The Effect of Duvelisib, a Dual Inhibitor of PI3K-δ,γ on the Components of the Tumor Microenvironment in Previously Untreated Follicular Lymphoma Patients

Poster 216
Abstract 7579

Carla Casulo[1], Andre Goy[2], Koen Van Eygen[3], Juan M. Sancho[4], Santiago Mercadal[5], Rod Johnson[6], Sven DeVos[7], Kamal-Krimo Bouabdallah[8], Kam Sprott[9], NgocDiep Le[9], Jonathan A. Pachter[9], David T. Weaver[9]

[1]Wilmot Cancer Institute University of Rochester Medical School, Rochester, New York, USA; [2]Hackensack University Medical Center, Hackensack, New Jersey, USA; [3]AZ GROENINGE, Kortrijk, Belgium; [4]Hospital Universitario Germans Trias i Pujol, Badalona, Spain; [5]Department of Hematology, ICO L'Hospitalet-Hospital Duran i Reynals L'Hospitalet de Ll., Spain; [6]St. James's Univ. Hospital Trust, Leeds, Great Britain; [7]Division of Hematology/Oncology, Ronald Reagan UCLA Medical Center, Los Angeles, California, USA; [8]University Hospital of Bordeaux, CHU Haut-Leveque, Bordeaux, France; [9]Verastem Oncology, Needham, Massachusetts, USA

Background

- PI3K-δ inhibition directly targets malignant cells, while PI3K-γ inhibition disrupts the supportive tumor microenvironment
- Duvelisib, an oral dual inhibitor of PI3K-δ and PI3K-γ, has shown clinical activity in iNHL (including FL), CLL/SLL, and TCL[1-3]
- Duvelisib has shown an acceptable safety profile in Phase 1-3 studies in patients with advanced hematologic malignancies[1-3]
- The CONTEMPO study is a two-arm, open-label, Phase 1b/2 study that examined the safety and efficacy of duvelisib in combination with either rituximab or obinutuzumab in previously untreated CD20+ FL

Figure 1: ORR by Investigator assessment per IWG Criteria in the Phase 2 CONTEMPO™ study[2]

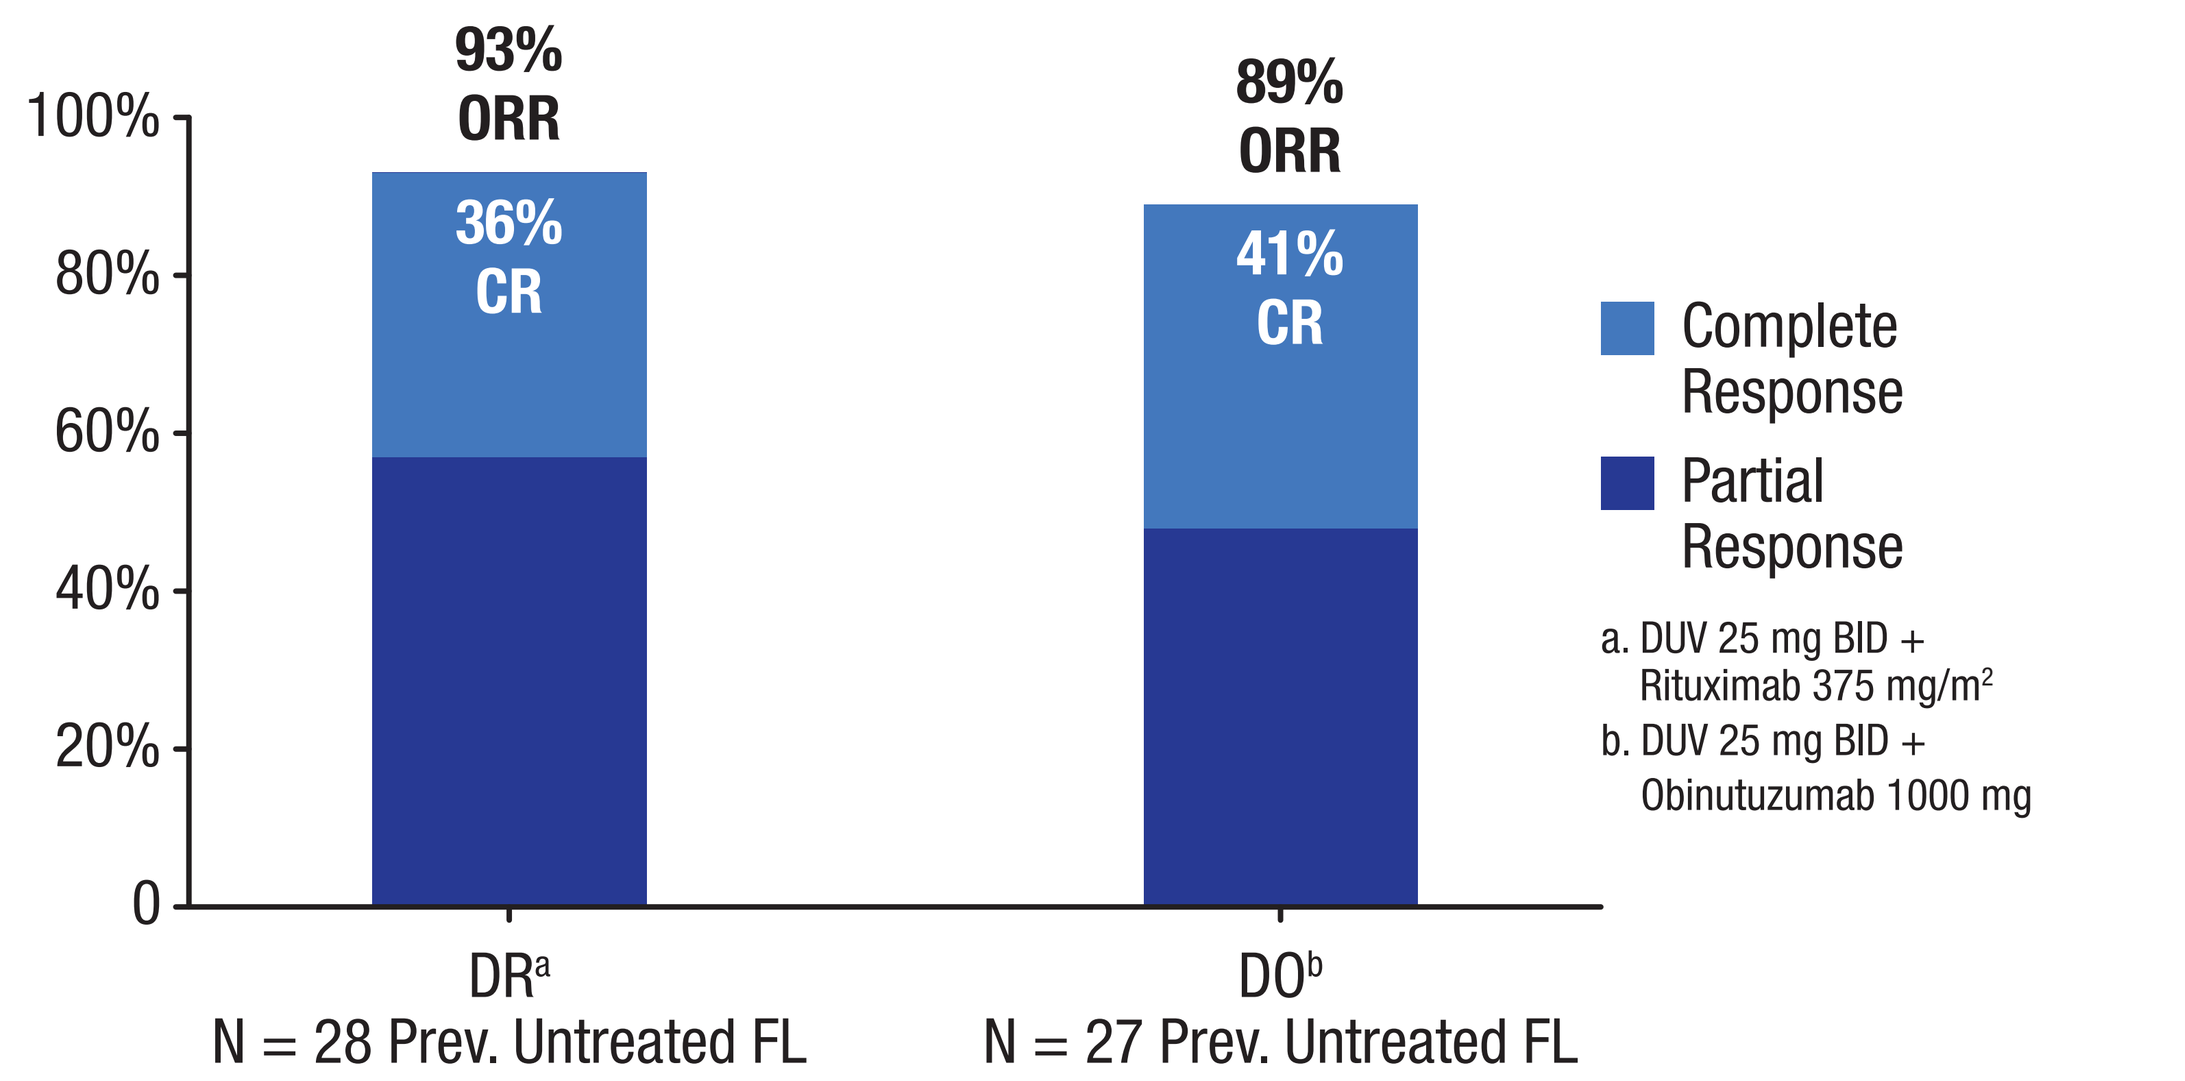
93% ORR
36% CR
89% ORR
41% CR
100%
80%
60%
40%
20%
0
Complete Response
Partial Response
a. DUV 25 mg BID + Rituximab 375 mg/m²
b. DUV 25 mg BID + Obinutuzumab 1000 mg
DR[a]
N = 28 Prev. Untreated FL
DO[b]
N = 27 Prev. Untreated FL

