocytes from human donors. In the table, whole blood assay IC_{50} values, which encompass enzyme inhibition, cell penetration and protein binding of inhibitors, are related to reported clinical plasma exposures of each agent at RP2D. Only duvelisib covers IC_{50} values for both PI3K-δ and PI3K-γ at clinically achievable exposures.

- Duvelisib human PK. C_{max} @ 25 mg BID (RP2D) = 1062 ng/ml; MW = 417 g/mol
- Idelalisib human PK data from Webb, ASH 2010. C_{max} @ 150 mg BID (RP2D) = 2000 ng/ml; MW = 415 g/mol
- IPI-549 human PK from Hong, SITC 2017. $C_{max\text{-}ss}$ @ 60 mg QD (RP2D) = 4800 ng/ml, MW = 529 g/mol
- TGR-1202 human PK from Savona, ASH 2013. $C_{max\text{-}ss}$ @ 800 mg QD (RP2D) = 1800 ng/ml, MW = 572 g/mol

Combination of Duvelisib + anti-PD-1 is Synergistic in Induction of Tumor Growth Inhibition and Survival in A20 B Cell Lymphoma Model

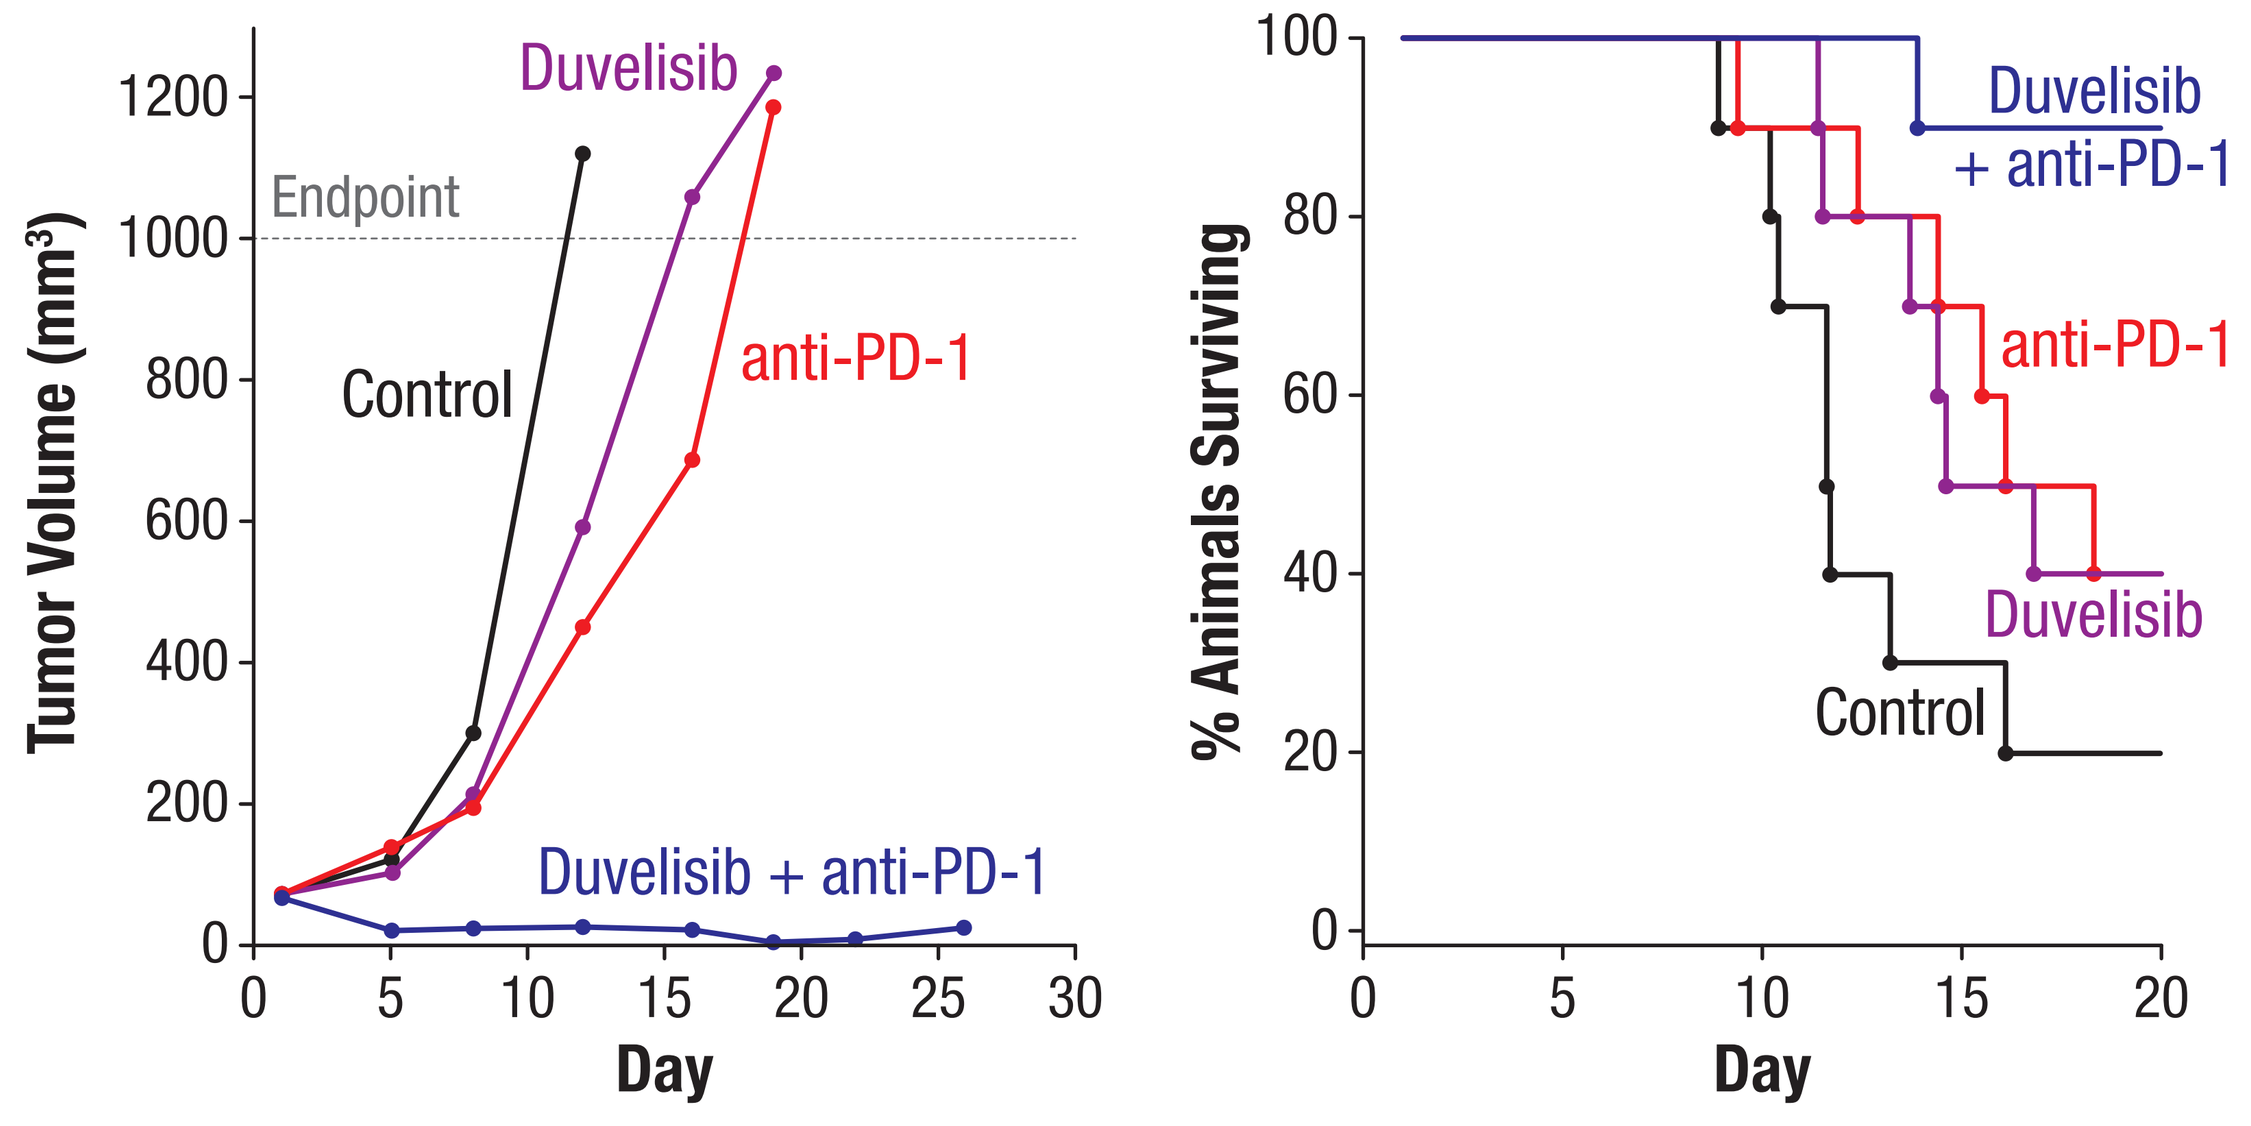
Tumor Volume (mm³)
Endpoint
Control
Duvelisib
anti-PD-1
Duvelisib + anti-PD-1
% Animals Surviving
Day

