PI3K/mTOR Inhibitor VS-5584 Targets Cancer Stem Cells and Prevents Tumor Regrowth After Chemotherapy in Preclinical Models of Small Cell Lung Cancer

Abstract# 439

Vihren N. Kolev[1], Mahesh V. Padval[1], Quentin G. Wright[1], Jill Ricono[2], David T. Weaver[1], Jonathan A. Pachter[1], and Qunli Xu[1]

[1]Verastem Inc., Boston, MA; [2]Molecular Response, San Diego, CA

ABSTRACT

Small cell lung cancer (SCLC) is a highly aggressive malignancy with a 5-year overall survival rate of only 5-10%. Most patients with SCLC initially respond to chemotherapy but subsequently experience aggressive tumor recurrence, that may be attributed to the presence of a sub-population of tumor initiating cancer stem cells (CSCs). We have previously demonstrated that the oral dual PI3K/mTOR inhibitor VS-5584, currently in phase 1 clinical development, preferentially targets CSCs in breast and ovarian cancer preclinical models.

Here we report that VS-5584 inhibits proliferation and induces apoptosis in SCLC cell lines *in vitro* and exhibits antitumor activity in SCLC xenograft models *in vivo*. VS-5584 is also synergistic with standard of care chemotherapy cisplatin and etoposide in reducing the viability of NCI-H69 SCLC cells. Importantly, VS-5584 is more potent against CSCs than bulk tumor cells and thus reduces the proportion of CSCs in preclinical models of SCLC. A drug resistant population of small cell lung cancer CSCs (side population) can be monitored by enhanced ability to exclude Hoechst 33342 dye. In the H69 SCLC cell line, VS-5584 effectively eliminated the side population CSCs, while cisplatin and etoposide enriched this CSC population. *In vivo*, VS-5584 caused significant tumor growth inhibition and reduced the proportion of CSCs in NCI-H841 SCLC tumors as evidenced by a significant decrease in the percentage of side population cells and approximately 70-fold reduction in tumor-initiating capability when tumor cells from VS-5584-treated animals were injected into secondary immunodeficient mice. Consistent with the notion that CSCs are responsible for cancer relapse after chemotherapy, VS-5584 significantly reduced tumor regrowth following cessation of cisplatin treatment in the H69 SCLC xenograft model and in a SCLC patient-derived xenograft (PDX) model.

The preferential targeting of CSCs by VS-5584 in preclinical models of SCLC provides the rationale for clinical development of VS-5584 either as a single agent or in combination with chemotherapeutic agents to potentially extend time to relapse and improve outcome for patients with small cell lung cancer.

INTRODUCTION

Structure & selectivity profile of VS-5584

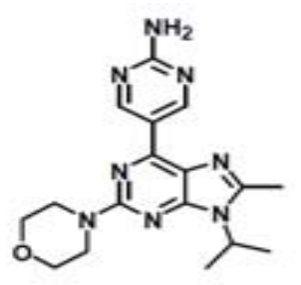

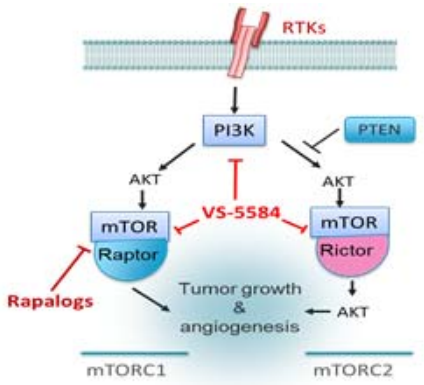
RTKs
PI3K
PTEN
AKT
AKT
VS-5584
mTOR
Raptor
mTOR
Rictor
Rapalogs
Tumor growth & angiogenesis
AKT
mTORC1
mTORC2

Biochemical Potencies (IC_{50}, nM)*					
mTOR	PI3Kα	PI3Kαmut H1047R	PI3Kβ	PI3Kγ	PI3Kδ
3.4	2.6	3.3	21	2.7	3

*Selective for these kinases among a panel of over 400 kinases (Hart et al., Mol Cancer Ther. 2013 Feb;12(2):151-61.)