Figure 2: ORR with duvelisib monotherapy in Phase 1 and Phase 2 studies

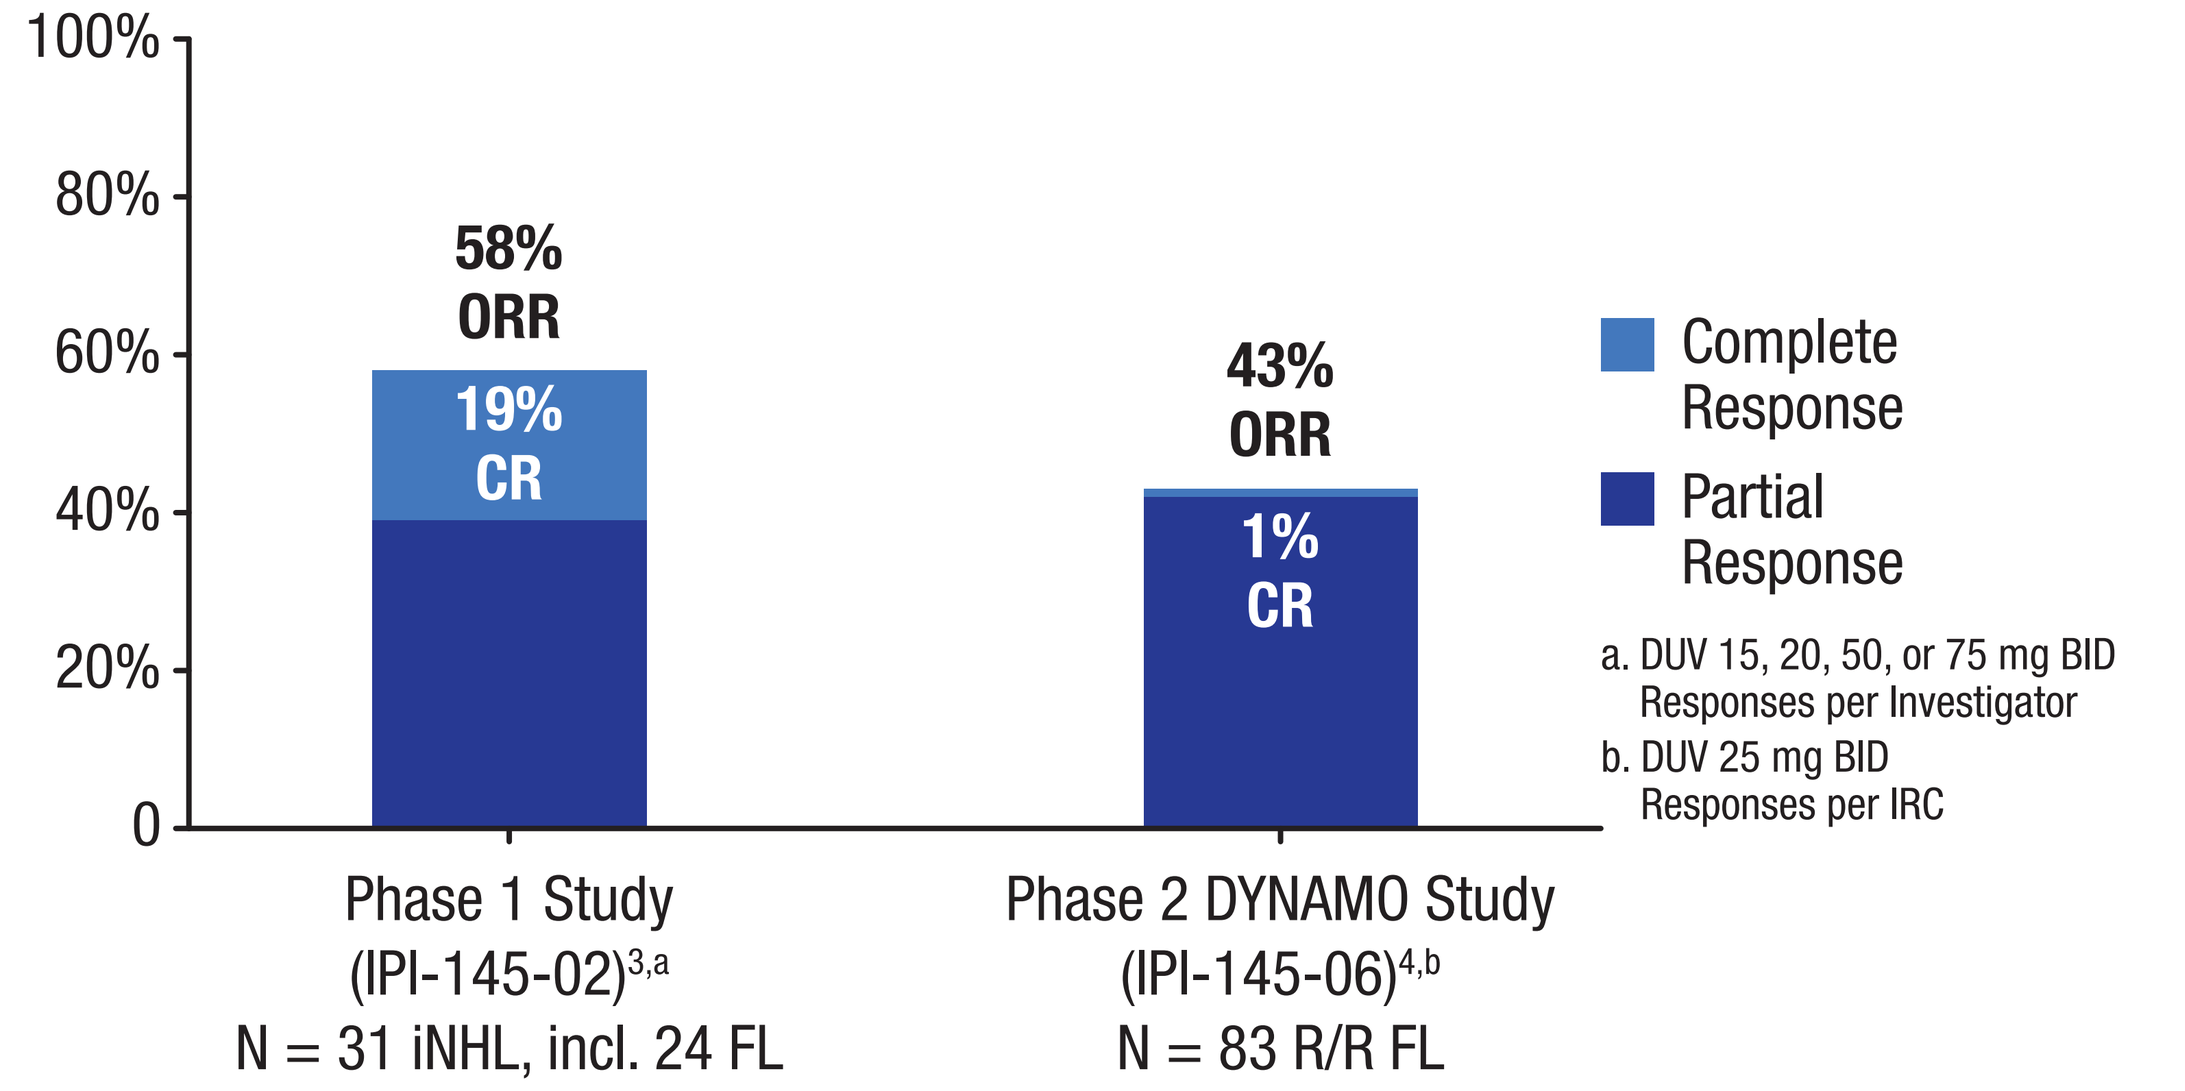
58% ORR
19% CR
43% ORR
1% CR
100%
80%
60%
40%
20%
0
Complete Response
Partial Response
a. DUV 15, 20, 50, or 75 mg BID Responses per Investigator
b. DUV 25 mg BID Responses per IRC
Phase 1 Study (IPI-145-02)[3,a]
N = 31 iNHL, incl. 24 FL
Phase 2 DYNAMO Study (IPI-145-06)[4,b]
N = 83 R/R FL

PI3K-δ and PI3K-γ-Selective Pharmacodynamic Assessment in Circulating Monocytes§

- Whole blood *ex vivo* assay for PI3K-δ- and PI3K-γ-selective PD
- Flow cytometry-based measurements for CD14+ cells
- Healthy donor samples to assess duvelisib and other agents (marked with ** below)