Figure 2: Mice bearing syngeneic A20 B cell lymphoma tumors were randomized once tumors reached 60-90 mm^3 and treated with either vehicle + rat IgG2a control, duvelisib (50 mg/kg, BID, po through end of experiment) + rat IgG2a control, anti-PD-1 (clone RMP1-14, 100 mg/mouse, i.p. biweekly x 2), or duvelisib + anti-PD-1. Tumor volume was measured by caliper. For Kaplan-Meier, an event was defined by death or tumor growth beyond 1000 mm^3.

Combination of Duvelisib + anti-OX40 Induces Tumor Regressions and Long-Term Survival in A20 B Cell Lymphoma Model

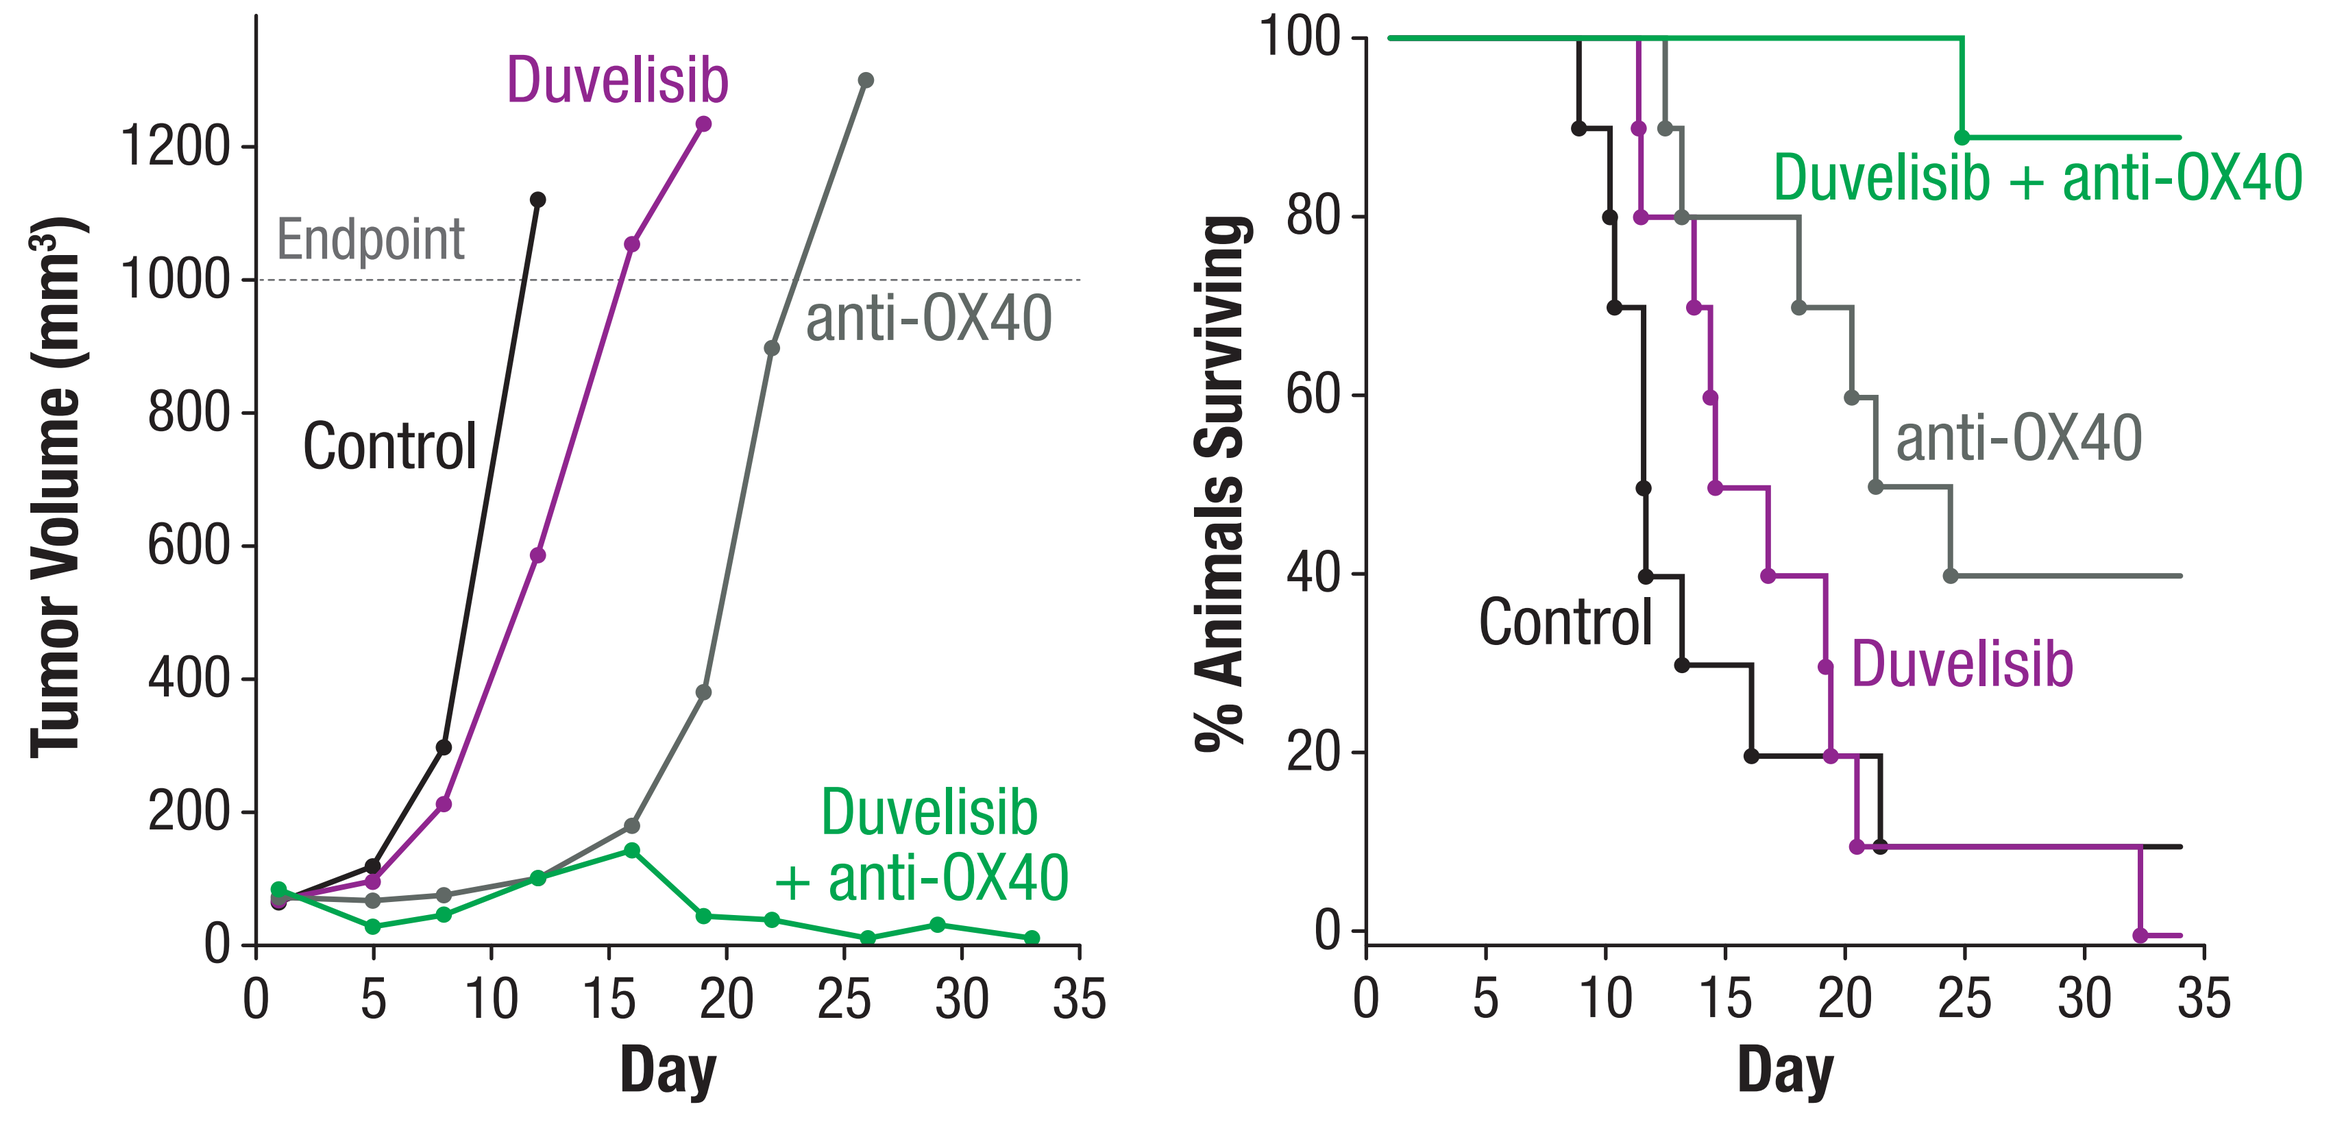
Tumor Volume (mm³)
Endpoint
Control
Duvelisib
anti-OX40
Duvelisib + anti-OX40
% Animals Surviving
Day

Figure 3: Mice bearing syngeneic A20 B cell lymphoma tumors were randomized once tumors reached 60-90 mm^3 and treated with either vehicle + rat IgG2a control, duvelisib (50 mg/kg, BID, po through end of experiment) + rat IgG2a control, anti-OX40 (clone OX86, 100 mg/mouse, i.p. biweekly x 2), or duvelisib + anti-OX40.