• We have previously reported that dual PI3K/mTOR inhibition is critical for preferential targeting of CSCs: *Kolev et al, 2014 AACR Abstract #3906*

RESULTS

Fig 1: A subset of SCLC cell lines harboring PIK3CA^{mut} or PTEN-loss exhibit increased sensitivity to VS-5584.

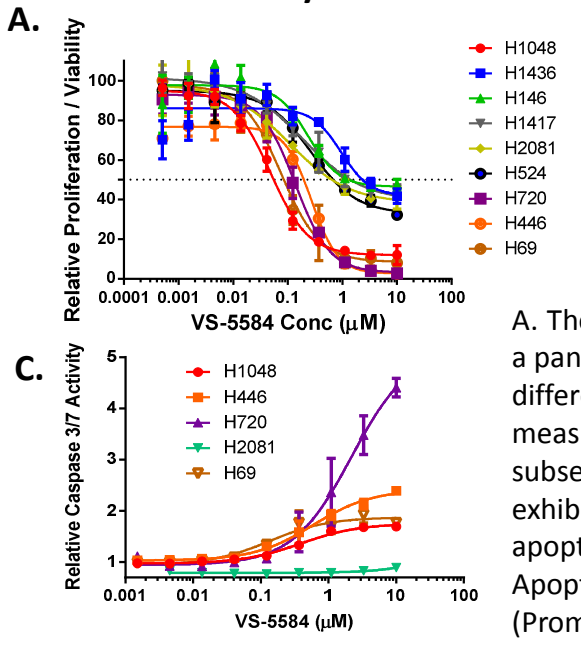
A.
Relative Proliferation / Viability
VS-5584 Conc (μM)
H1048
H1436
H146
H1417
H2081
H524
H720
H446
H69
C.
Relative Caspase 3/7 Activity
VS-5584 (μM)
H1048
H446
H720
H2081
H69

B.

	PIK3CA	PTEN	TP53	RB
H1048	H1047R		Mut	Mut
H446		Mut	Mut	Mut
H69	G106_R108del		Mut	Mut
H1436		Mut	Mut	Mut
H2081		Mut		Mut
H1417			Mut	Mut
H524			Mut	Mut
H146			Mut	Mut
H720			Mut	Mut

A. The anti-proliferative effect of VS-5584 was evaluated in a panel of 9 SCLC cell lines. A. Cells were treated VS-5584 at different concentrations for 3 days and cell viability was measured with CellTiter Glo (Promega). Interestingly a subset of cell lines with PIK3CA^{mut} or PTEN-loss (B., arrows) exhibited hypersensitivity to VS-5584. C. VS-5584 induces apoptosis in cell lines bearing PIK3CA^{mut} or PTEN-loss. Apoptosis was measured by CaspaseGlo 3/7 assay (Promega).

Fig 2: VS-5584 exhibits synergistic anticancer activity with standard-of-care agents cisplatin and etoposide in SCLC cell lines *in vitro*.

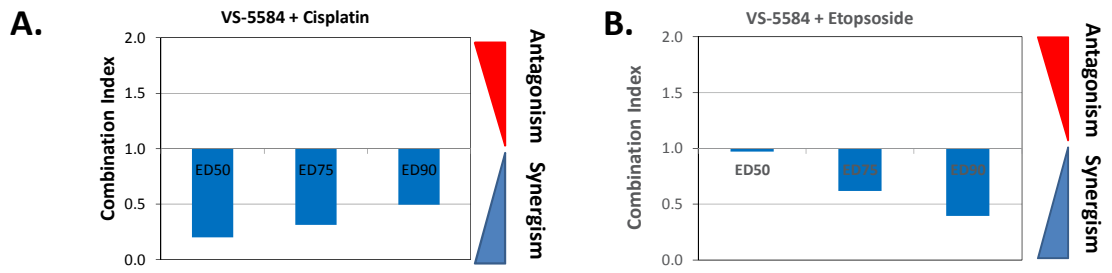
A.
VS-5584 + Cisplatin
Combination Index
ED50
ED75
ED90
Antagonism
Synergism
B.
VS-5584 + Etopsoside
Combination Index
ED50
Antagonism
Synergism

SCLC cell line NCI-H69 was treated with combination of VS-5584 and cisplatin (A) or etoposide (B) in 10x10 concentration matrix and cell viability was determined by CellTiter Glo (Promega). Combination index analysis using CalcuSyn (Biosoft, Cambridge, UK) at effective dose (ED) 50, 75, or 90 for both agents are shown.