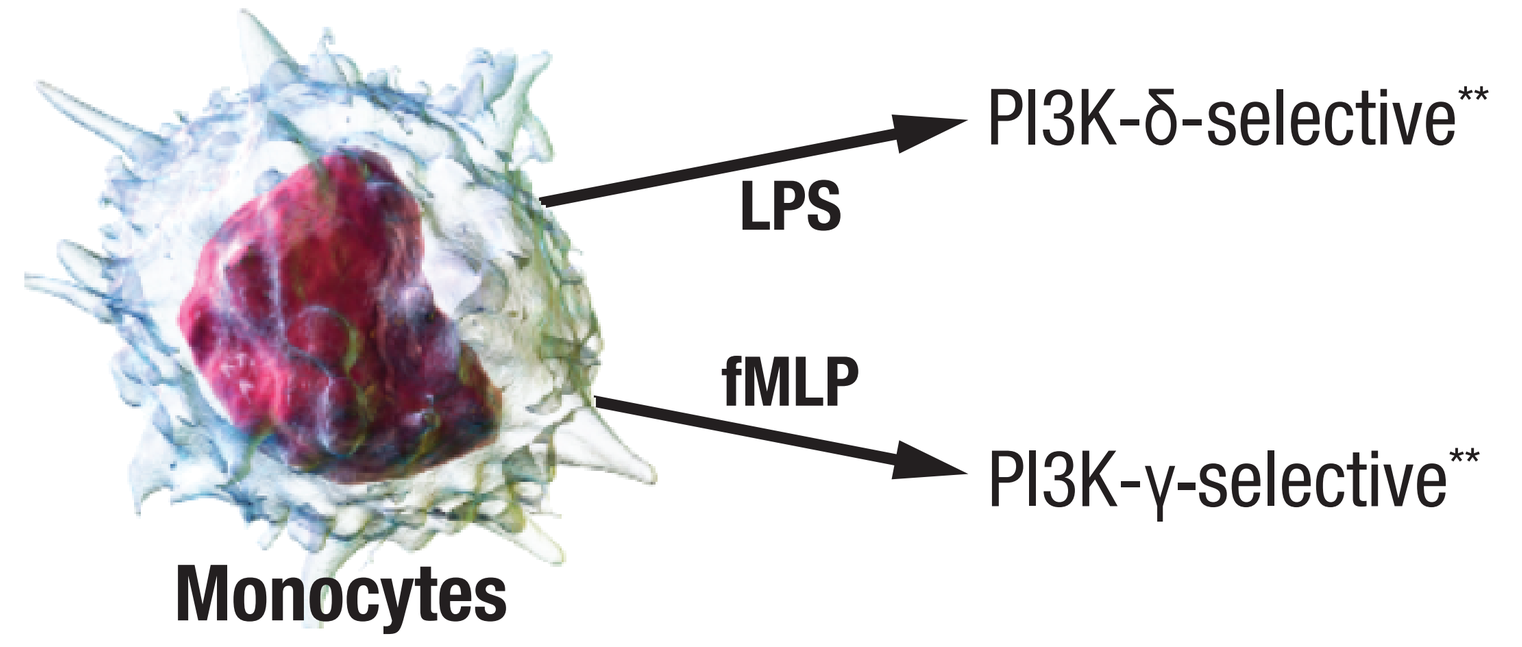
PI3K-δ-selective**
LPS
fMLP
PI3K-γ-selective**
Monocytes

PI3K-δ,γ Inhibition in Selective Whole Blood Assays§

- *Ex vivo* whole blood assays were conducted from 32 FL patients treated on CONTEMPO
- PI3K-δ assay: LPS-stimulated monocyte pAKT (T308)
- PI3K-γ assays: fMLP-stimulated monocyte pAKT (T308)

PI3K-δ Inhibition by Duvelisib 25 mg BID

- pAKT T308 inhibition 95% at 1 hr postdose, 92% at 4 hours postdose, and 70% predose at Cycle 2 Day 1

Figure 3: pAKT T308 Inhibition by Duvelisib in LPS-Stimulated CD14+ Monocytes

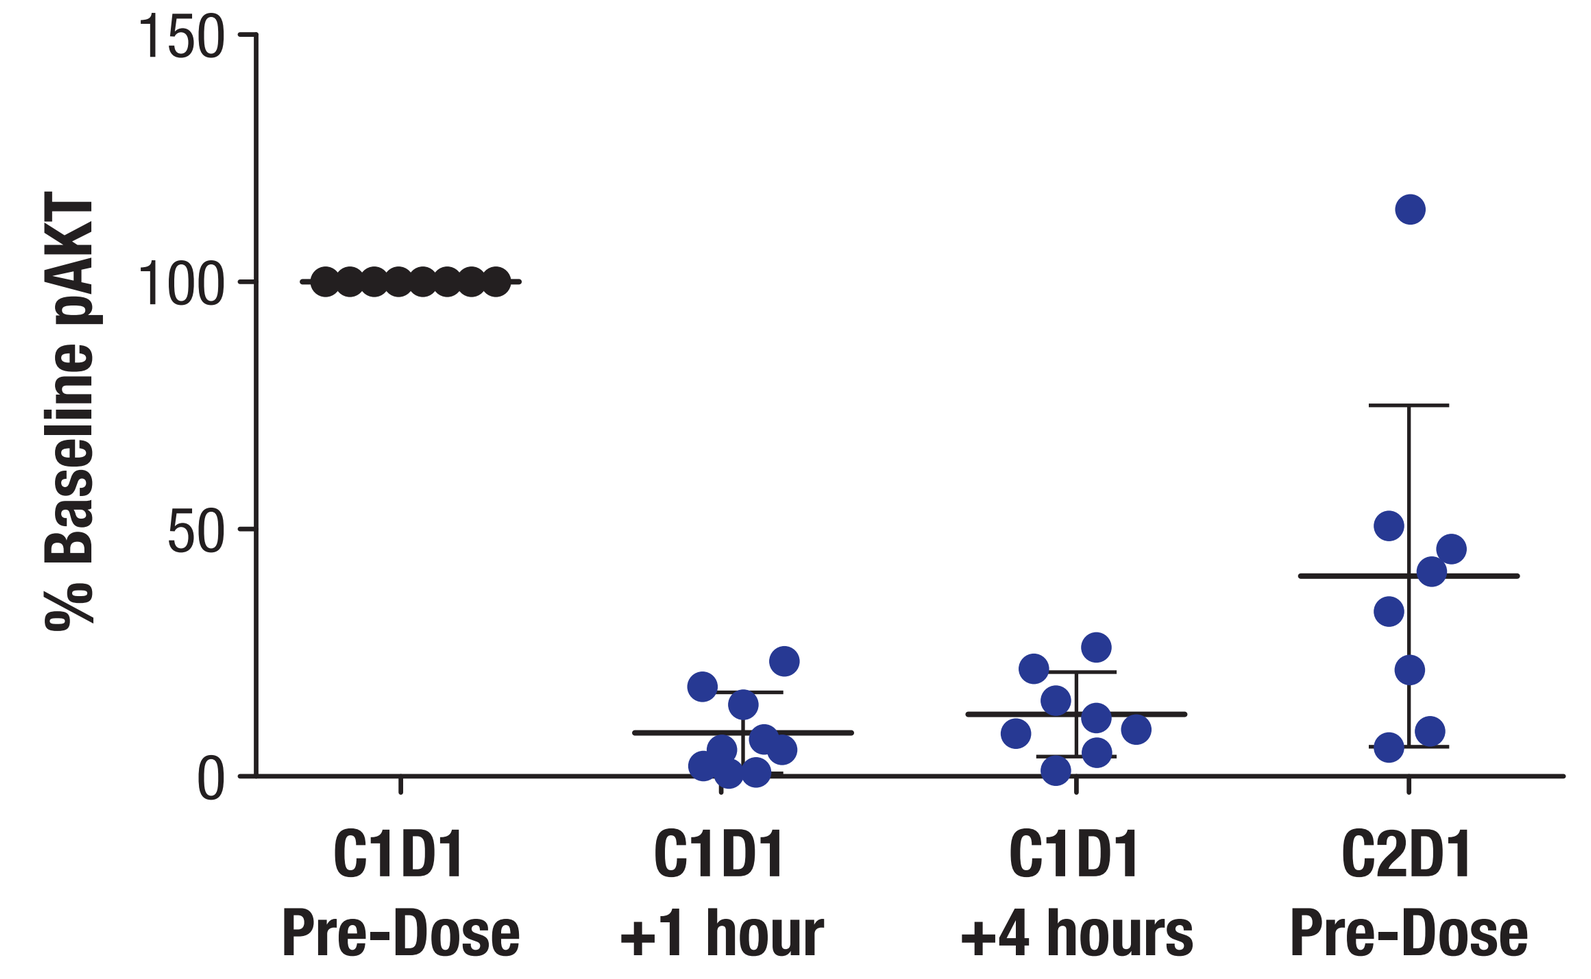
% Baseline pAKT
150
100
50
0
C1D1 Pre-Dose
C1D1 +1 hour
C1D1 +4 hours
C2D1 Pre-Dose

PI3K-γ Inhibition by Duvelisib 25 mg BID

- pAKT T308 inhibition 62% at 1 hour postdose, 69% at 4 hours postdose, and 14% predose at Cycle 2 Day 1