Combination of Duvelisib + anti-OX40 Induces Tumor Regressions

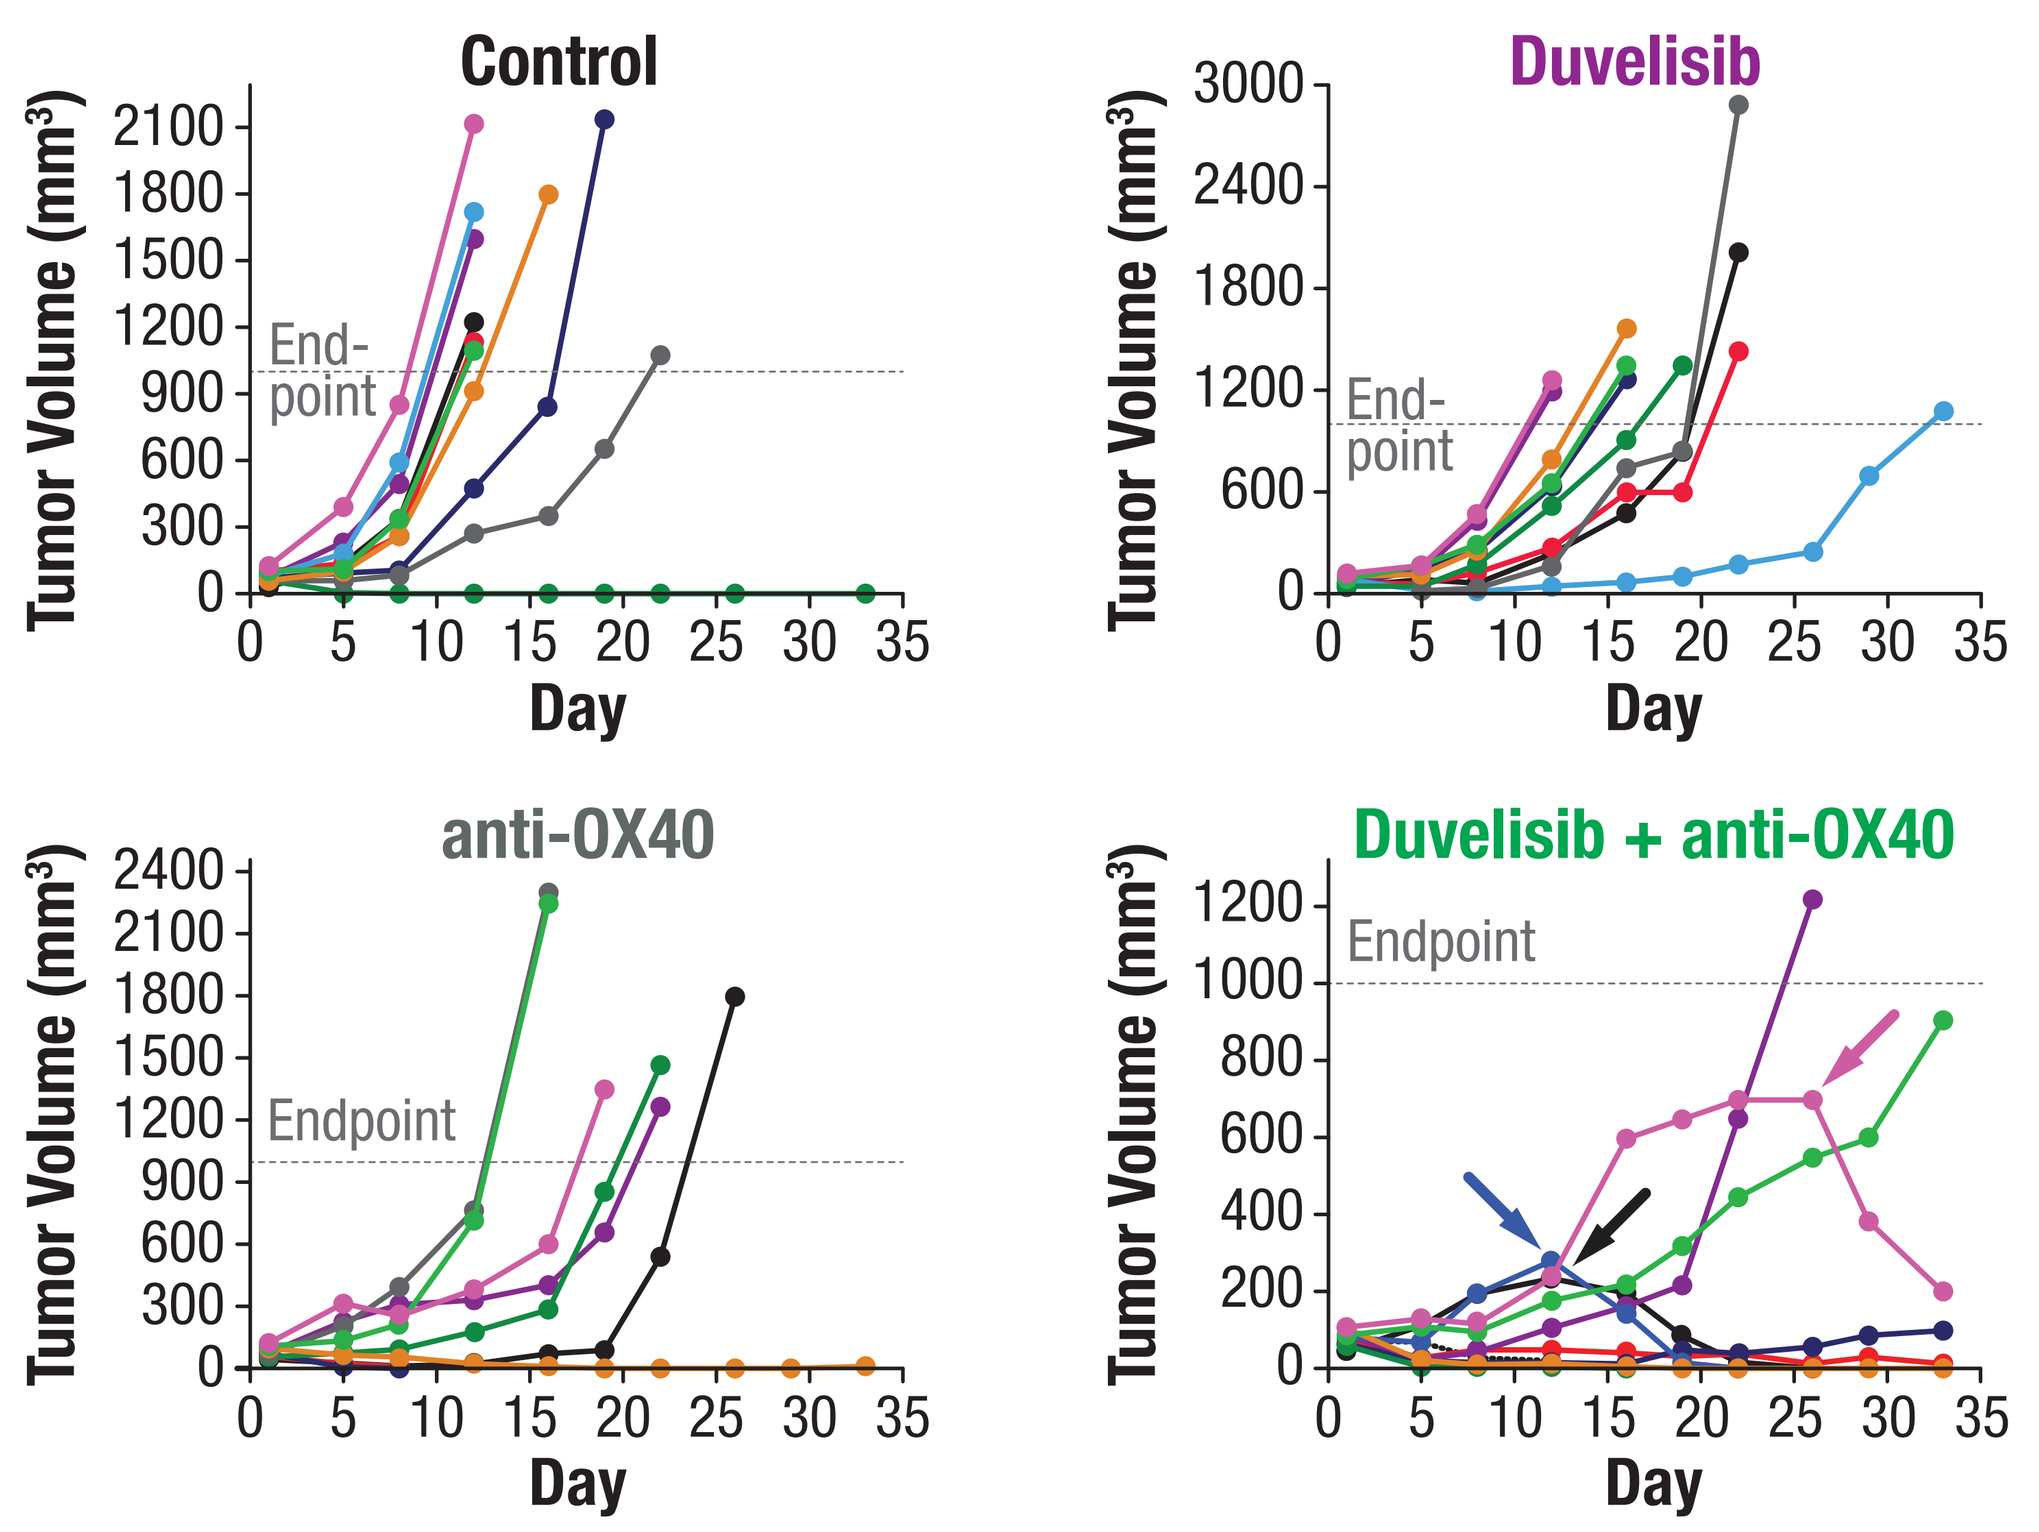
Control
Duvelisib
anti-OX40
Duvelisib + anti-OX40
Tumor Volume (mm³)
End-point
Endpoint
Day

Figure 4: Mice bearing A20 B cell lymphoma tumors were treated with vehicle + rat IgG2a control, duvelisib (50 mg/kg, BID, po through end of experiment) + rat IgG2a control, anti-OX40 (100 mg/mouse, i.p. biweekly x 2), or duvelisib + anti-OX40. Curves show tumor growth for individual mice in each group. Only mice treated with the combination of duvelisib + anti-OX40 show tumor regression.