Fig 3: VS-5584 preferentially targets SCLC CSCs.

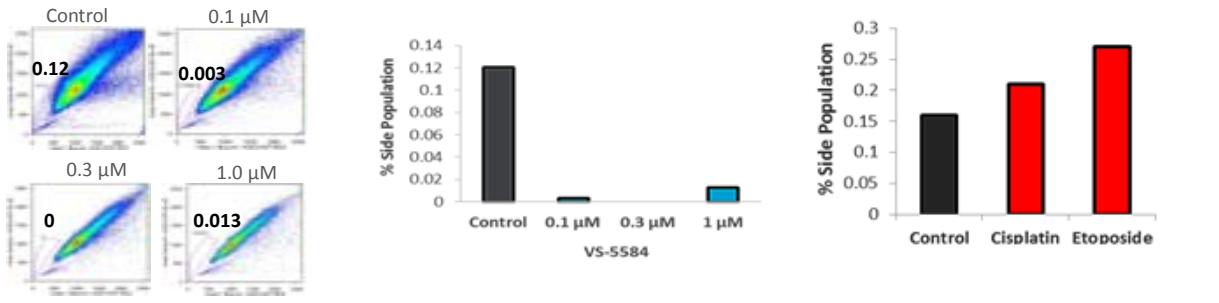
Control
0.12
0.1 μM
0.003
0.3 μM
0
1.0 μM
0.013
% Side Population
Control
0.1 μM
0.3 μM
1 μM
VS-5584
% Side Population
Control
Cisplatin
Etoposide

H69 cells were cultured under hypoxic conditions and treated with VS-5584, cisplatin or etoposide for 2 days. Cells were incubated with Hoechst 33342 dye and SP was determined by FACS.

Verastem, Inc. | 117 Kendrick St| Suite 500| Needham| Massachusetts | 02494

Fig 4: VS-5584 preferentially targets CSCs *in vivo* and delays tumor regrowth after chemotherapy in small cell lung cancer models.

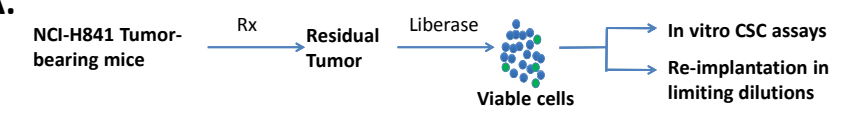
A.
NCI-H841 Tumor-bearing mice
Rx
Residual Tumor
Liberase
Viable cells
In vitro CSC assays
Re-implantation in limiting dilutions

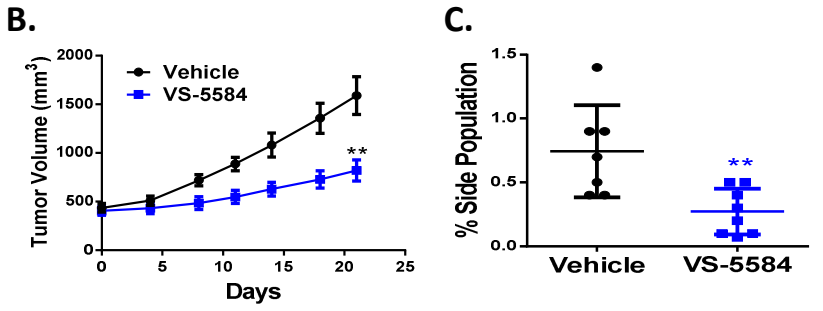
B.
Tumor Volume (mm3)
Vehicle
VS-5584
Days
C.
% Side Population
Vehicle
VS-5584

D.