Figure 4: pAKT T308 Inhibition by Duvelisib in fMLP-Stimulated CD14+ Monocytes

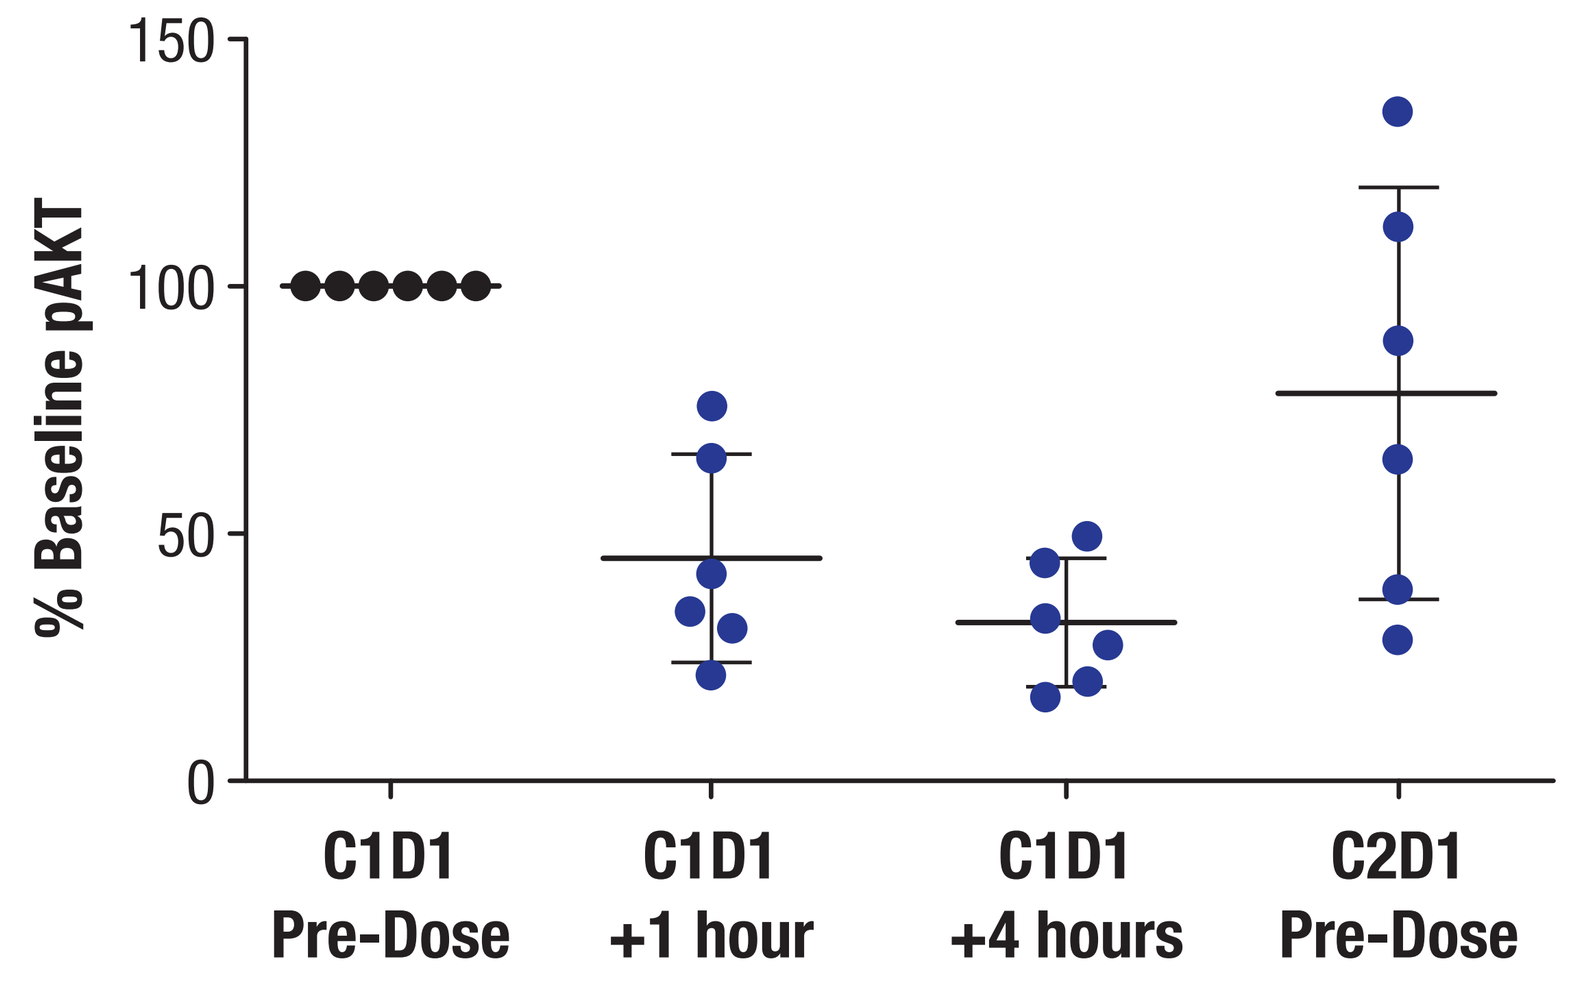
% Baseline pAKT
150
100
50
0
C1D1 Pre-Dose
C1D1 +1 hour
C1D1 +4 hours
C2D1 Pre-Dose

Abbreviations: ANOVA = analysis of variance; ARG1 = arginase-1; BID = twice daily; BMDM = bone marrow-derived macrophage; C1D1 = Cycle 1 Day 1; C2D1 = Cycle 2 Day 1; CD14 = cluster of differentiation 14; CLL = chronic lymphocytic leukemia; C_{max} = maximum concentration observed; CR = complete response; CXCL = CXC chemokine ligand; DO = DUV + ofatumumab; DR = DUV + rituximab; DUV = duvelisib; EC_{50} = half maximal effective concentration; FL = follicular lymphoma; fMLP = formyl-methionyl-leucyl-phenylalanine; IC_{50} = half maximal inhibitory concentration; iNHL = indolent non-Hodgkin's lymphoma; IRC = Independent Review Committee; IWG = International Working Group; LPS = lipopolysaccharide; ORR = objective response rate; pAKT = phospho-AKT; PD = pharmacodynamics; PI3K = phosphoinositide 3-kinase; PK = pharmacokinetics; RP2D = recommended Phase 2 dose; R/R = relapsed/refractory; SLL = small lymphocytic lymphoma; TCL = T cell lymphoma; TME = tumor microenvironment

References: 1) Flinn, et al. ASH 2017. 2) Flinn, et al. 2018. Blood.131(8):877-887. doi: 10.1182/blood-2017-05-786566. 3) Zinzani, et al. EHA 2017. 4) Casulo, et al. ASH 2016 5) Chiu, et al. 2017. Front Immunol. 2017 Jun 30;8:747. doi: 10.3389/fimmu.2017.00747. 6) Hong, et al. SITC 2017. 7) Webb, et al. ASH 2010. 8) Burris, et al. Lancet Oncol. 2018 Apr;19(4):486-496. doi: 10.1016/S1470-2045(18)30082-2.

Duvelisib PI3K-γ Activity Inhibits Macrophage Migration

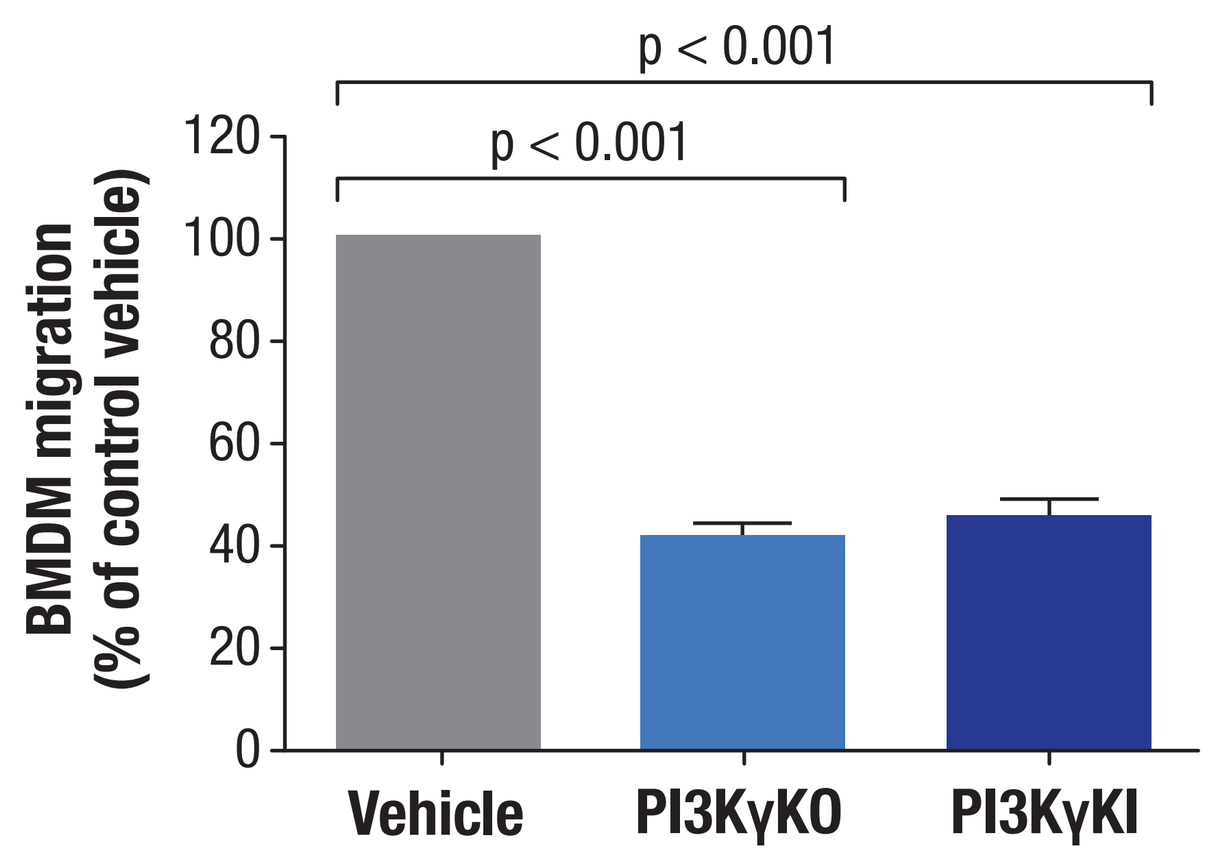
p < 0.001
p < 0.001
BMDM migration (% of control vehicle)
120
100
80
60
40
20
0
Vehicle
PI3KγKO
PI3KγKI