Combination of Duvelisib + anti-OX40 Induces Immune Memory

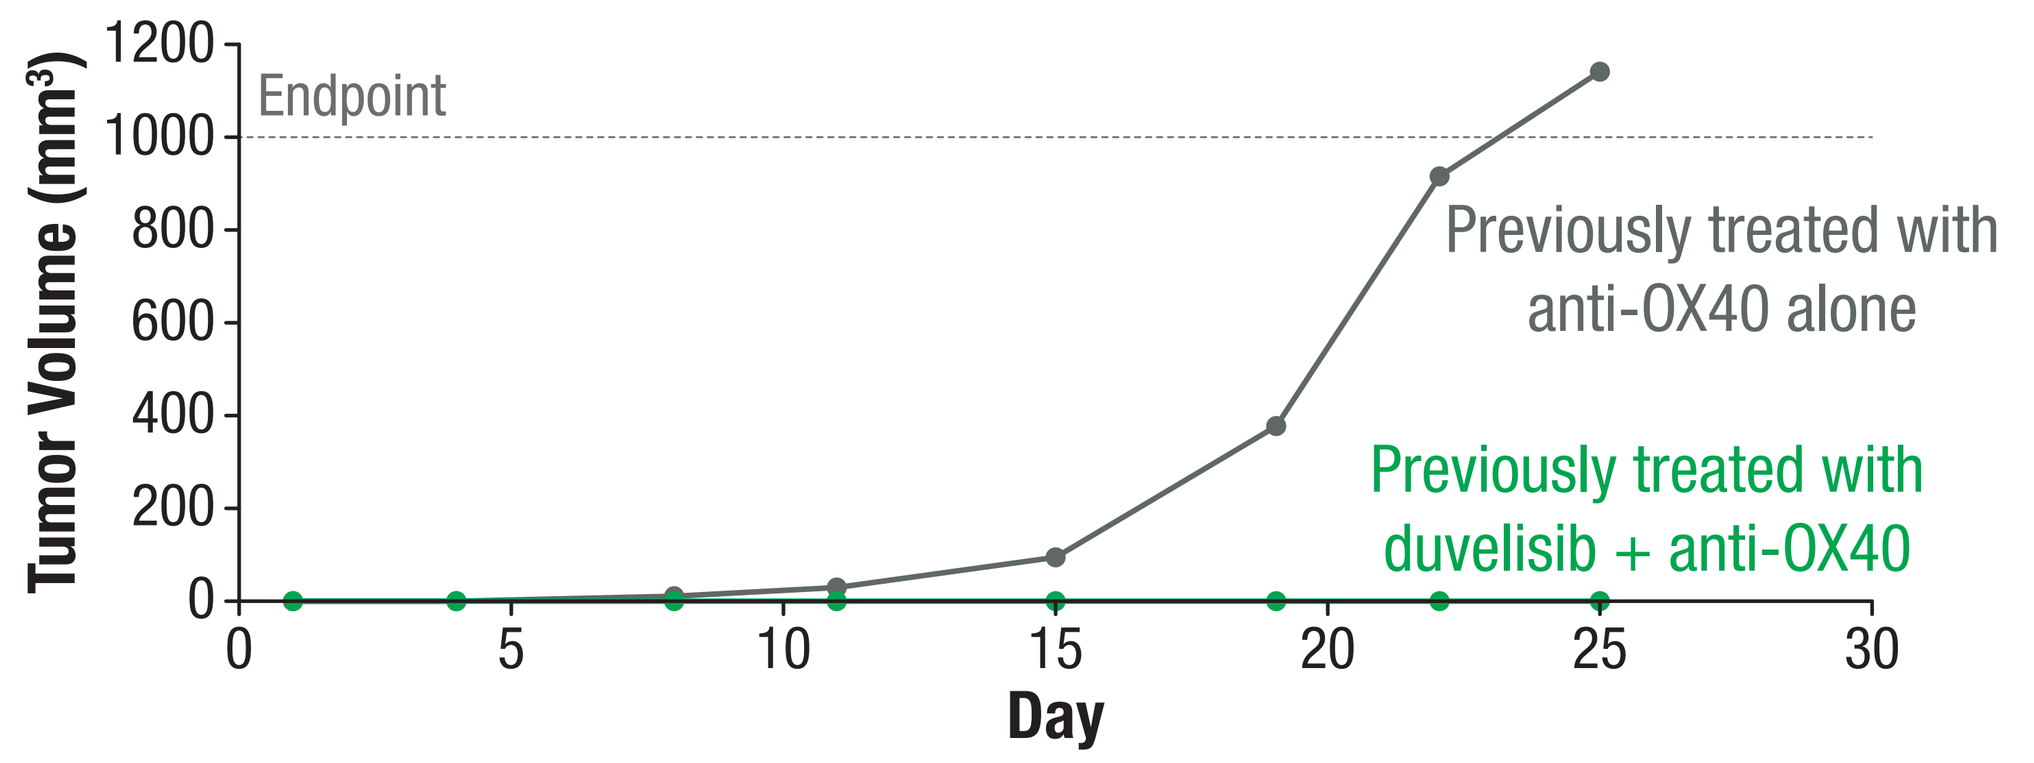
Tumor Volume (mm³)
Endpoint
Previously treated with anti-OX40 alone
Previously treated with duvelisib + anti-OX40
Day

Figure 5: Mice bearing A20 tumors were treated with anti-OX40 alone or duvelisib + anti-OX40. On day 44, all mice with no detectable tumor from the anti-OX40 (n=2) and duvelisib + anti-OX40 (n=5) groups were re-injected with A20 B cell lymphoma cells in the contralateral flank with no further treatment to assess immune memory. Both mice previously treated with anti-OX40 alone grew new tumors, whereas all mice previously treated with duvelisib + anti-OX40 remained tumor-free.