Limiting dilutions

# Cells	Control	VS-5584
1,000,000	4(4)	2(4)
100,000	2(3)	0(3)
10,000	2(3)	0(3)
1,000	1(3)	0(3)
TIF	1/24,000	1/1,615,000
P		$5x10^{-6}$

VS-5584 reduced TIF by 67 fold.

Mice bearing NCI-H841 SCLC tumors were treated with either vehicle or 20 mg/kg VS-5584 thrice weekly for 3 weeks. A. Experimental outline. B. Tumor volume was measured. C, D. Cells were dissociated from tumors and subject to side population analysis by FACS (C) or implanted in secondary mice in limiting dilution assay (D).

Fig 5: VS-5584 delays tumor regrowth after chemotherapy in small cell lung cancer models.

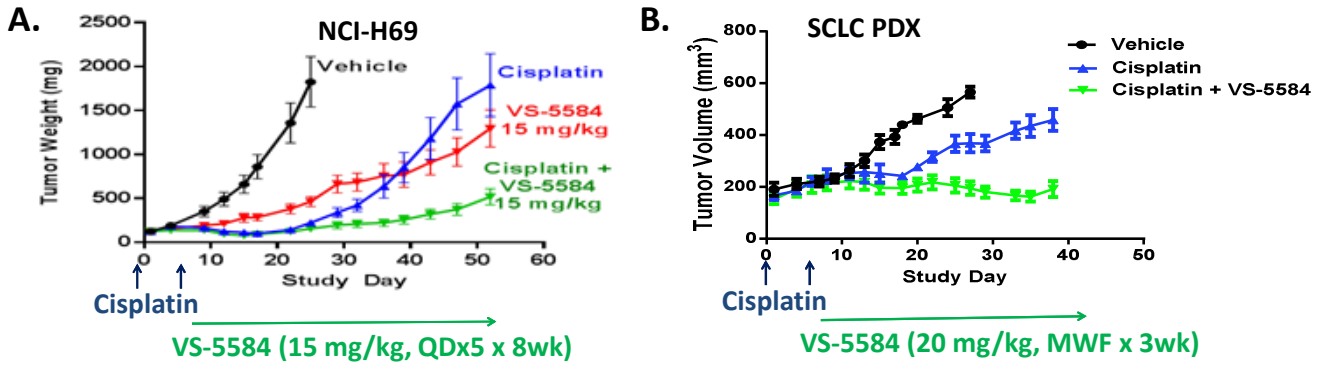
A.
NCI-H69
Tumor Weight (mg)
Vehicle
Cisplatin
VS-5584 15 mg/kg
Cisplatin + VS-5584 15 mg/kg
Study Day
Cisplatin
VS-5584 (15 mg/kg, QDx5 x 8wk)
B.
SCLC PDX
Tumor Volume (mm3)
Vehicle
Cisplatin
Cisplatin + VS-5584
Study Day
Cisplatin
VS-5584 (20 mg/kg, MWF x 3wk)

A. Mice bearing NCI-H69 xenografts were treated with vehicle, cisplatin (weekly dosing of 5 mg/kg cisplatin for 2 weeks), VS-5584 (oral gavage at 15 mg/kg on a QDx5 schedule for 8 weeks), alone or in combination. B. Mice bearing SCLC PDX tumors were treated with vehicle, cisplatin (weekly dosing of 5 mg/kg cisplatin for 2 weeks) or cisplatin followed by VS-5584 (oral gavage at 20 mg/kg on MWF dosing schedule for 3 weeks).

SUMMARY

- **VS-5584 is a highly selective dual PI3K/mTOR inhibitor that potently inhibits mTORC1, mTORC2 and all Class I PI3K isoforms.**
- **VS-5584 exhibits synergistic activity with standard-of-care agents cisplatin and etoposide in SCLC.**
- **VS-5584 preferentially inhibits cancer stem cells as assessed in orthogonal assays *in vitro and in vivo.***
- **Consistent with its preferential targeting of CSCs, VS-5584 delays tumor regrowth after treatment with the cytotoxic agent cisplatin in SCLC xenograft models.**
- **VS-5584 is currently being evaluated in a Phase 1 clinical trial in patients with solid tumors or lymphoma NCT01991938.**