Figure 5: BMDM (murine) migration to C5α or CXCL12. PI3K-γ knockout or PI3K-γ kinase-deficient macrophages significantly reduced for migration. (Analysis Test: One-Way ANOVA Bonferroni post-test)

Figure 6: Duvelisib and IPI-549 (γ-selective) exhibit similar inhibitory activity against macrophage migration by CXCL12 (left) or C5α (right)

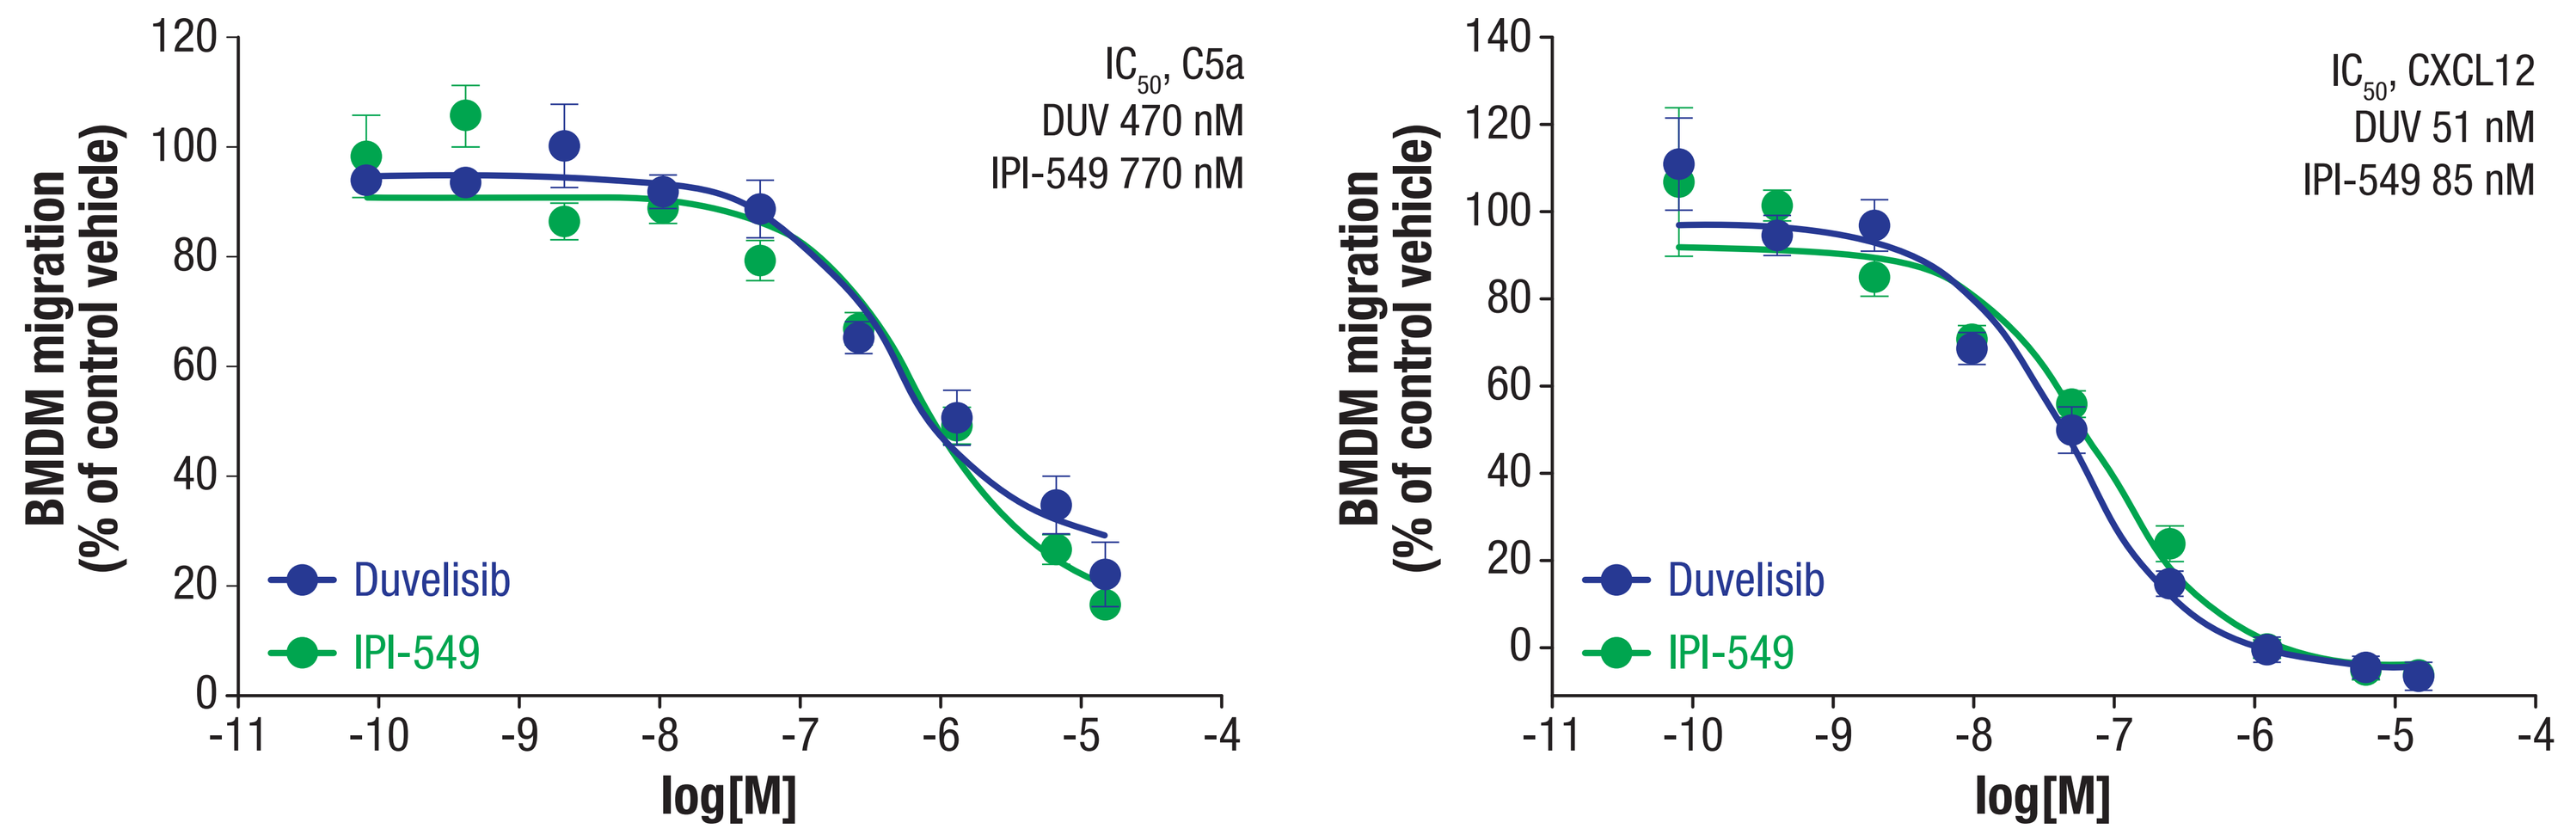
BMDM migration (% of control vehicle)
IC50, C5a
DUV 470 nM
IPI-549 770 nM
Duvelisib
IPI-549
log[M]
IC50, CXCL12
DUV 51 nM
IPI-549 85 nM
Duvelisib
IPI-549
log[M]

Duvelisib Activity Inhibits Macrophage Polarization and Modulates TME

- Macrophage polarization is PI3K-γ-dependent
- Duvelisib and IPI-549 inhibit M2 macrophage polarization

Figure 7: IC_{50} Values for Duvelisib, IPI-549, and IPI-3063 in M2 Macrophage Polarization