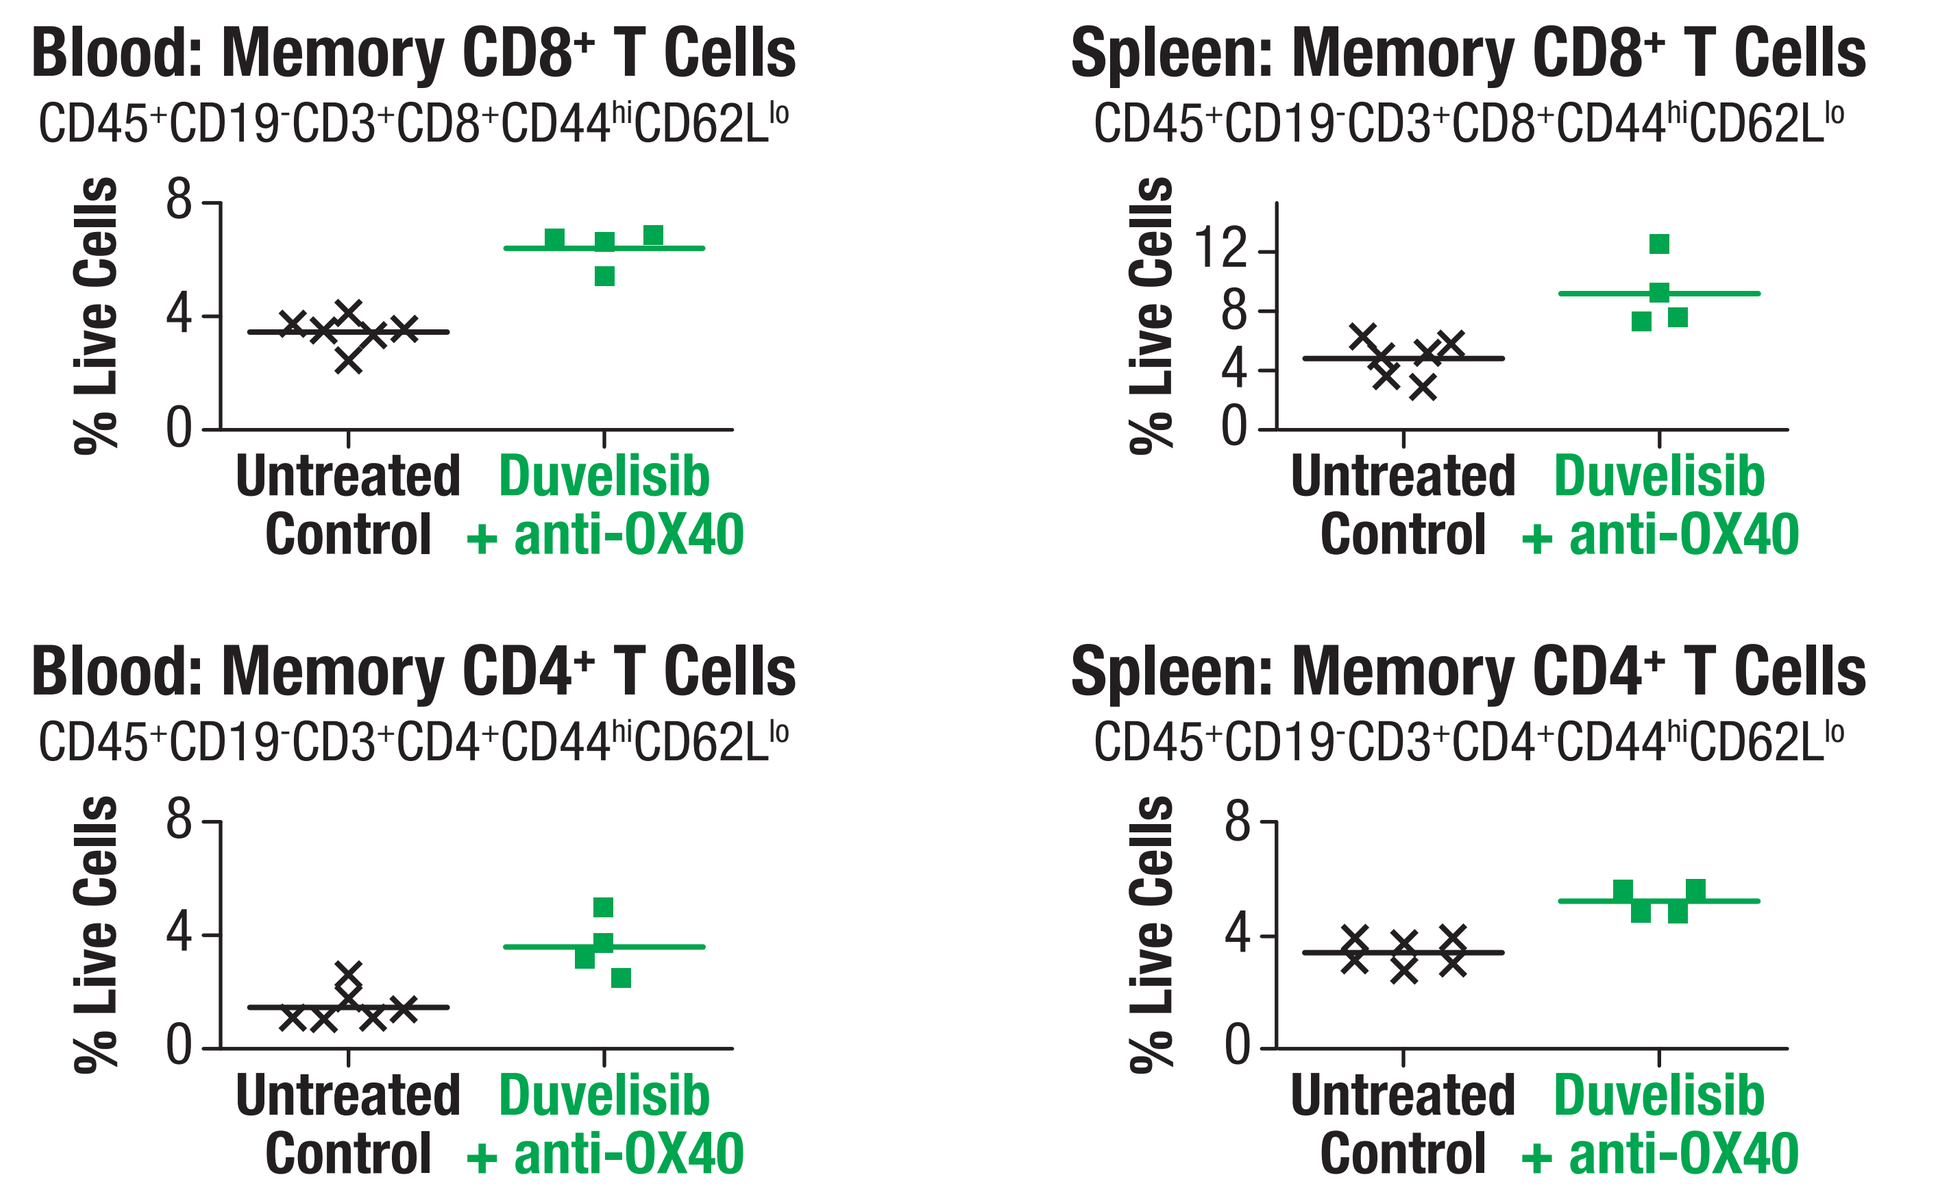
Blood: Memory CD8⁺ T Cells
CD45⁺CD19⁻CD3⁺CD8⁺CD44ʰⁱCD62Lˡᵒ
Spleen: Memory CD8⁺ T Cells
CD45⁺CD19⁻CD3⁺CD8⁺CD44ʰⁱCD62Lˡᵒ
Blood: Memory CD4⁺ T Cells
CD45⁺CD19⁻CD3⁺CD4⁺CD44ʰⁱCD62Lˡᵒ
Spleen: Memory CD4⁺ T Cells
CD45⁺CD19⁻CD3⁺CD4⁺CD44ʰⁱCD62Lˡᵒ
% Live Cells
Untreated Control
Duvelisib + anti-OX40

Figure 6: Mice previously treated with the duvelisib + anti-OX40 combination show increased memory T cells in the blood and spleen compared to untreated control mice bearing A20 tumors.