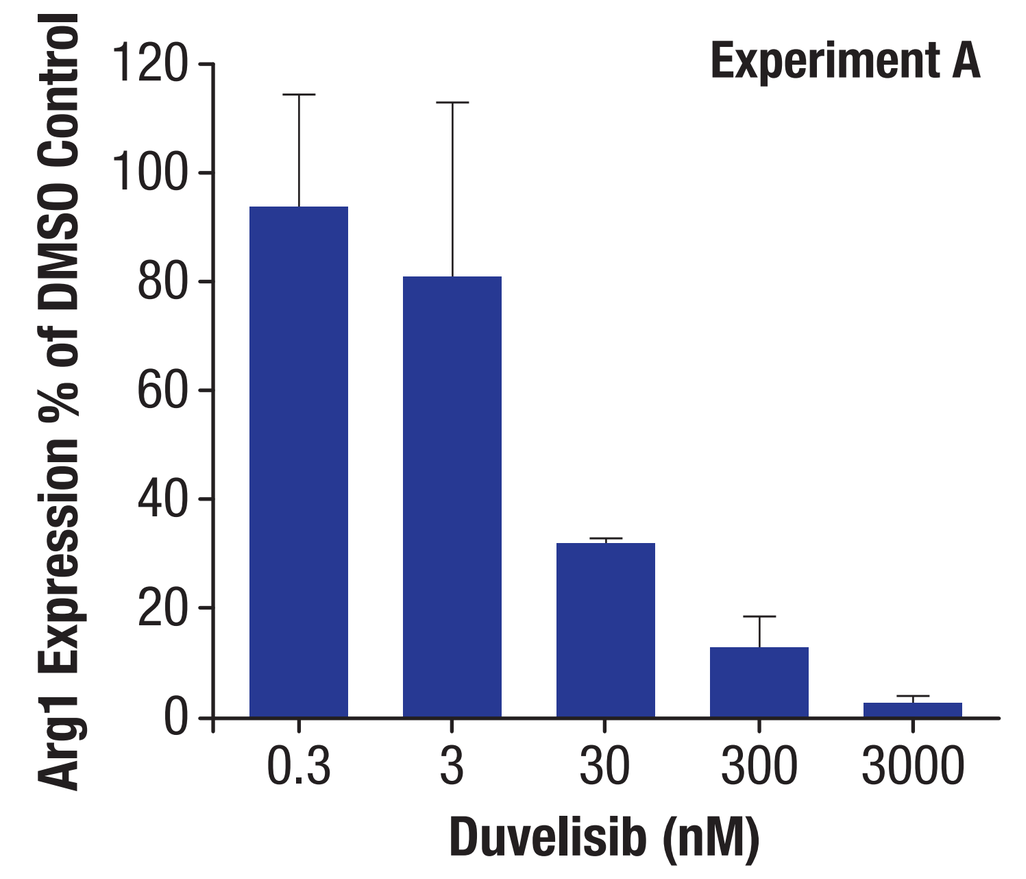
Experiment A
Arg1 Expression % of DMSO Control
120
100
80
60
40
20
0
0.3
3
30
300
3000
Duvelisib (nM)

Drug	Mechanism of Action	Experiment A	Experiment B
Duvelisib	Dual PI3K-δ,γ Inhibitor	15 nM	4 nM
IPI-3063[5]	PI3K-δ inhibitor	Not Tested	Only active at highest concentration (1 μM)
IPI-549	PI3K-γ Inhibitor	82 nM	44 nM

Figure 8: Median Percent Change from Baseline in Serum Analytes following DO/DR Treatment (Combined Data)

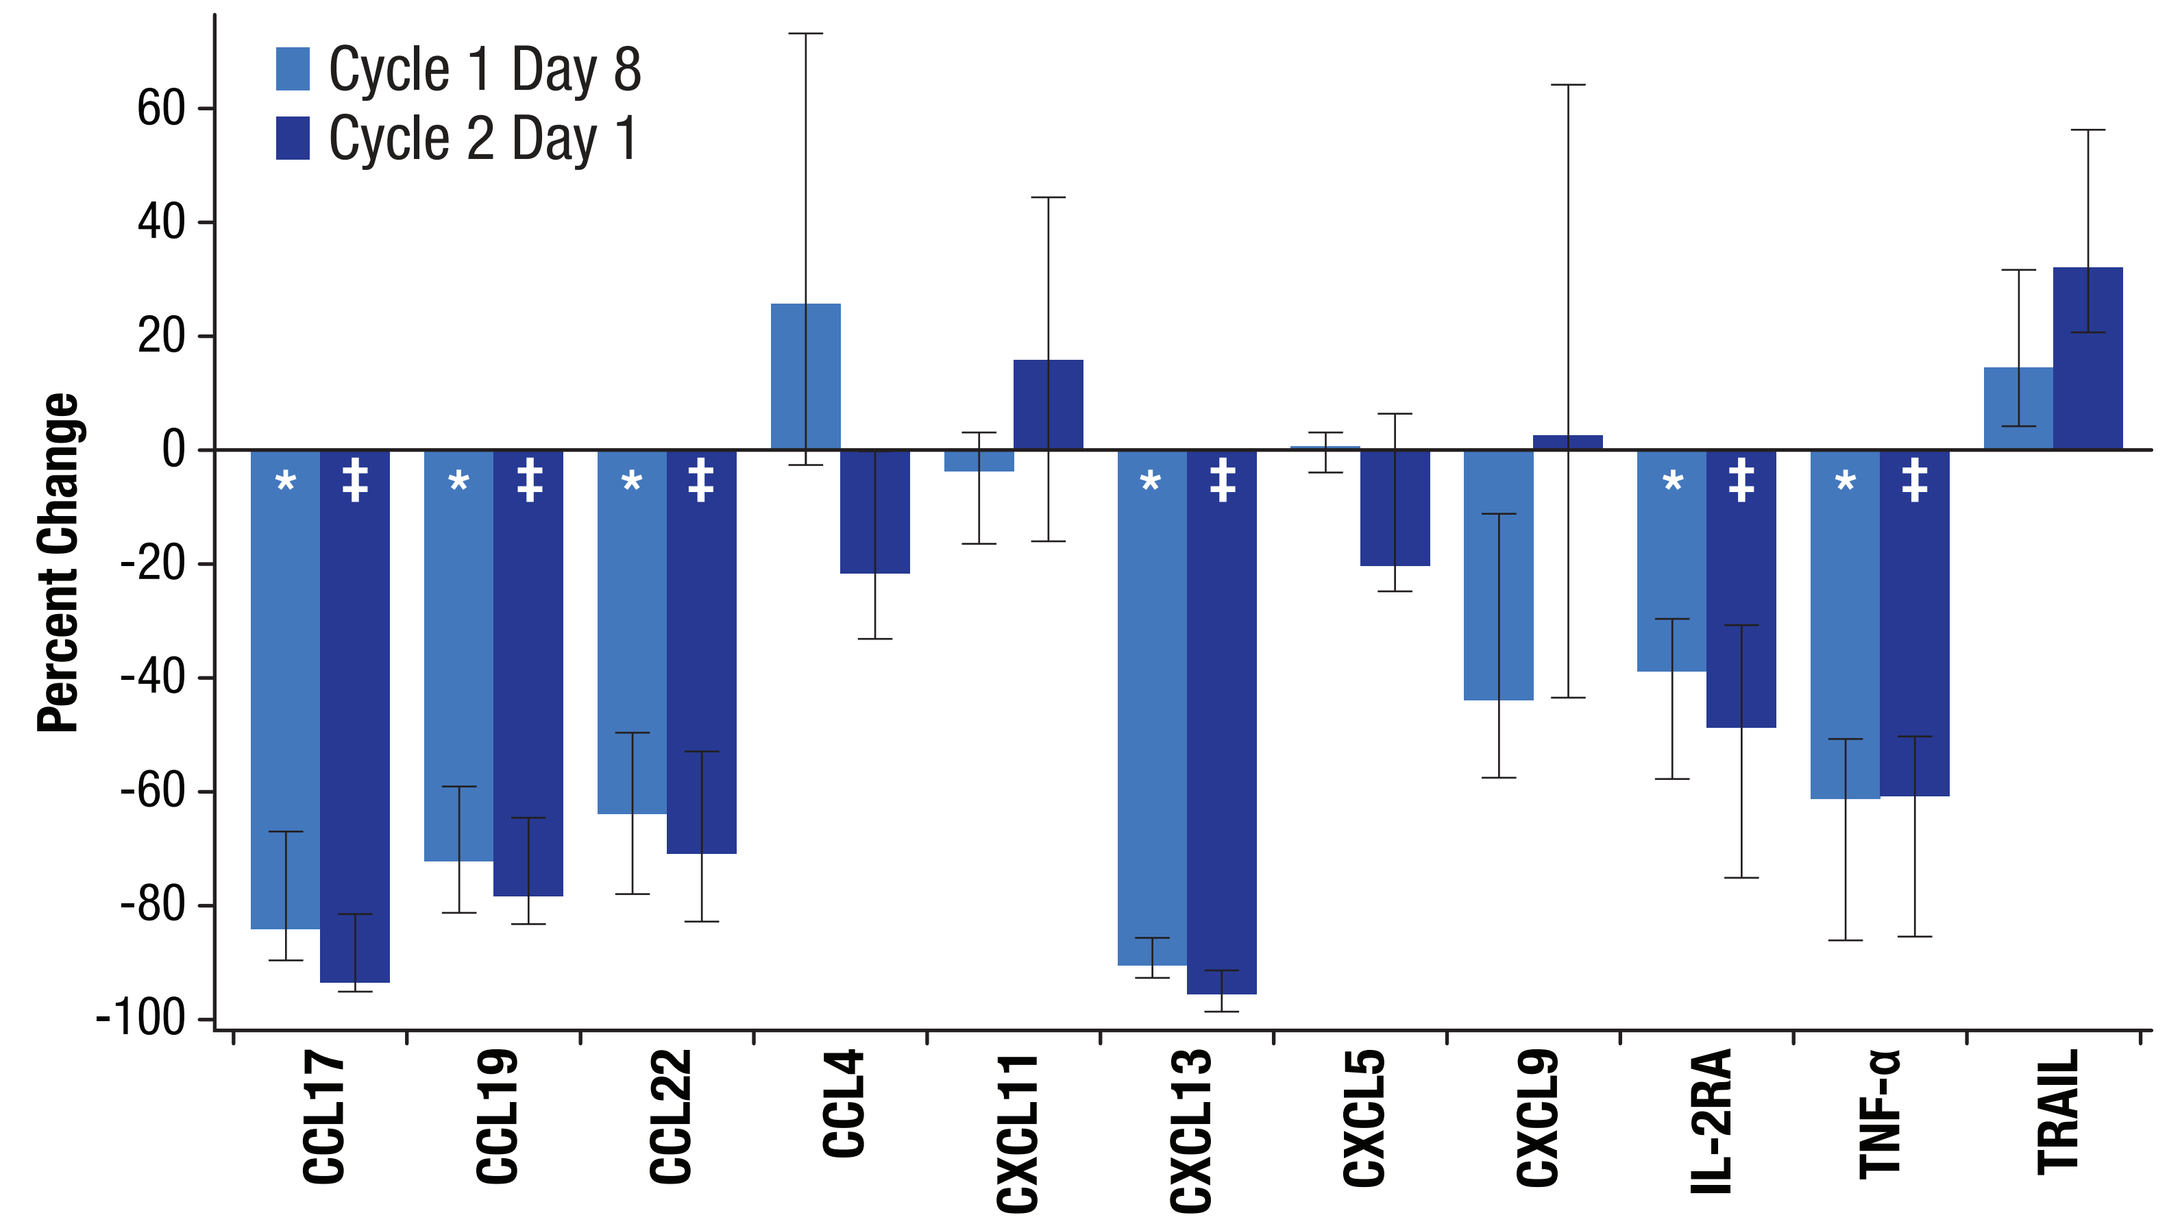
Cycle 1 Day 8
Cycle 2 Day 1
Percent Change
60
40
20
0
-20
-40
-60
-80
-100
CCL17
CCL19
CCL22
CCL4
CXCL11
CXCL13
CXCL5
CXCL9
IL-2RA
TNF-α
TRAIL

- Analytes decreased with duvelisib monotherapy in R/R iNHL were also decreased in this untreated FL population (CCL17, CCL22, CXCL13, TNF-α); modulation of these factors may impact tumor cell migration and recruitment of regulatory T cells[2]
- Decrease in serum chemokine and cytokine shows biological activity via modulation of the TME

Error bars represent 25th & 75th percentiles. Paired t-test with Bonferroni correction to evaluate change from baseline: * $p < 0.005$ and ‡ $p < 0.008$.

Duvelisib Exhibits Robust Dual Inhibition of PI3K-δ,γ Compared to Isoform Class-Specific Compounds

δ,γ Inhibition as Measured by IC_{50}

- Whole blood assay IC_{50} values encompass enzyme inhibition, cell penetration and protein binding of inhibitors **(see items marked with § at left)**
- Duvelisib (IC_{50} 0.4 ± 0.1 μM) and idelalisib (IC_{50} 1.0 ± 0.2 μM) potently inhibited LPS-induced human monocytes via PI3K-δ compared with the PI3K-γ-selective IPI-549 (IC_{50} 12 ± 0.5 μM)
- For TGR-1202, the IC_{50} (25 ± 8 μM) was below the RP2D clinical exposure
- Duvelisib (IC_{50} 0.5 ± 0.2 μM) and IPI-549 (IC_{50} 1.6 ± 0.2 μM) potently inhibited PI3K-γ-dependent fMLP-stimulated human monocytes compared to idelalisib (IC_{50} 9.4 ± 2.3) and TGR-1202 (IC_{50} 55 ± 16 μM)

Duvelisib is a Dual Inhibitor of PI3K-δ & PI3K-γ

Figure 9: PI3K-δ and PI3K-γ inhibition by drug

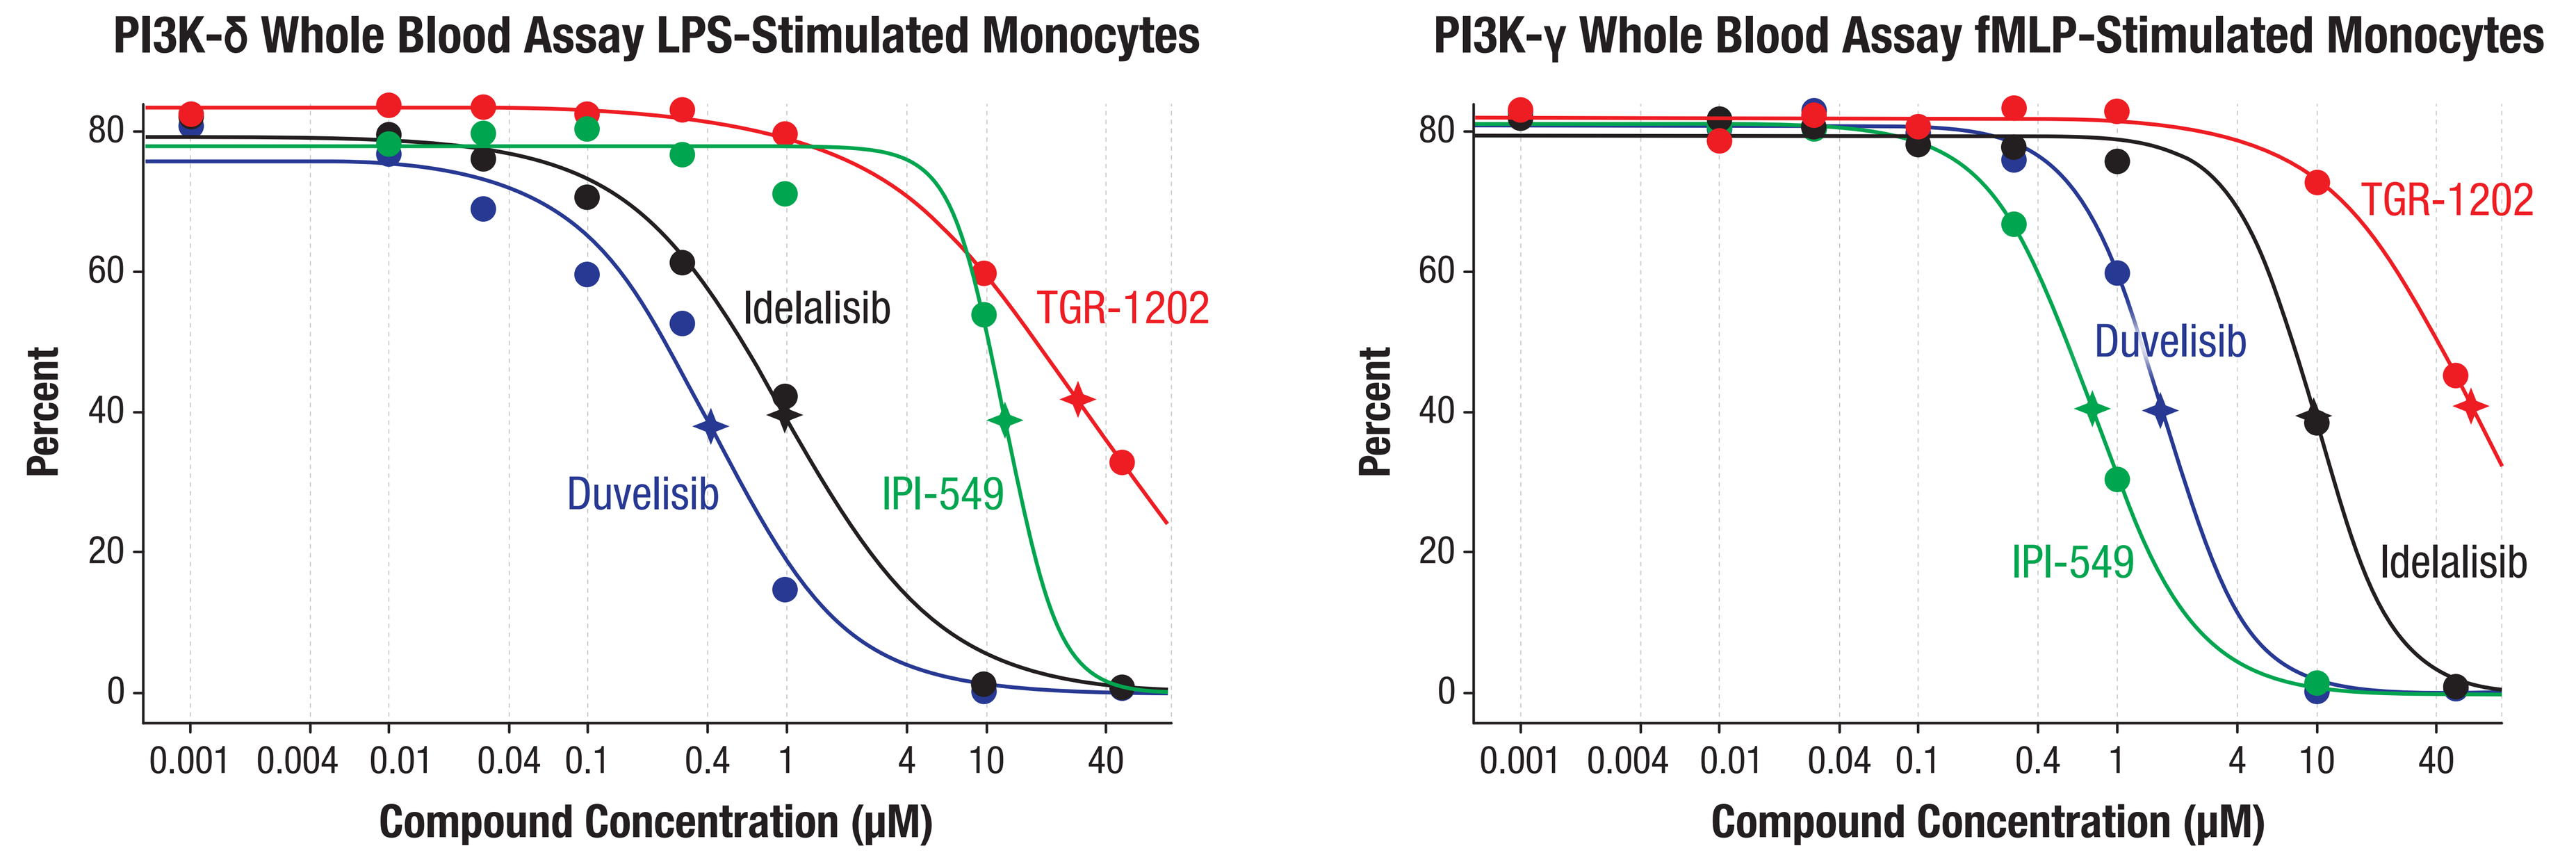
PI3K-δ Whole Blood Assay LPS-Stimulated Monocytes
Percent
Idelalisib
TGR-1202
Duvelisib
IPI-549
Compound Concentration (μM)
PI3K-γ Whole Blood Assay fMLP-Stimulated Monocytes
Percent
TGR-1202
Duvelisib
IPI-549
Idelalisib
Compound Concentration (μM)