The Dual PI3K-δ/PI3K-γ Inhibitor Duvelisib Suppresses Both Immunosuppressive Tregs and Immunosuppressive Myeloid Cells

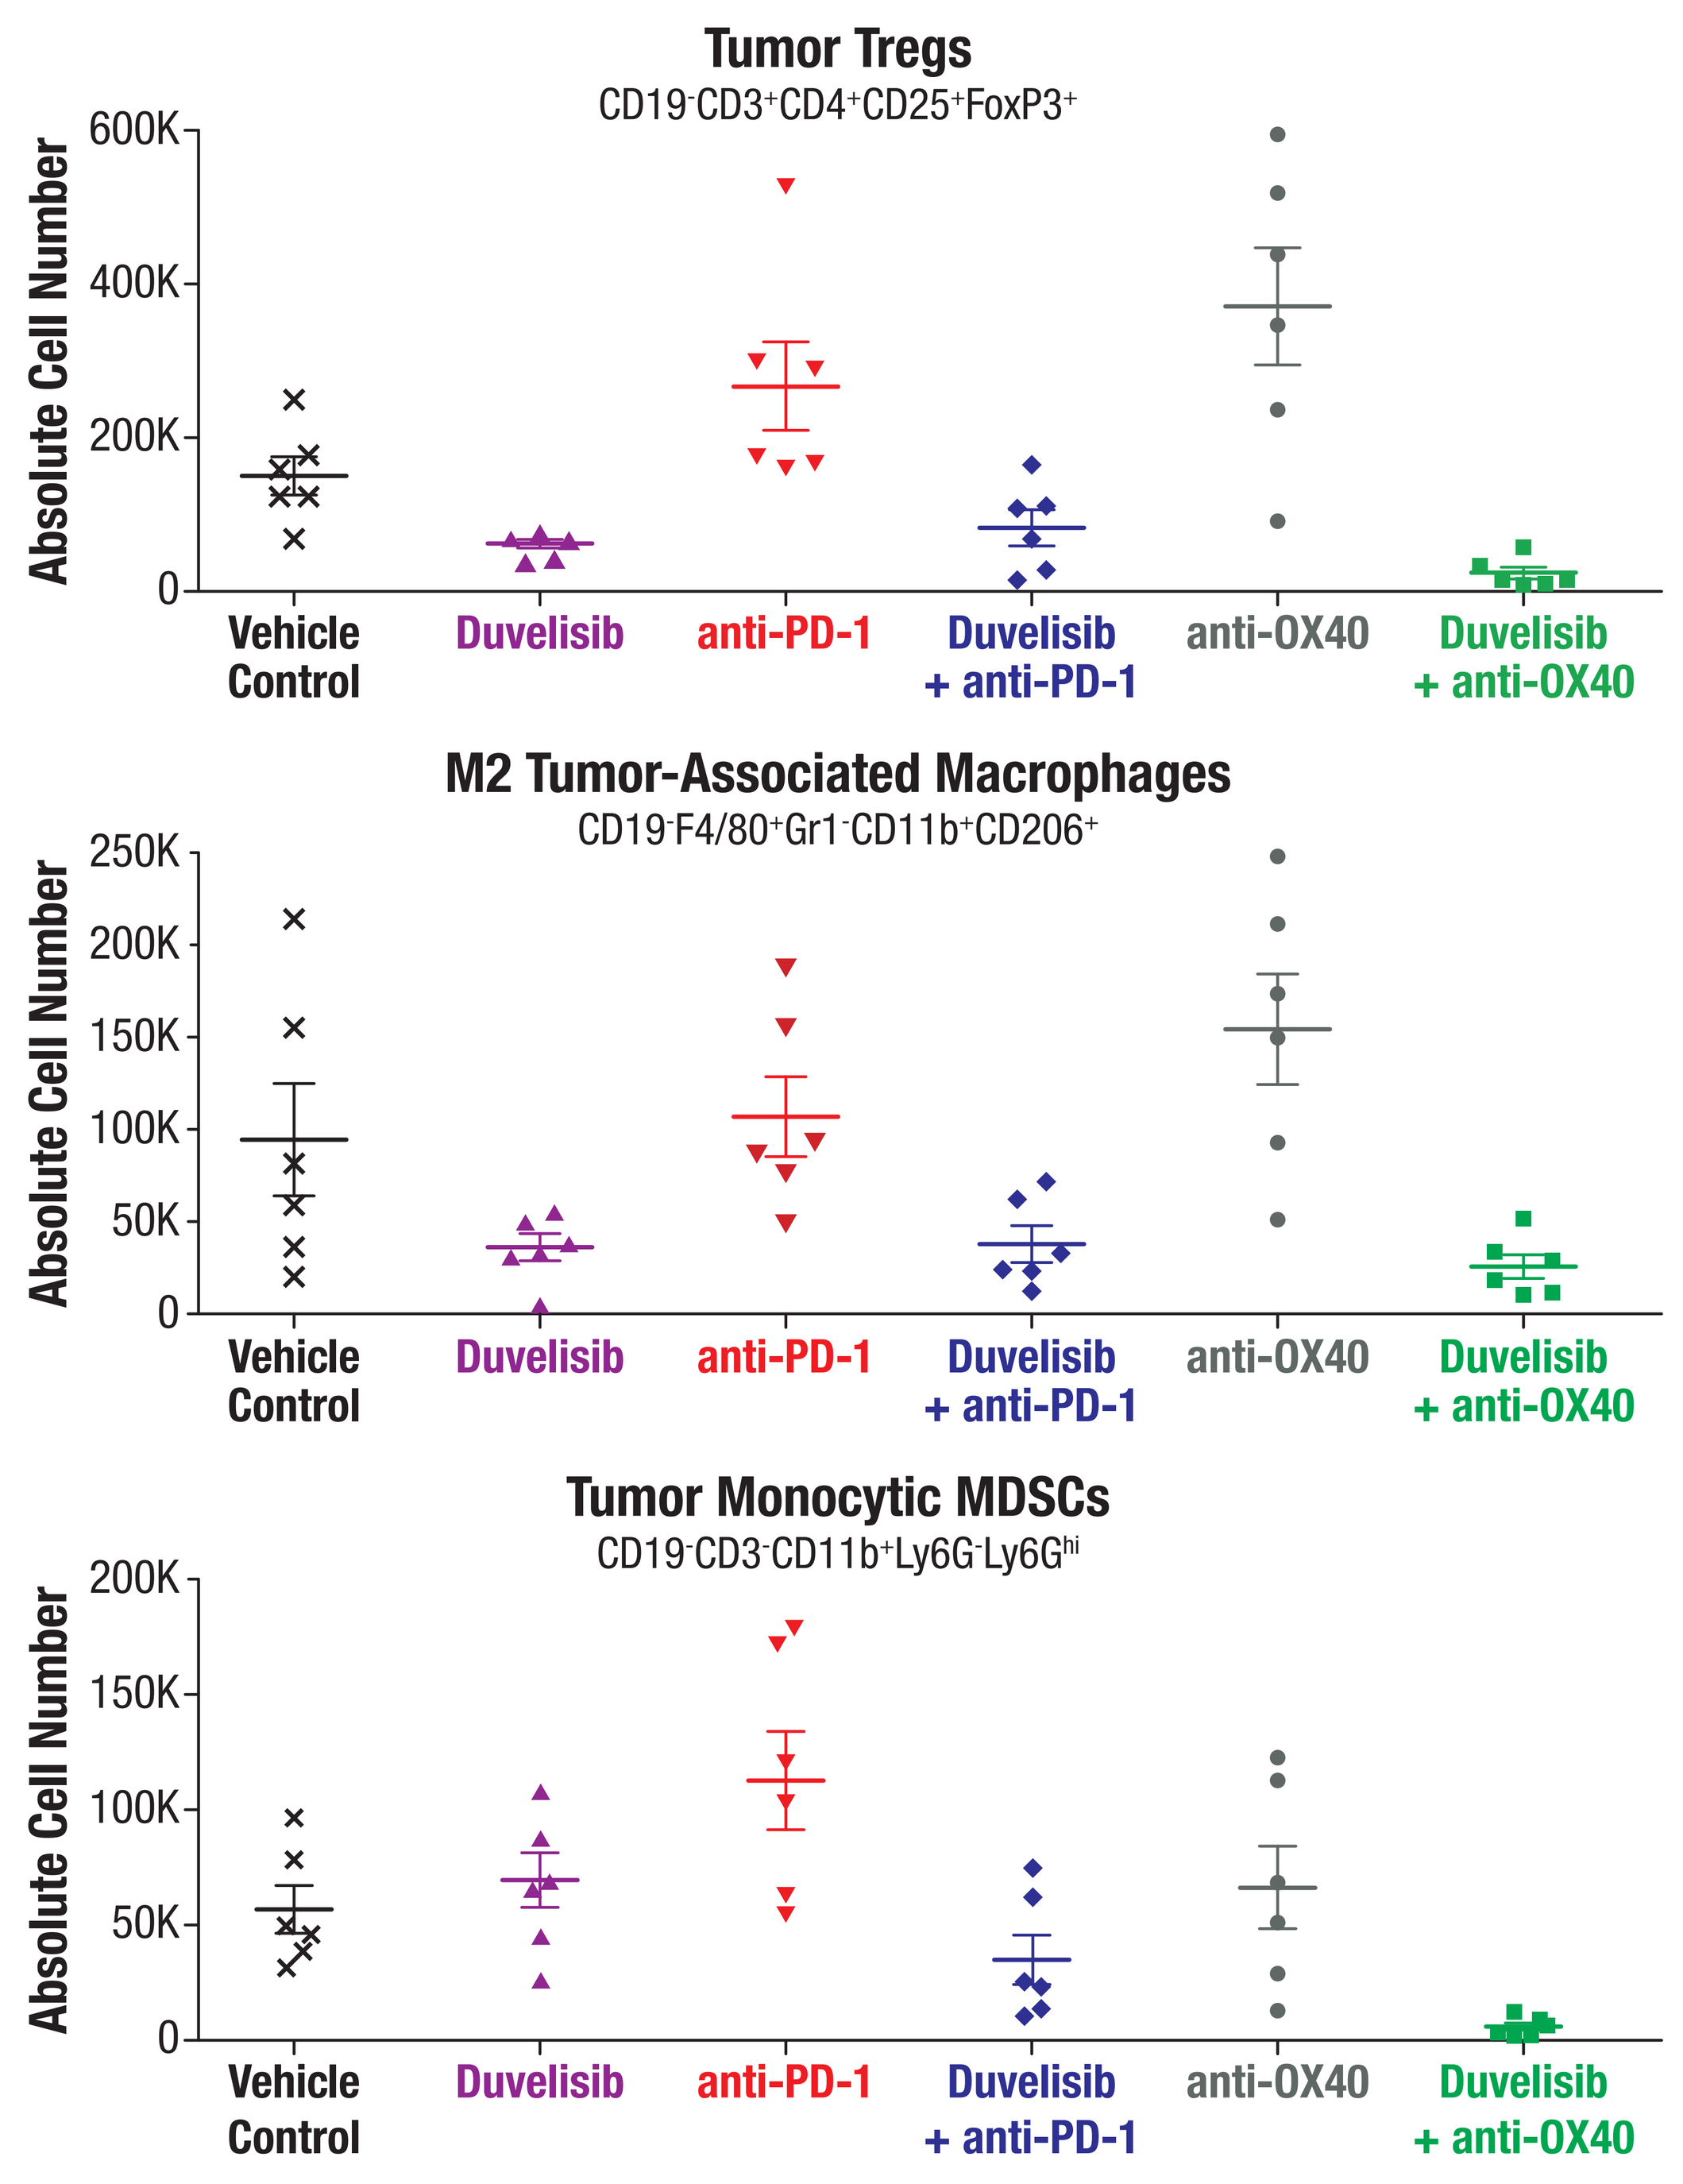
Tumor Tregs
CD19⁻CD3⁺CD4⁺CD25⁺FoxP3⁺
M2 Tumor-Associated Macrophages
CD19⁻F4/80⁺Gr1⁻CD11b⁺CD206⁺
Tumor Monocytic MDSCs
CD19⁻CD3⁻CD11b⁺Ly6G⁻Ly6Gʰⁱ
Absolute Cell Number
Vehicle Control
Duvelisib
anti-PD-1
Duvelisib + anti-PD-1
anti-OX40
Duvelisib + anti-OX40