Figure 10: % Inhibition of Whole Blood PI3K-δ and PI3K-γ at Clinical C_{max}

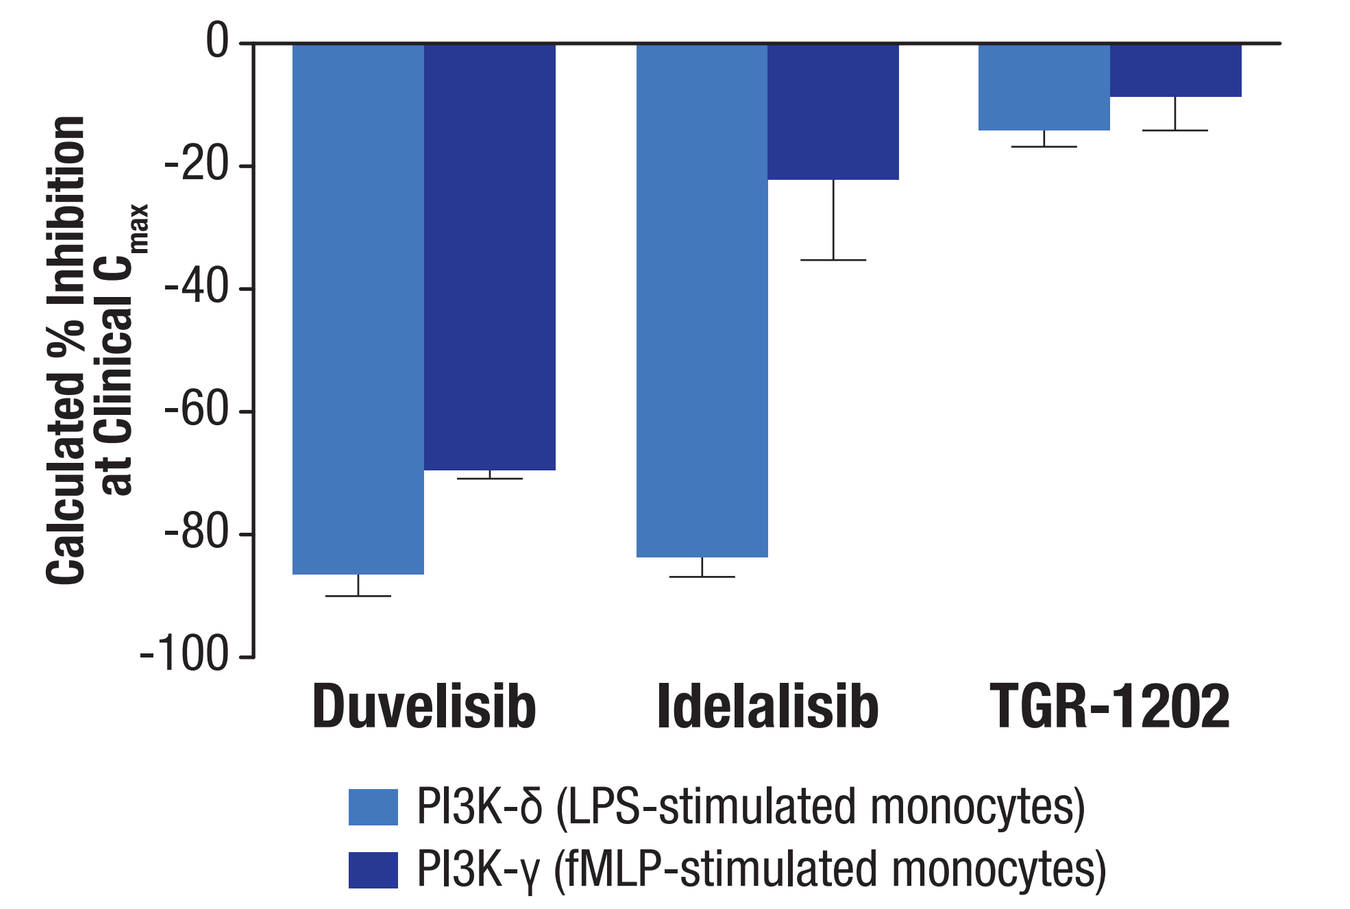
Calculated % Inhibition at Clinical Cmax
0
-20
-40
-60
-80
-100
Duvelisib
Idelalisib
TGR-1202
PI3K-δ (LPS-stimulated monocytes)
PI3K-γ (fMLP-stimulated monocytes)

Table 2: Potencies vs PI3K-δ and PI3K-γ in Human Whole Blood Assays

Drug	Mechanism of Action	PI3K-δ IC_{50} (μM)	PI3K-γ IC_{50} (μM)	C_{max} (ss) (μM)
Duvelisib	Dual PI3K-δ,γ Inhibitor	0.4 ± 0.1	1.6 ± 0.2	2.5
Idelalisib	PI3K-δ Inhibitor	1.0 ± 0.2	9.4 ± 2.3	4.8
IPI-549	PI3K-γ Inhibitor	12 ± 0.5	0.5 ± 0.2	9.1
TGR-1202		25 ± 8	55 ± 16	9.2

Green = at or below predicted IC_{50} Red = above predicted IC_{50}

- Duvelisib human PK: C_{max} @ 25 mg BID (RP2D) = 1062 ng/ml; MW = 417 g/mol
- Idelalisib human PK:[7] C_{max} @ 150 mg BID (RP2D) = 2000 ng/ml; MW = 415 g/mol
- IPI-549 human PK:[6] C_{max}-ss @ 60 mg QD (RP2D) = 4800 ng/ml, MW = 529 g/mol
- TGR-1202 human PK:[8] C_{max}-ss @ 800 mg QD (RP2D) = 5276 ng/ml; MW = 572

δ,γ Inhibition, Macrophage Polarization, and Macrophage and T cell Migration in FL Patients

- Results from selective assays demonstrated that duvelisib inhibited PI3K-δ and PI3K-γ for 1-4 hours post-treatment in FL patients
- Consistent with a PI3K-γ mechanism, duvelisib:
 - Inhibited macrophage polarization
 - Reduced CXCL12-induced macrophage migration (duvelisib IC_{50} 51 nM; IPI-549 IC_{50} 85 nM)
 - Blocked CXCL12-induced T cell migration (duvelisib EC_{50} 128 ± 39 nM; IPI-549 EC_{50} 17 ± 17 nM); to a greater extent than with PI3K-δ inhibitor IPI-3063 (IC_{50} 630 ± 71 nM)
- Comparable inhibitory activities were observed for duvelisib and IPI-549 (PI3K-γ inhibitor) in these assays

Conclusions

- Duvelisib inhibits both PI3K-δ and PI3K-γ in non-tumor cells from patients with untreated FL
- Duvelisib's disruption of PI3K-δ,γ function in patients with FL supports its inhibition of the TME through cancer-supportive macrophages and T cells

For More Information

+1-781-292-4200

MedicalAffairs@verastem.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Presented at the 2018 American Society for Clinical Oncology Annual Meeting • Chicago, Illinois, USA • June 1-5, 2018