Figure 7: Effects of duvelisib ± anti-PD-1 or anti-OX40 on Tregs and myeloid immunosuppressive cells in A20 B cell lymphoma tumors. Female BALB/c mice bearing syngeneic A20 B cell lymphoma tumors were randomized once tumors reached 60-90 mm^3 and treated as indicated. On day 9 of treatment, mice were euthanized, tumors removed, and immune cell populations in the tumors were analyzed by flow cytometry using the indicated marker sets.

Summary

- Duvelisib is a dual PI3K-δ,γ inhibitor
- Duvelisib has proven activity as monotherapy in patients with B cell malignancies
- Dual inhibition of PI3K-δ & PI3K-γ confers reduction of both immunosuppressive Tregs (PI3K-δ) & myeloid cells (PI3K-γ)
- Duvelisib greatly enhanced efficacy of anti-PD-1 mAb and induced tumor regression and immune memory in combination with anti-OX40 mAb in an A20 B cell lymphoma model
- These data support further exploration of duvelisib in combination with anti-PD-1/PD-L1 or co-stimulatory antibodies in patients with B cell malignancies

References: 1) O'Brien ASH 2014; Flinn ASH 2014; Horwitz ASH 2014; Flinn ASH 2016; Flinn ASH 2017; **2)** Ali, *Nature*, 2014; **3)** Kaneda, *Nature*, 2016; De Henau, *Nature*, 2016

Presented at the ASCO-SITC Clinical Immuno-Oncology Symposium • January 26, 2018 • San Francisco, California, USA